Characterization of the non-ATP competitive PI3Kδ inhibitor IOA-244 in lymphoma models: from single agent to combination screen and clinical investigation

Poster n. 293

Francesco Bertoni [1,2], Chiara Tarantelli [1], Filippo Spriano [1], Luciano Cascione [1], Elisa Civanelli [1], Eleonora Cannas [1], Afua A Mensah [1], Alberto J Arribas [1], Sara Napoli [1], Andrea Rinaldi [1], Anastasios Stathis [2], Karolina Niewola-Staszkowska [3], Giusy Di Conza [3], Michael Lahn [3], Armando Santoro [4], Carmelo Carlo-Stella [4]

[1] Institute of Oncology Research, Faculty of Biomedical Sciences, USI, Bellinzona, Switzerland;
[2] Oncology Institute of Southern Switzerland, Bellinzona, Switzerland;
[3] IOnctura SA, Geneva, Switzerland;
[4] Humanitas University and IRCSS Humanitas Research Hospital, Milano-Rozzano, Italy.

BACKGROUND

IOA-244 is a highly selective PI3Kδ inhibitor with a differentiated chemical mechanism of action from other PI3K inhibitors. IOA-244 is currently being investigated in patients with solid tumors and lymphoma (NCT04328844). Here, we present preclinical and initial clinical data in lymphomas.

MATERIAL AND METHODS

Anti-proliferative activity by MTT assay at 72h. Transcriptome analysis by total and targeted RNA-Seq on Illumina platform. Follicular lymphoma patients (pts) were treated at 20 mg and 80 mg QD continuous dosing and assessed for safety.

CONCLUSIONS

Single-agent IOA-244 has moderate activity in vitro in lymphoma, correlated with PI3Kδ expression. Given its favorable monotherapy safety profile, IOA-244 may be used in combination with drugs identified in the present pharmacological screen.

Contact: Francesco Bertoni, MD, Institute of Oncology Research, via Francesco Chiesa 5, 6500 Bellinzona, Switzerland; phone: +41 58 666 72 06; e-mail: francesco.bertoni@ior.usi.ch

Study sponsored by iOnctura, by Innosuisse (Project no. 55179.1 IP-LS) and by the Swiss Cancer Research grant KFS-4713-02-2019.

RESULTS

IOA-244 showed moderate dose-dependent anti-proliferative activity, measured as area under the curve, across 66 human B and T cell lymphoma models, with IC50s < 10 μM in 18 cell lines (Figure 1A). The activity correlated with PI3Kδ expression as measured by total RNA-Seq (R^2=0.18, P=0.0009; 59 B and T cell lymphomas) or by HTG EdgeSeq Oncology Biomarker panel (R^2=0.13, P=0.028; 36 B cell lymphomas) (Figure 1B-C).

