AACR Poster # 2636

Targeting autotaxin to suppress stromal signaling in the tumor microenvironment to improve outcome to therapy in fibrotic tumor types

Zoë Johnson[1], Marcel A. Deken[1], Ragini Medhi[2], Lauren Maggs[2], Alan Carruthers[2], Anne Cheasty [2], Alessia Tagliavini[3], Andrea Nizzardo[3], Marco Pergher[3], Karolina Niewola-Staszkowska[1], Amy Fraser[2], Lars van der Veen[1], Pritom Shah[2], Luigi Ziviani[4], Stefano Milleri[4] & Michael Lahn[1]

[1]iOnctura SA, Avenue de Sécheron 15, 1202 Geneva, Switzerland; [2]Cancer Research UK Therapeutic Discovery Laboratories, Babraham Research Campus, Cambridge, UK; [3]Evotec, Verona, Italy; [4]Centro Ricerche Cliniche di Verona srl, Italy

Background

Recent findings indicate that in cancer, host immune suppression and resistance to therapy is orchestrated by the presence and activity of stromal cells in the tumor microenvironment. Therefore novel approaches to overcome stromal mediated resistance to therapy represent a potential strategy to improve the outcome to chemotherapy and immune checkpoint blockade, particularly in tumors that have a fibrotic microenviroment. Autotaxin is an enzyme that converts lysophosphatidylcholine (LPC) to lysophosphatidic acid (LPA), a key pathway that is aberrantly activated in fibrosis. LPA has also been described to act directly on tumor cells, promoting their growth and proliferation via G protein-coupled LPA Receptors (LPARs). IOA-289 is a novel, potent, selective and orally bioavailable inhibitor of autotaxin, a target with known clinical application in fibrotic diseases and with a strong rationale for exploration in cancer.

Methods

- Analysis of ATX (gene name ENPP2) expression in various cancers and sample types using TCGA and GTEX data.
- CA19-9 and ATX levels measured in patients' plasma using a commercial ELISA kit.
- Growth and proliferation of MIA PaCa-2 cells were assessed upon treatment using time-lapse imaging (Incucyte). Nuclight rapid red dye was used for live-cell nuclear labelling. For condition media generation, 0082T cells were treated with 12 µM IOA-289 or 0.1% DMSO for 72 hours post seeding in low nutrient serum free media supplemented with 0.5% BSA. Supernatant media was collected and concentrated to 30X.
- The 0082T cell line was derived from primary CAF sample 0082 and prepared from human PDAC surgical specimens PMID: 30837243, provided by Dr. Andrew Lowy (University of California, San Diego)
- Human fibroblast cell line HFFF2 was stimulated with rhTGFβ and supernatants analysed 48h later by ELISA for soluble mediators.
- *In vivo* efficacy was studied in an immuno-competent, orthotopic mouse model of pancreatic cancer derived from a GEMM PDAC model (KRAS (G12D); $p53^{-/-}$).
- PK/PD studies were performed following a single oral dose of IOA-289 in mice, and plasma LPA was used as a PD biomarker.
- Plasma concentrations of IOA-289 and LPA were measured following a single oral dose of IOA-289 in a healthy volunteer study.

PDAC samples have high ENPP2 / ATX levels

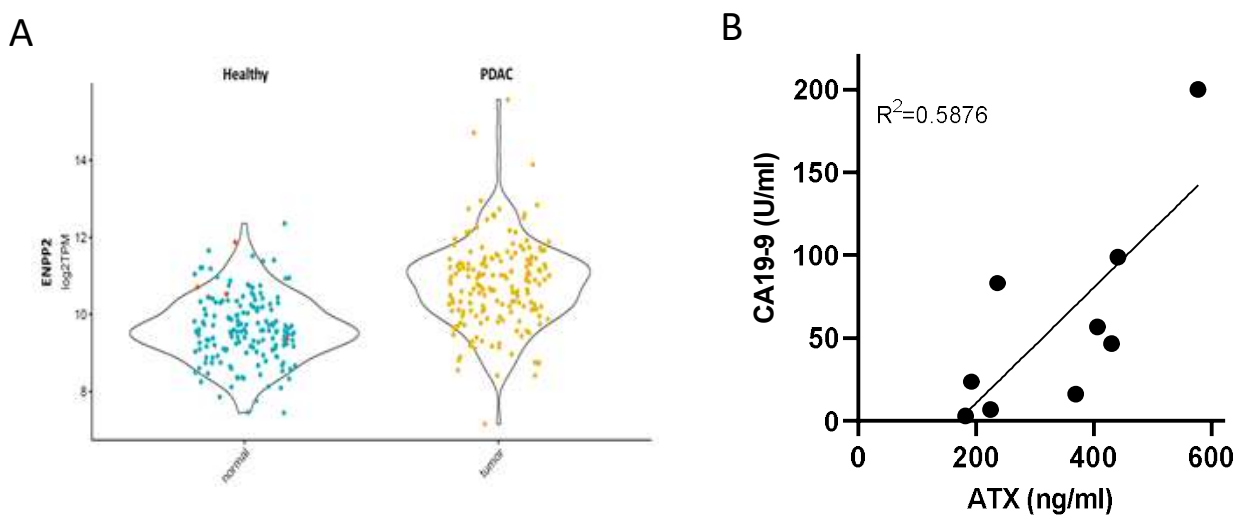
A
Healthy
PDAC
ENPP2
log2TPM
normal
tumor
B
R^2=0.5876
CA19-9 (U/ml)
ATX (ng/ml)

