3107 - First-in-human phase I study of the highly selective phosphoinositide 3-kinase inhibitor delta (PI3Kδ) inhibitor IOA-244 in patients with advanced cancer: Safety, activity, pharmacokinetic, pharmacodynamic results (Bd # 99)

AM Di Giacomo[1], F Santangelo[1], G Amato[1], E Simonetti[1], J Graham[2], M Lahn[3], L van der Veen[3], Z Johnson[4], C Pickering[5], M Durini[6], T Ziyang[7], T Lakshmikanth[7], P Brodin[7], Mariaelena Occhipinti[8], Matteo Simonelli[9], Carmelo Carlostella[9], Armando Santoro[9], P Spiliopoulou[10 11], TRJ Evans[11]*, Michele Maio[1]*

[1]University of Siena and Center for Immuno-Oncology, Department of Oncology, University Hospital, Siena, Italy, [2]Medical Oncology Dept BWSCC-Beatson West of Scotland Cancer Centre, NHS Greater Glasgow and Clyde, Glasgow, United Kingdom, [3]Oncology Department, iOnctura SA, Geneva, Switzerland, [4]Translational Research & Early Development, iOnctura SA, Geneva, Switzerland, [5]R&D Department, iOnctura SA, Geneva, Switzerland, [6]Oncology Department, Labcorp Clinical Development Ltd, Via Raimondo Montecuccoli 20/1, Milano, Italy, [7]Women´s and Children´s Health, Karolinska Institutet, National Cellular Immunomonitoring Facility, Stockholm, Sweden, [8]Radiomics, Clos Chanmurly 13, Liège, Belgium, [9]IRCSS Humanitas Research Hospital, Via Alessandro Manzoni 56, Milano-Rozzano Italy, [10]Drug Development Program, Phase I Unit, Princess Margaret Hospital, University of Toronto, 700 University Avenue, Toronto (ON), Canada, [11]Institute of Cancer Sciences, University of Glasgow, Glasgow, United Kingdom *Asterisk indicated both are senior authors