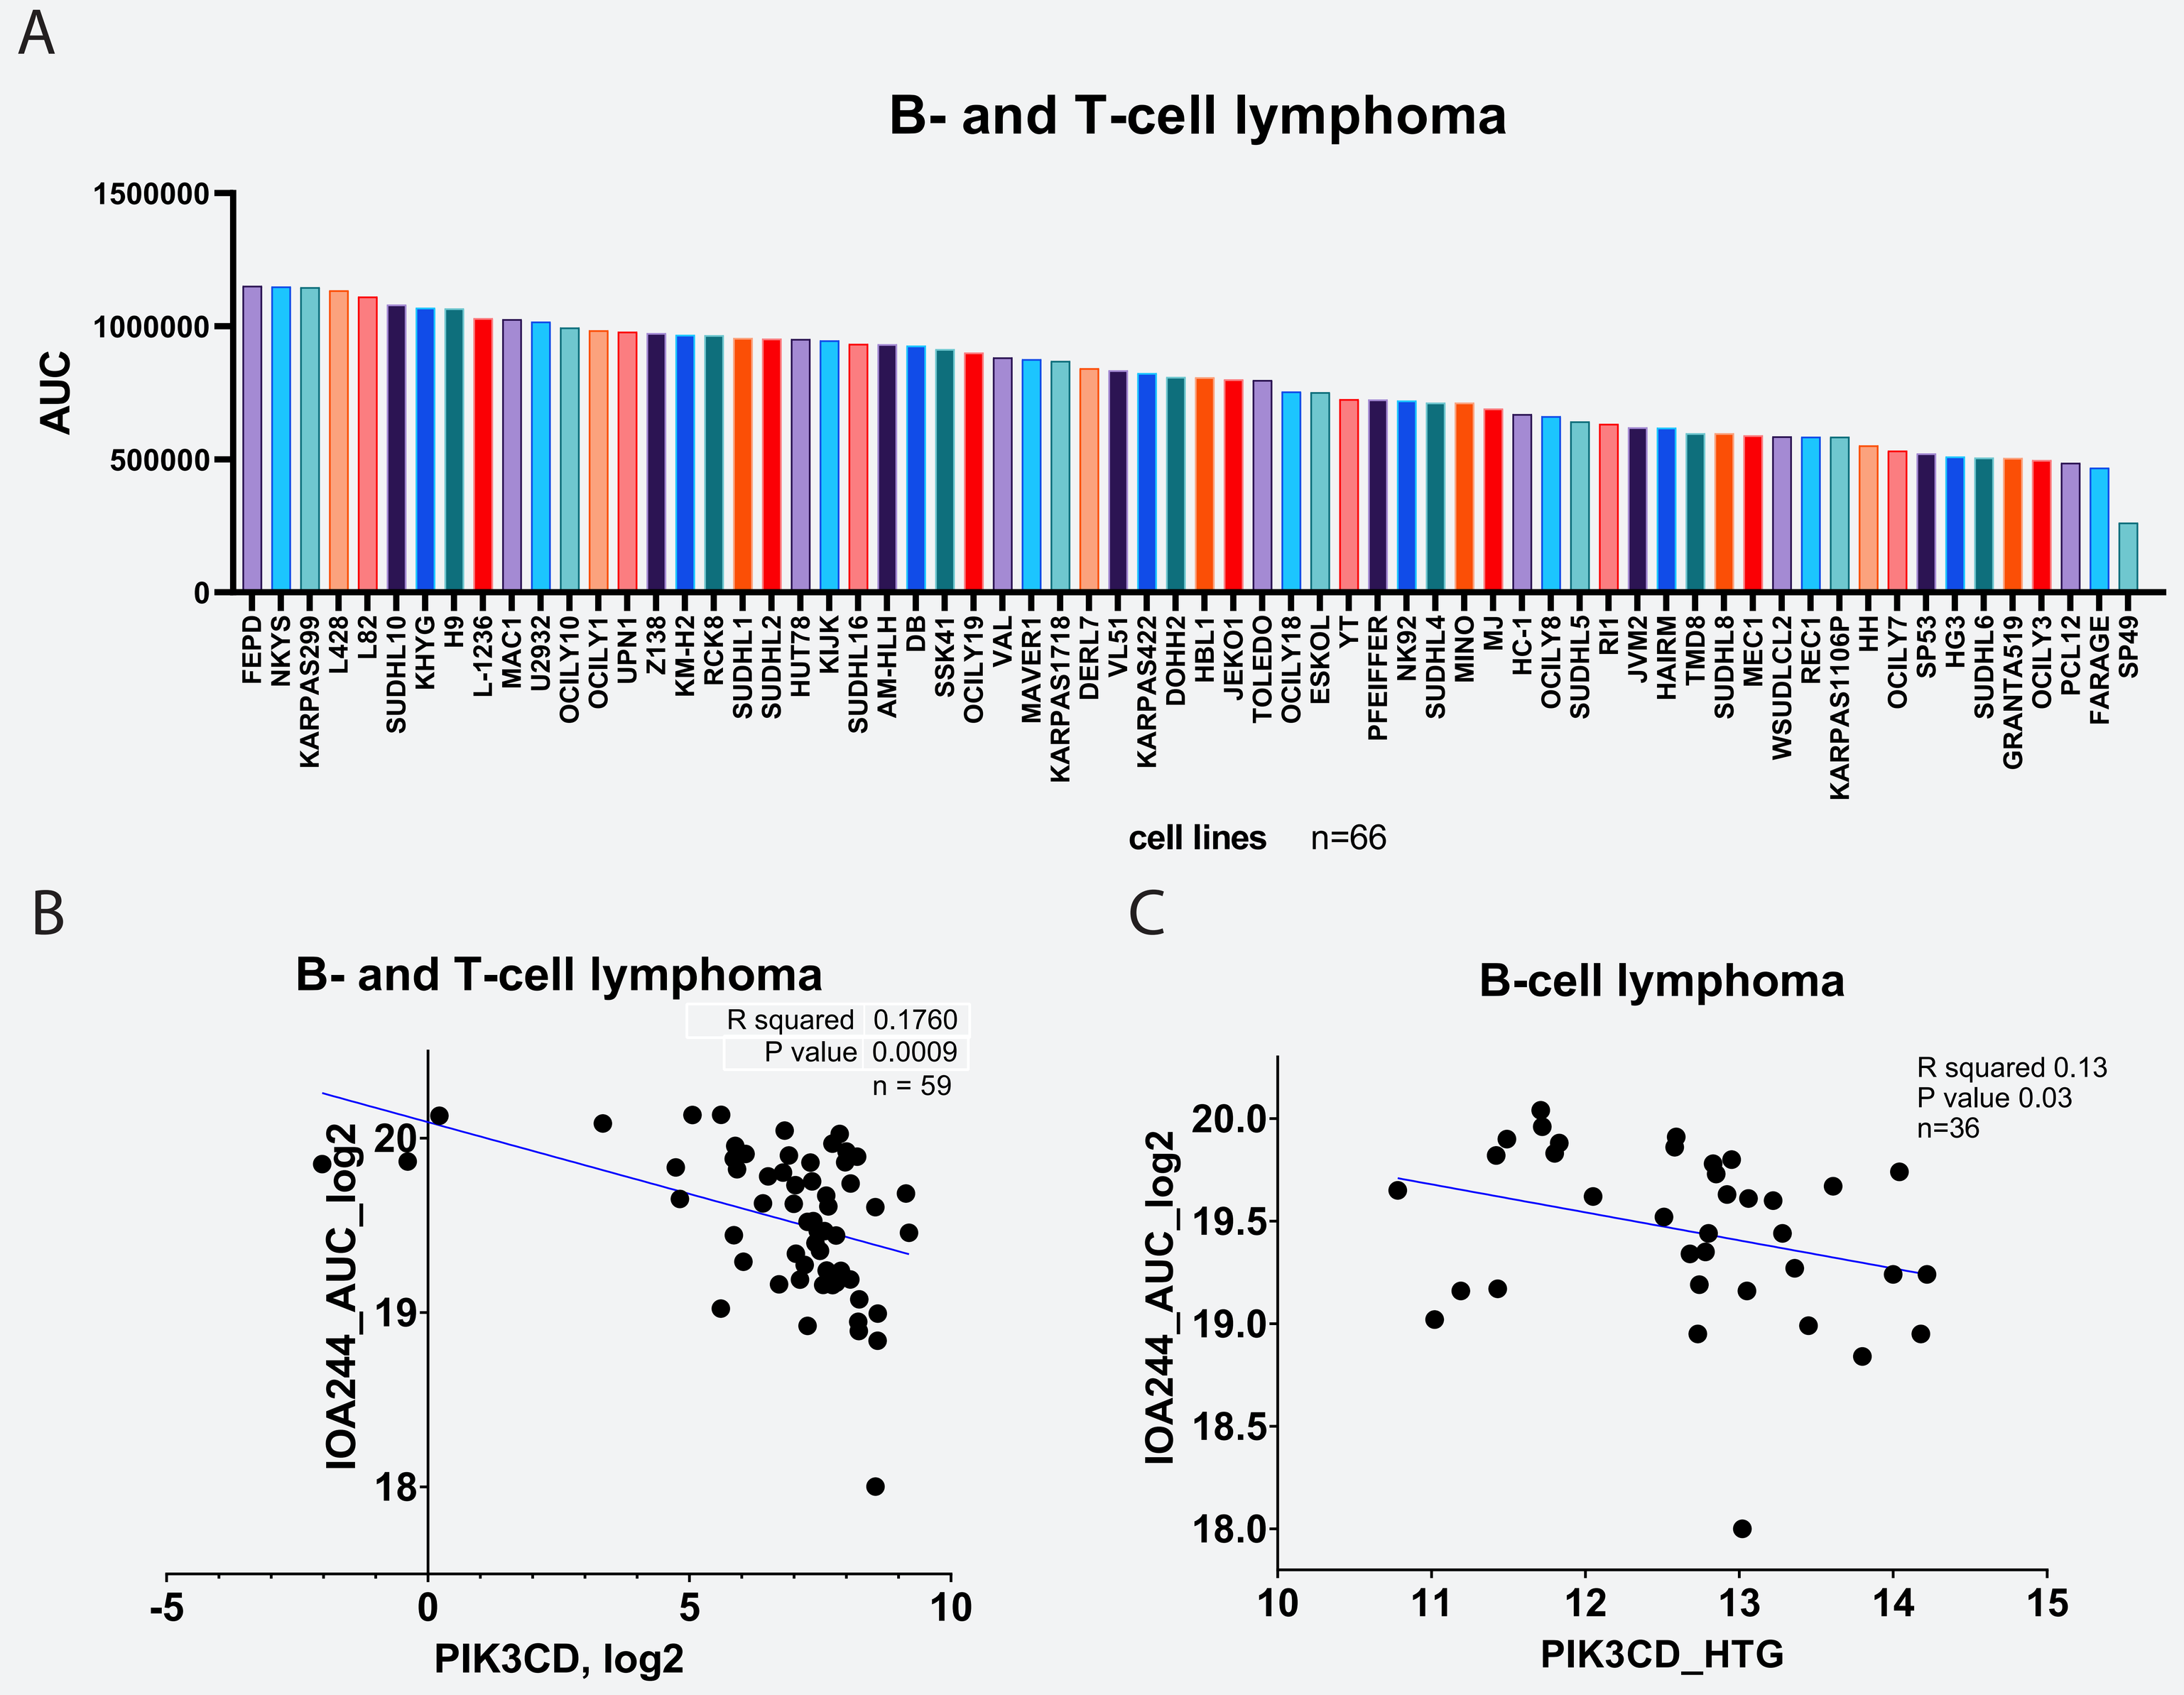
A
B- and T-cell lymphoma
AUC
1500000
1000000
500000
0
FEPD NKYS KARPAS299 L428 L82 SUDHL10 KHYG H9 L-1236 MAC1 U2932 OCILY10 OCILY1 UPN1 Z138 KM-H2 RCK8 SUDHL1 SUDHL2 HUT78 KIJK SUDHL16 AM-HLH DB SSK41 OCILY19 VAL MAVER1 KARPAS1718 DERL7 VL51 KARPAS422 DOHH2 HBL1 JEKO1 TOLEDO OCILY18 ESKOL YT PFEIFFER NK92 SUDHL4 MINO MJ HC-1 OCILY8 SUDHL5 RI1 JVM2 HAIRM TMD8 SUDHL8 MEC1 WSUDLCL2 REC1 KARPAS1106P HH OCILY7 SP53 HG3 SUDHL6 GRANTA519 OCILY3 PCL12 FARAGE SP49
cell lines n=66
B
B- and T-cell lymphoma
R squared 0.1760
P value 0.0009
n = 59
IOA244_AUC_log2
PIK3CD, log2
C
B-cell lymphoma
R squared 0.13
P value 0.03
n=36
IOA244_AUC_log2
PIK3CD_HTG