Figure 1. ATX expression in PDAC: Pancreatic adenocarcinoma (PDAC) samples have high ENNP2 expression compared to normal tissue (Fig. 1A). ATX levels in PDAC patient plasma correlate with CA19-9 levels (Fig. 1B).

IOA-289 inhibits PDAC cells and CAFs *in vitro*

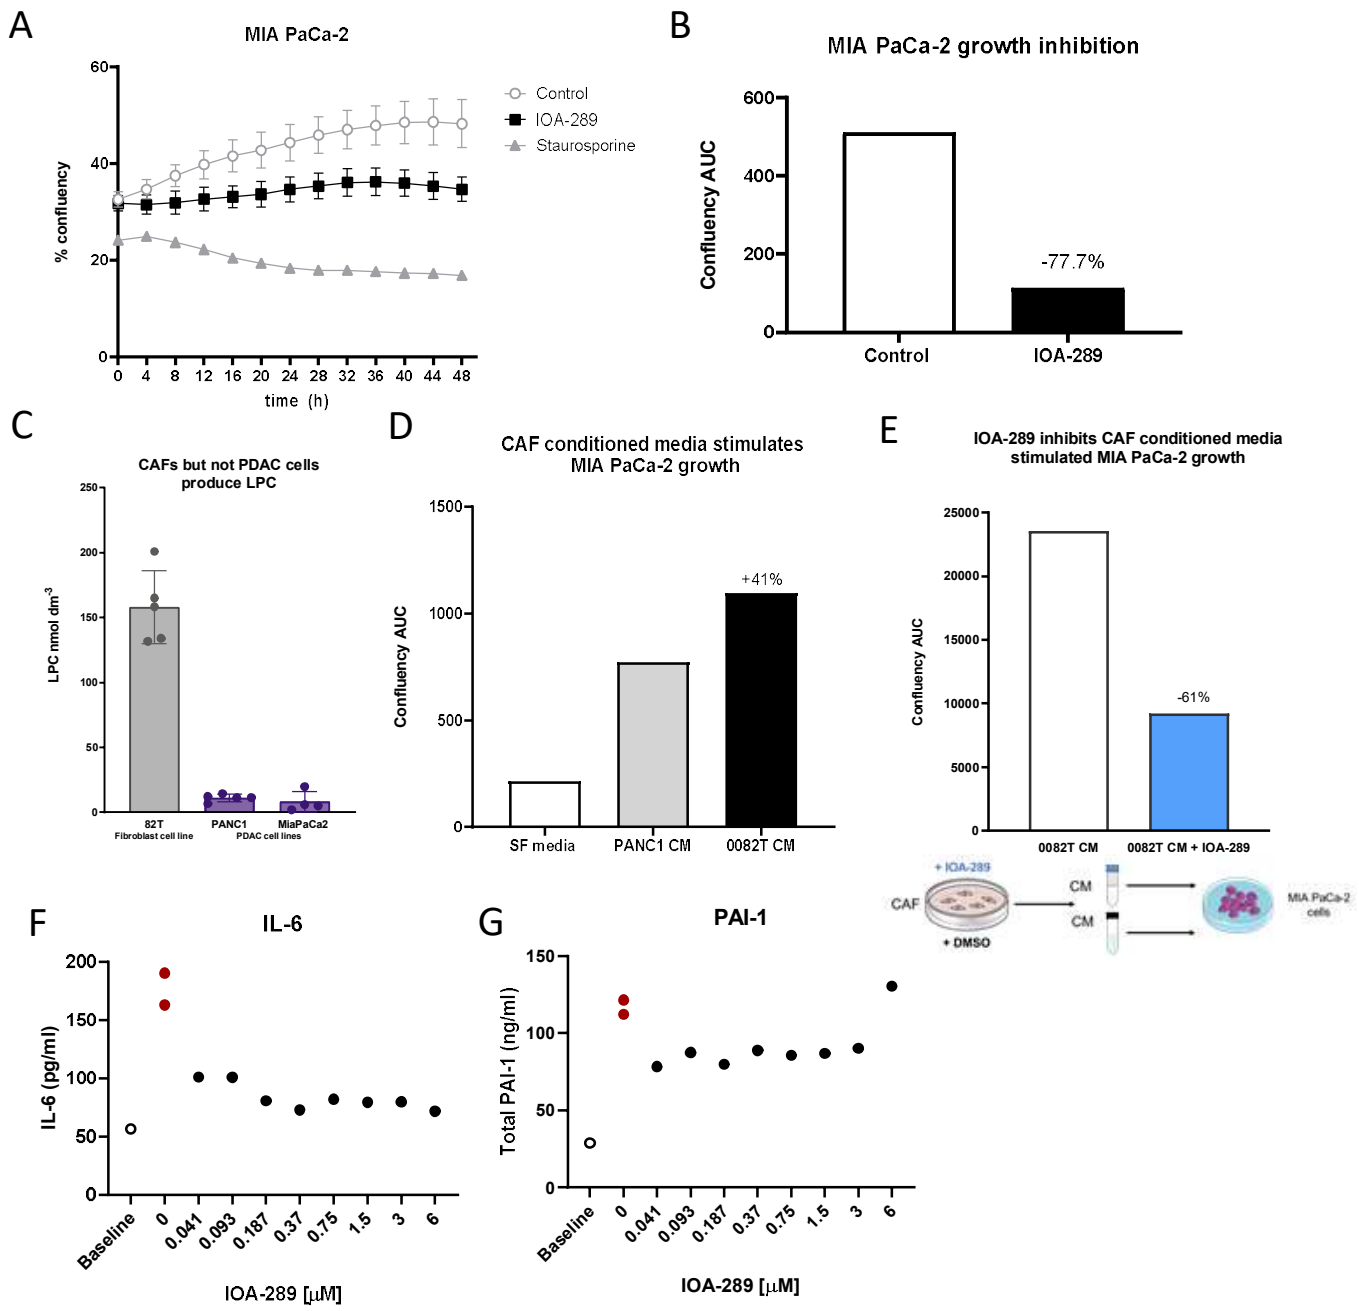
A
MIA PaCa-2
Control
IOA-289
Staurosporine
% confluency
time (h)
B
MIA PaCa-2 growth inhibition
Confluency AUC
-77.7%
Control
IOA-289
C
CAFs but not PDAC cells produce LPC
LPC nmol dm-3
82T
Fibroblast cell line
PANC1
MiaPaCa2
PDAC cell lines
D
CAF conditioned media stimulates MIA PaCa-2 growth
Confluency AUC
+41%
SF media
PANC1 CM
0082T CM
E
IOA-289 inhibits CAF conditioned media stimulated MIA PaCa-2 growth
Confluency AUC
-61%
0082T CM
0082T CM + IOA-289
CAF
+ IOA-289
+ DMSO
CM
CM
MIA PaCa-2 cells
F
IL-6
IL-6 (pg/ml)
Baseline
IOA-289 [µM]
G
PAI-1
Total PAI-1 (ng/ml)
Baseline
IOA-289 [µM]

Figure 2. IOA-289 inhibits the growth and proliferation of PDAC cell lines *in vitro* (Fig. 2A, B). CAFs but not PDAC cells are the main source of LPC (Fig. 2C). Conditioned media (CM) from CAFs stimulate the growth of PDAC cells (Fig. 2D). IOA-289 inhibits the growth stimulating effect of CAFs (Fig. 2E). IOA-289 also inhibits key mediators secreted by activated fibroblasts that play a role in tumor-stroma-immune cross-talk (Fig. 2F-H).