BACKGROUND

Figure 1

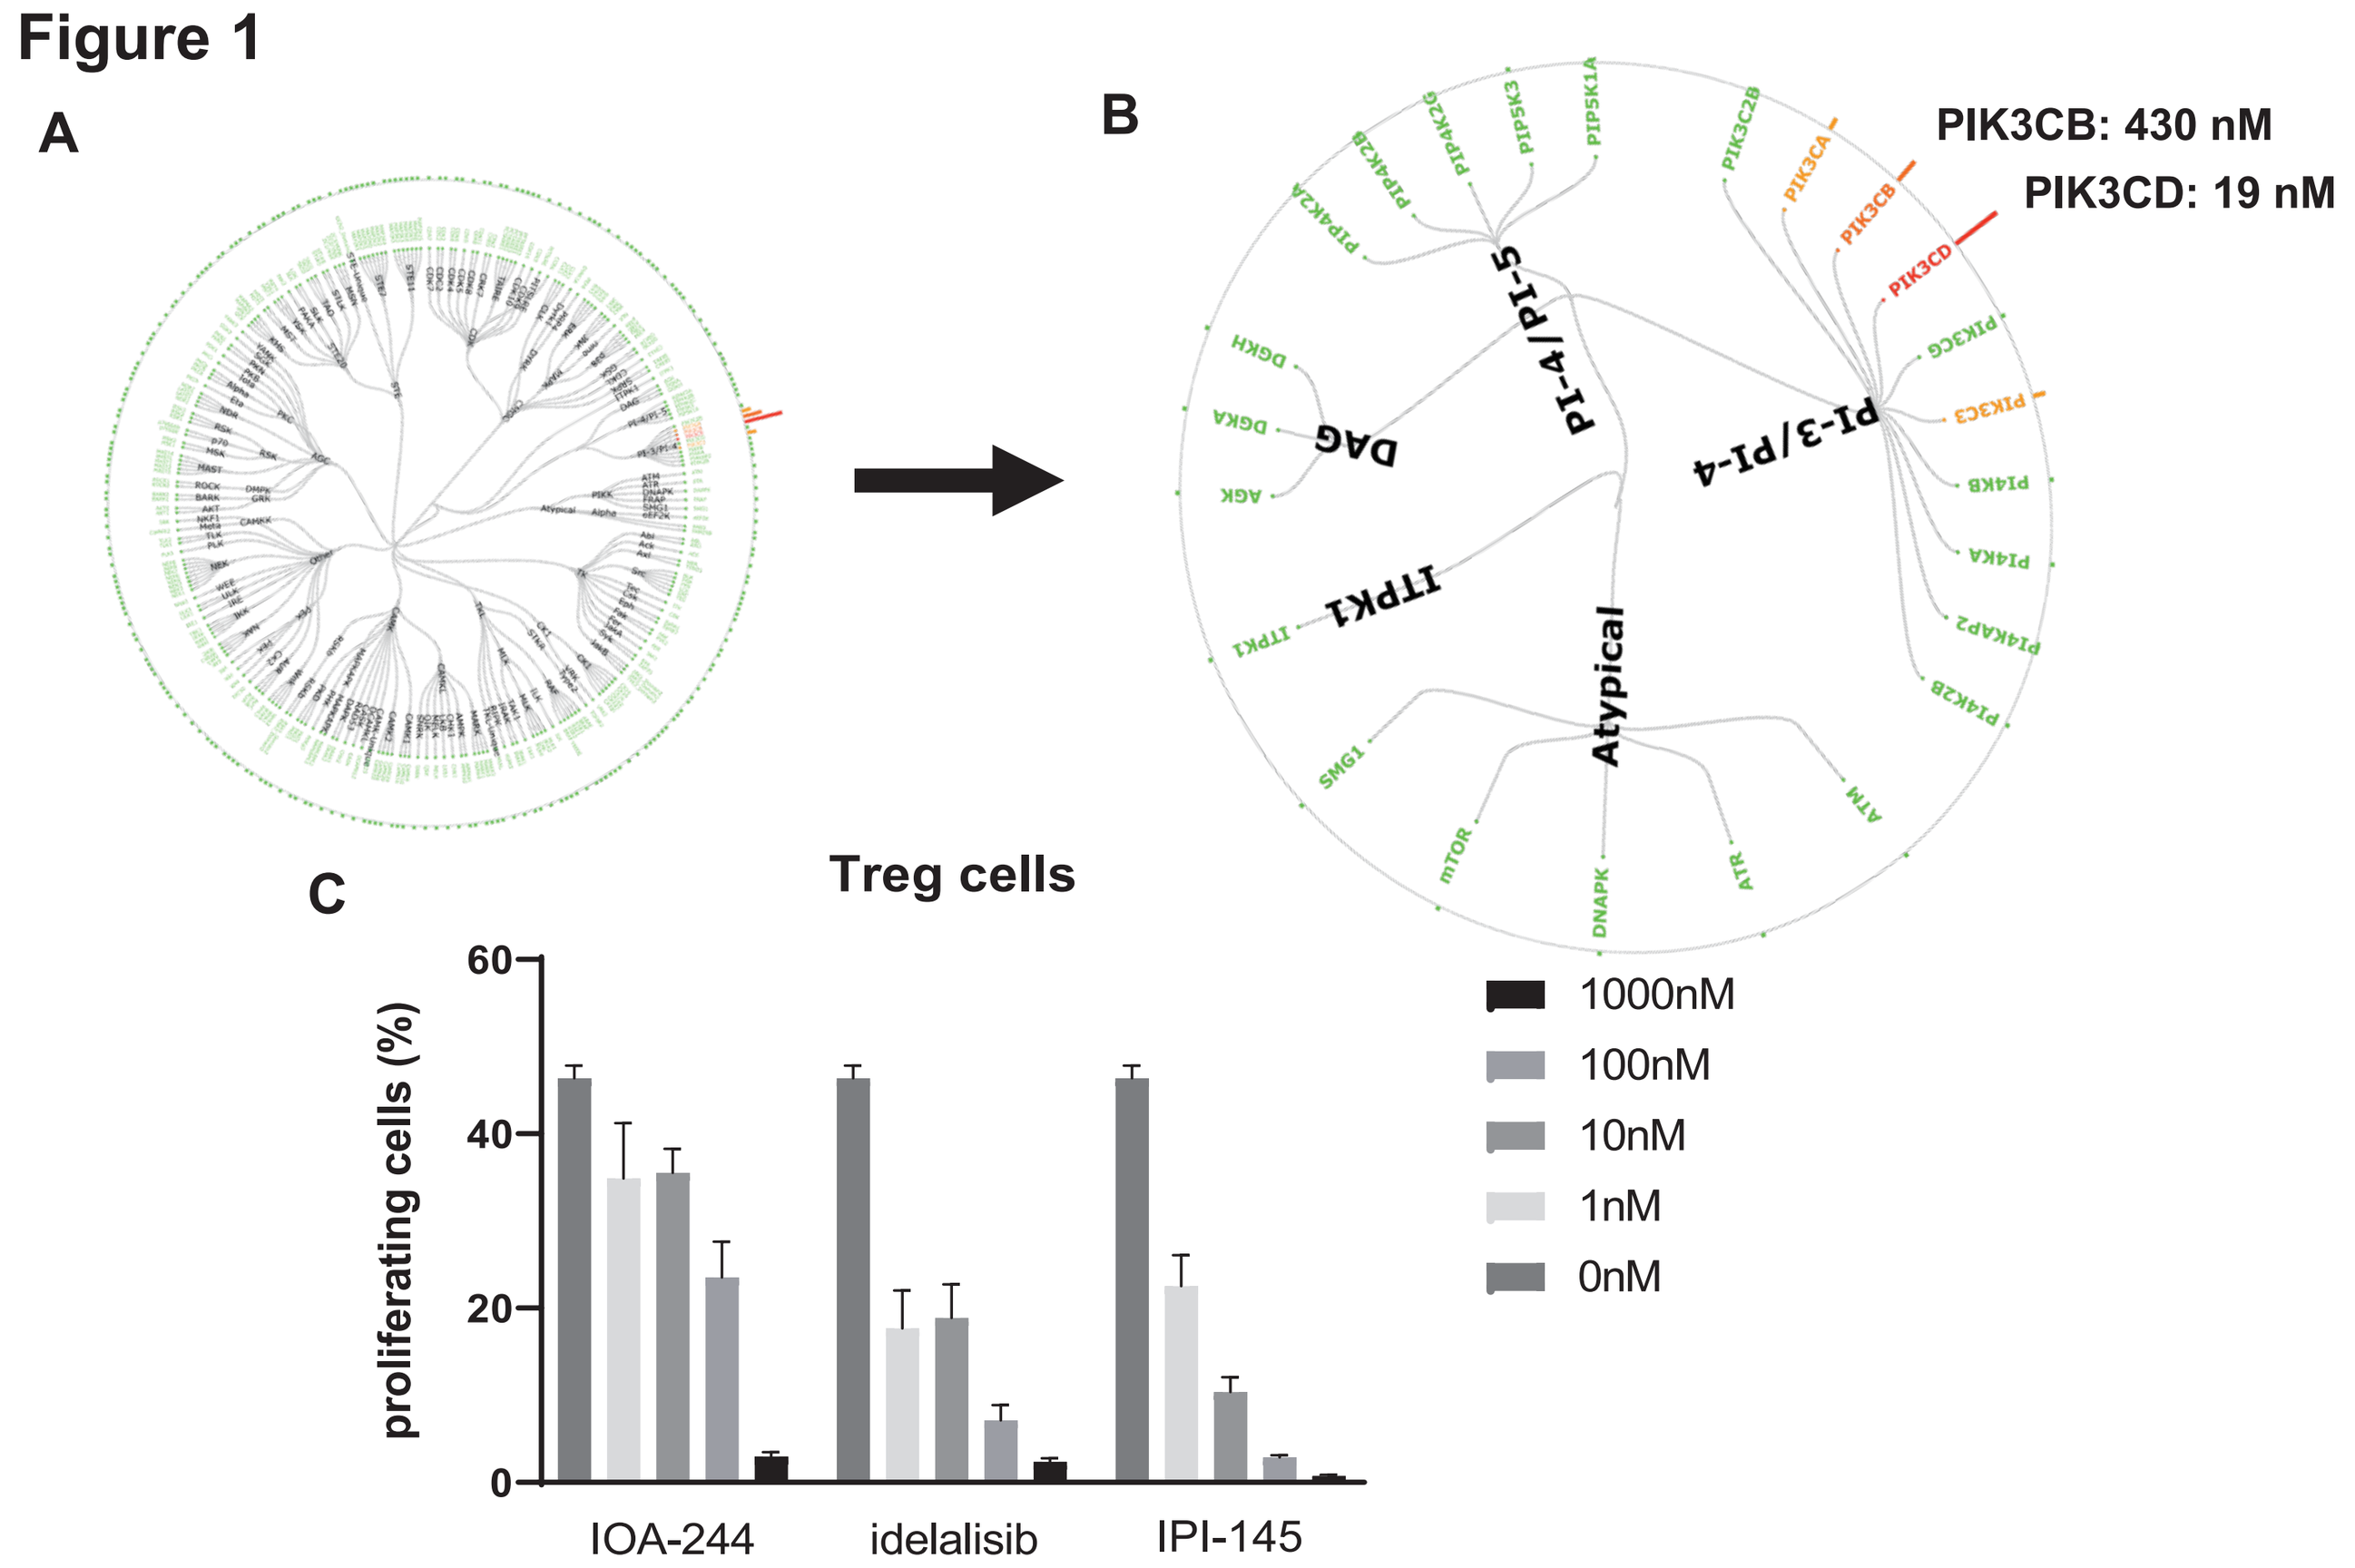
A
B
PIK3CB: 430 nM
PIK3CD: 19 nM
PI-4/PI-5
PI-3/PI-4
DAG
ITPK1
Atypical
C
Treg cells
proliferating cells (%)
1000nM
100nM
10nM
1nM
0nM
IOA-244
idelalisib
IPI-145