Figure 1. Antiproliferative activity of IOA-244 in in a panel of different subtypes of lymphoma and correlation between AUC and PIK3CD transcript expression.
(A) Cell lines were treated with increasing concentration of IOA-244 compound. MTT assay was performed to evaluate anti-tumoral activity of the drug and Area Under the Curve was evaluated (AUC). Y-axis: AUC values. Scatter plots of (B) B and T cell lymphoma in correlation with total RNA-Seq and (C) B cell lymphoma cell lines in correlation with targeted RNA-Seq HTG panel are showed here. PIK3CD RNA expression was estrapolated by RNA sequencing of each cell line at baseline conditions. R², Spearman correlation.

In MCL SP53 cells, IOA-244 (5μM; 24, 48, 72h) downregulated BCR, MYD88, NF-κB, MTOR and NOTCH signaling and upregulated cell cycle arrest genes (adj.P<10-10) (Figure 2). Changes overlapped with signatures obtained with other PI3K and BTK inhibitors (adj.P $<10^{-10}$).

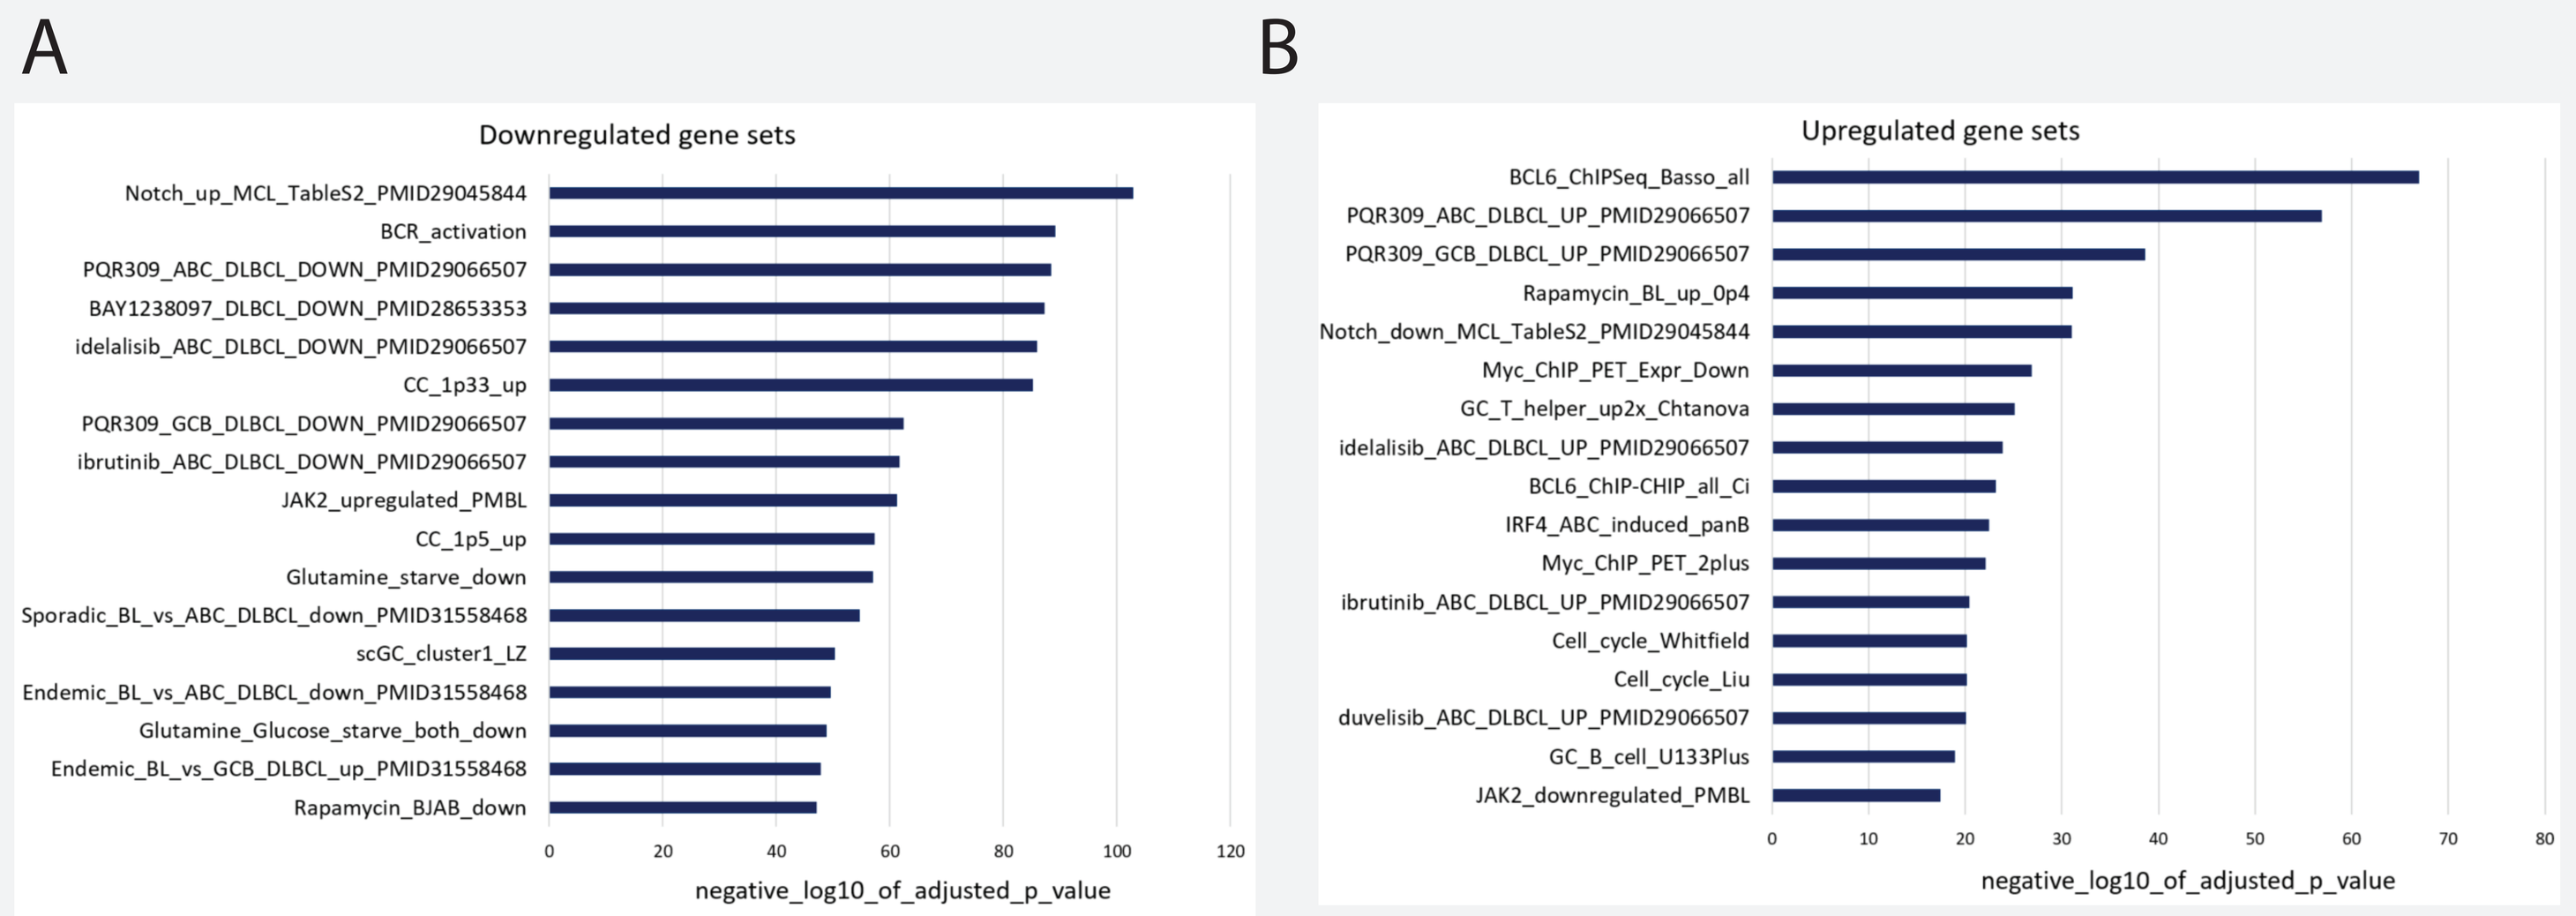
A
Downregulated gene sets