IOA-289 inhibits PDAC growth *in vivo*

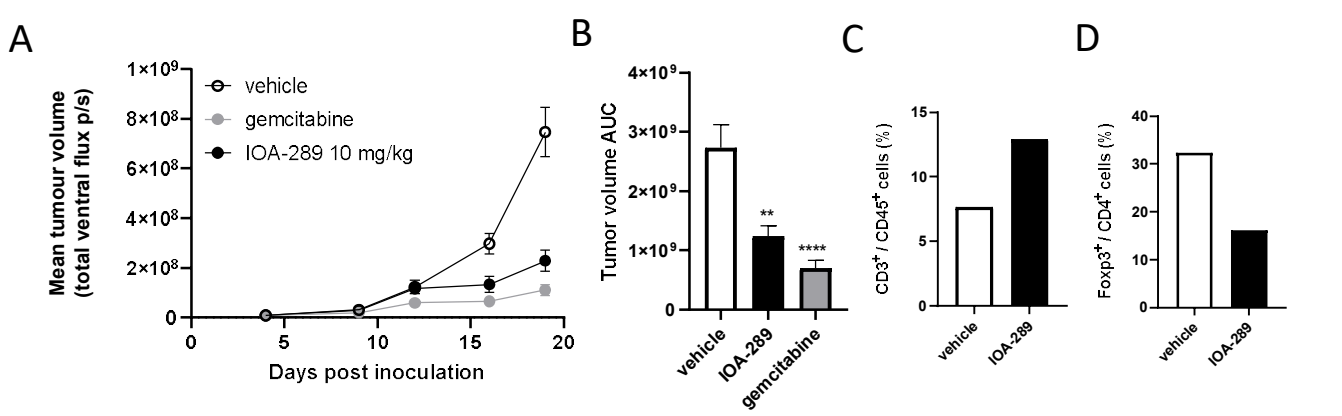
A
vehicle
gemcitabine
IOA-289 10 mg/kg
Mean tumour volume (total ventral flux p/s)
Days post inoculation
B
Tumor volume AUC
**

vehicle
IOA-289
gemcitabine
C
CD3+ / CD45+ cells (%)
vehicle
IOA-289
D
Foxp3+ / CD4+ cells (%)
vehicle
IOA-289

Figure 3. Effect of IOA-289 on PDAC growth *in vivo*: IOA-289 has single agent activity on orthotopic pancreatic cancer growth *in vivo* (Fig. 3A-B). IOA-289 increased the infiltration of $CD3^+$ cells into the tumors (Fig. 3C), with a simultaneous reduction in Treg cells (Fig. 3D).

IOA-289 PK/PD from mouse to human

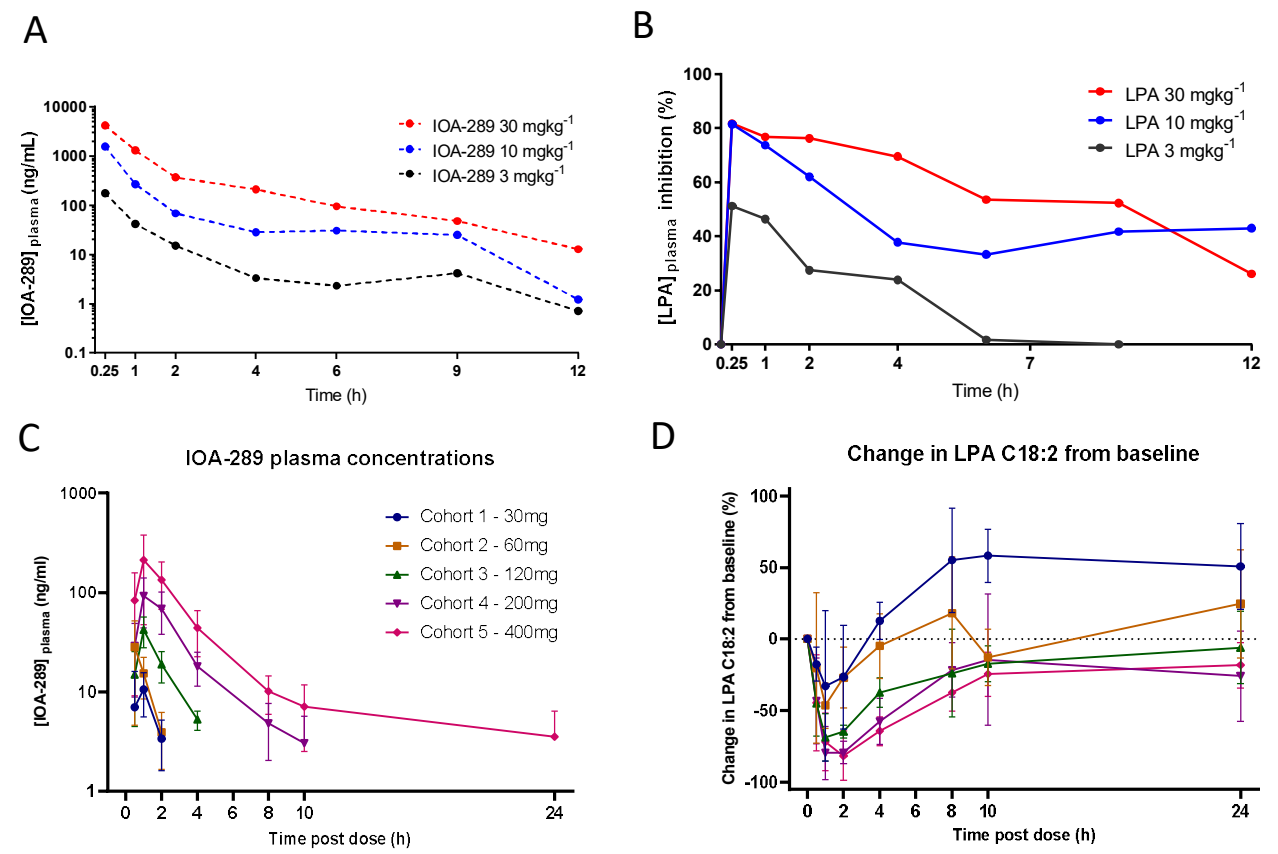
A
IOA-289 30 mgkg-1
IOA-289 10 mgkg-1
IOA-289 3 mgkg-1
[IOA-289] plasma (ng/mL)
Time (h)
B
LPA 30 mgkg-1
LPA 10 mgkg-1
LPA 3 mgkg-1
[LPA] plasma inhibition (%)
Time (h)
C
IOA-289 plasma concentrations
Cohort 1 - 30mg
Cohort 2 - 60mg
Cohort 3 - 120mg
Cohort 4 - 200mg
Cohort 5 - 400mg
[IOA-289] plasma (ng/ml)
Time post dose (h)
D
Change in LPA C18:2 from baseline
Change in LPA C18:2 from baseline (%)
Time post dose (h)