Notch_up_MCL_TableS2_PMID29045844
BCR_activation
PQR309_ABC_DLBCL_DOWN_PMID29066507
BAY1238097_DLBCL_DOWN_PMID28653353
idelalisib_ABC_DLBCL_DOWN_PMID29066507
CC_1p33_up
PQR309_GCB_DLBCL_DOWN_PMID29066507
ibrutinib_ABC_DLBCL_DOWN_PMID29066507
JAK2_upregulated_PMBL
CC_1p5_up
Glutamine_starve_down
Sporadic_BL_vs_ABC_DLBCL_down_PMID31558468
scGC_cluster1_LZ
Endemic_BL_vs_ABC_DLBCL_down_PMID31558468
Glutamine_Glucose_starve_both_down
Endemic_BL_vs_GCB_DLBCL_up_PMID31558468
Rapamycin_BJAB_down
negative_log10_of_adjusted_p_value
B
Upregulated gene sets
BCL6_ChIPSeq_Basso_all
PQR309_ABC_DLBCL_UP_PMID29066507
PQR309_GCB_DLBCL_UP_PMID29066507
Rapamycin_BL_up_0p4
Notch_down_MCL_TableS2_PMID29045844
Myc_ChIP_PET_Expr_Down
GC_T_helper_up2x_Chtanova
idelalisib_ABC_DLBCL_UP_PMID29066507
BCL6_ChIP-CHIP_all_Ci
IRF4_ABC_induced_panB
Myc_ChIP_PET_2plus
ibrutinib_ABC_DLBCL_UP_PMID29066507
Cell_cycle_Whitfield
Cell_cycle_Liu
duvelisib_ABC_DLBCL_UP_PMID29066507
GC_B_cell_U133Plus
JAK2_downregulated_PMBL
negative_log10_of_adjusted_p_value

Figure 2. Gene Set Enrichment Analysis (GSEA) of SP53 cell line RNA sequencing after treatment with IOA-244 for 24, 48 and 72 hours. X-axis: negative log10 of adjusted p-value. Top 17 down (A) and up (B) gene sets are shown here. Fold change (FC) <0, False discovery rate (FDR) < 0.05

IOA-244 was then combined with 474 compounds (each given at 5μM) in cell lines derived from MCL (SP-53) and from cutaneous T cell lymphoma (HH). Increased anti-proliferative activity was observed for various combinations, including with inhibitors of other kinases, chemotherapy agents as well as with compounds active on metabolism and nuclear targets (Table 1).

drug in combo	cell line	Median CI	95% range
Bendamustine	HH	0.69	0.56-0.85
	SP53	1.3	0.96-1.7
Pixantrone	HH	0.75	0.5-0.84
	SP53	0.77	0.28-1.6
Selinexor	HH	0.77	0.67-0.95
	SP53	1	0.85-1.5
Tretionin	HH	0.36	0.25-0.43
	SP53	0.8	0.53-1.1
Bromopyruvic acid	HH	0.53	0.41-0.84
	SP53	1.3	0.55-1.9

Table 1. Determination of synergy Combination Indexes (CI) for IOA244 and/or of listed molecules. HH and SP53 cell lines were exposed to selected compounds for 72 h. The combination effect was considered as beneficial if synergistic (<0.9) or additive (0-9-1.1), using the Chou-Talalay Combination Index. Values higher than 1.1 are suggestive of antagonism or no benefit.

NHL-FL pts treated with 20mg QD (4/4; 2 female and 2 males) and 80mg QD daily (4/4; 3 female and 1 male) had no DLT (Table 2). Transient platelet reduction (G3) (N=1), AST/ALT elevation (G2) and Neutropenia (G3) (N=1) were observed in 3/8 pts, which returned to baseline without dose modifications (Figure 3).

	20 mg n=4		80 mg n=4		Overall n=8	
	N (%)	Events	N (%)	Events	N (%)	Events
All Causality TEAEs						
Any Grade	4 (100%)	20	4 (100%)	25	8 (100%)	45
Grade 1	4 (100%)	13	4 (100%)	20	8 (100%)	33
Grade 2	1 (25%)	2	3 (75%)	4	4 (50%)	6
Grade 3	1 (25%)	1	1 (25%)	1	2 (25%)	2
Grade 4	0	0	0	0	0	0
Grade 5*	0	0	0	0	0	0
Drug-related TEAEs n (%)						
Any TEAEs related to IOA 244	3 (75%)	9	1 (25%)	1	4 (50%)	10
Grade 1	3 (75%)	5	0	0	3 (38%)	5
Grade 2	1 (25%)	3	0	0	1 (13%)	3
Grade 3ᵻ	1 (25%)	1	1 (25%)	1	2 (25%)	2
Grade 4	0	0	0	0	0	0
Grade 5	0	0	0	0	0	0

Table 2. Summary of Grade Common Terminology Criteria for Adverse Events (CTCAE) toxicities are shown as all-cause and drug-related adverse events.