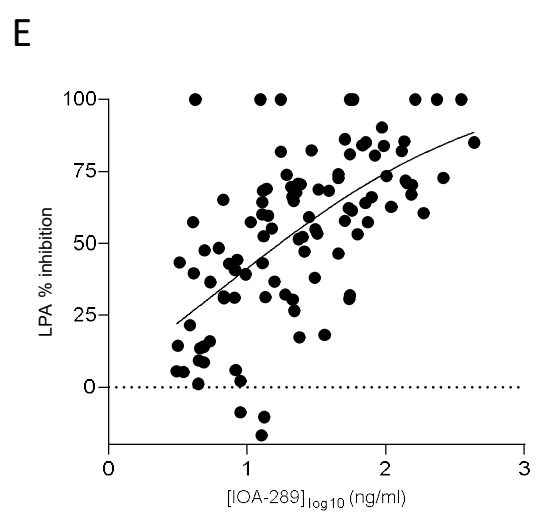
E
LPA % inhibition
[IOA-289] log10 (ng/ml)

Figure 4: Mice dosed with IOA-289 p.o. showed dose-dependent reduction of circulating LPA C18:2 with an ED_{50} value at 1 h post-dose of around 3 mg/kg (Fig. 4A-B). Plasma exposure was measured up to 24 h post dose in HV subjects (Fig. 4C). LPA C18:2 in plasma was measured at baseline and up to 24 h post dose (Fig. 4D). Using the PK and PD data, the IC_{50} of IOA-289 was calculated as 15 ng/mL in human plasma (Fig. 4E).

Conclusion

- ATX is elevated in PDAC and directly impacts cancer cell line growth *in vitro*
- Cancer associated fibroblasts stimulate PDAC cell growth *in vitro* and ATX is a key mediator driving the tumor supporting action of CAFs.
- ATX is a novel solid tumor target that orchestrates the interplay between stromal cells, immune cells and tumor cells.
- IOA-289 is the first ATX inhibitor to be investigated in solid tumors and here we demonstrate that IOA-289 reduced tumor burden in a mouse model of PDAC.
- IOA-289 is a potent, orally bioavailable ATX inhibitor with a PK/PD relationship directly translatable from mouse to human with a plasma IC_{50} of 15 ng/mL.
- IOA-289 is in clinical development for the treatment of solid tumors burdened with a high degree of fibrosis.

z.johnson@ionctura.com & m.lahn@ionctura.com

AACR 2022 Annual Meeting | April 8-13, 2022

Conflict of Interest Statement:
ZJ, MD, KNS, LV & ML are employees and shareholders of iOnctura