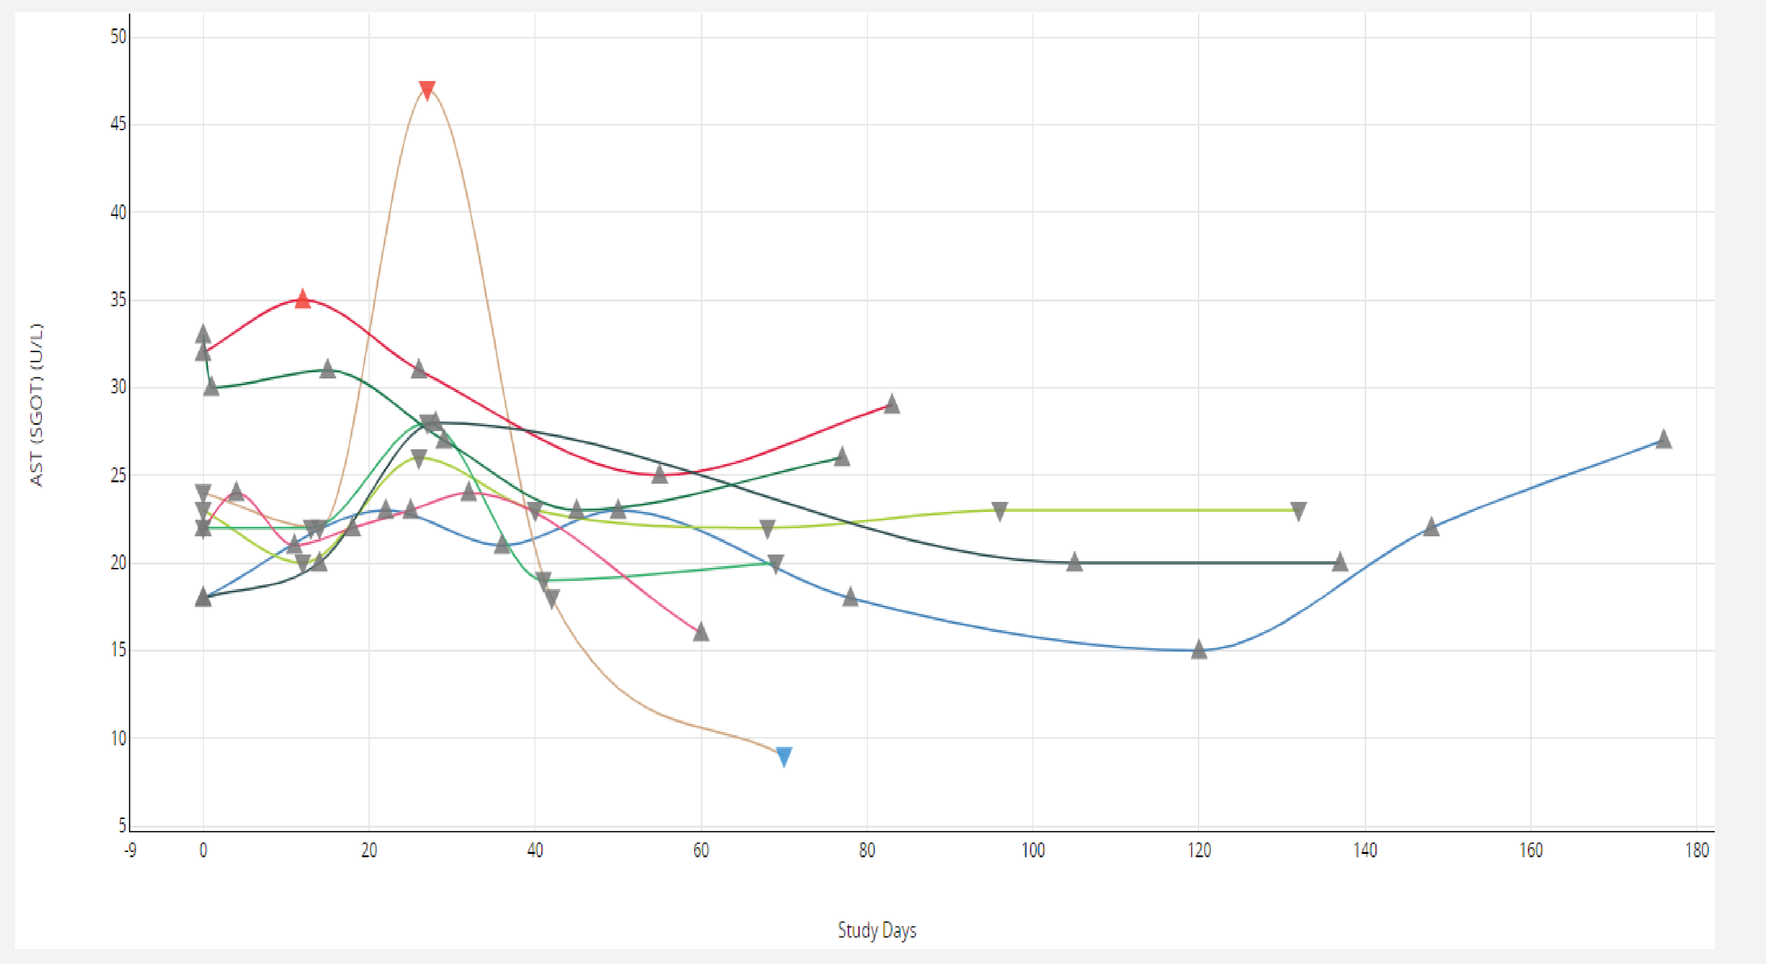
AST (SGOT) (U/L)
Study Days

Figure 3. IOA-244 was given without dose modifications in patients treated for up to 5 months. All 8 patients showed normal AST (U/L) and ALT levels. One patient had a transient increase in AST and ALT, that subsequently returned to baseline while patient continued to receive IOA-244 without any other drug intervention.