Figure 1: IOA-244 is a highly selective PI3Kδ inhibitor (A and B show the selectivity profile in Jurkat lysate using the KiNativ® assay: A: all kinases; B: lipid kinase panel only); C: IOA-244 inhibits the proliferation of human T regulatory (T_{reg}) cells similar to other PI3Kδ inhibitors

OBJECTIVES

Primary:

- Safety and tolerability of escalating doses of IOA-244 to the predicted biological effective dose (BED)

Secondary:

- To assess the pharmacokinetic (PK) profile
- Characterize pharmacodynamic (PD) effect as determined by inhibition of CD63 expression on basophils in response to IOA-244
- To document antitumor activity, including overall response rate (ORR), duration of response (DoR), progression free survival (PFS) and overall survival (OS)

Exploratory:

Changes in immune cell numbers within pre- and post-treatment biopsies and in the circulating blood (multiplex IHC and Cytometry by Time of Flight, CyTOF)

Figure 2

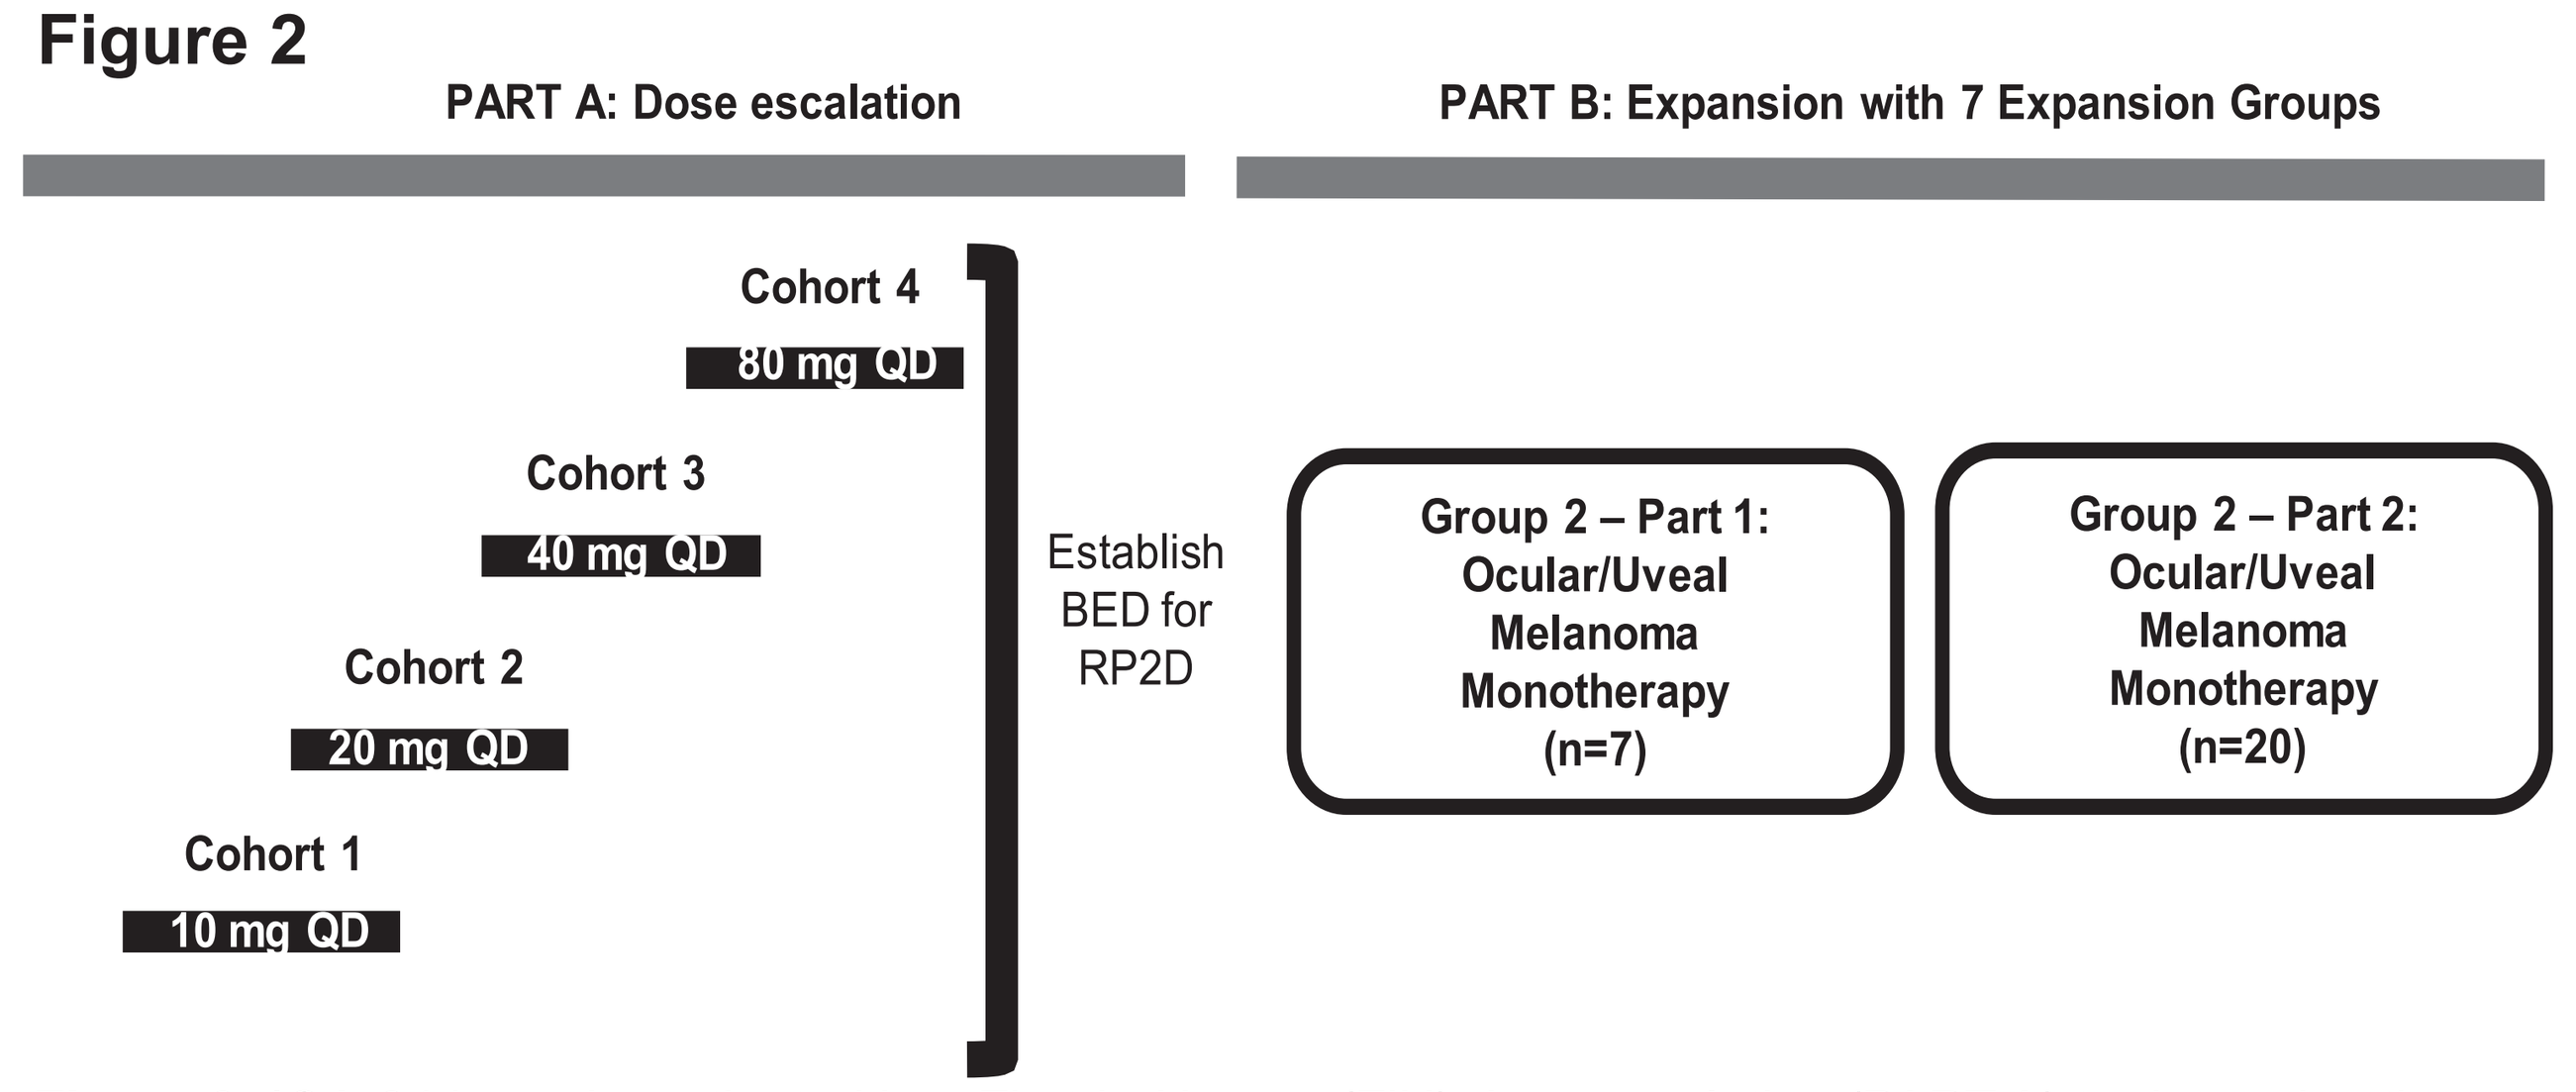
PART A: Dose escalation
PART B: Expansion with 7 Expansion Groups
Cohort 4
80 mg QD
Cohort 3
40 mg QD
Cohort 2
20 mg QD
Cohort 1
10 mg QD
Establish BED for RP2D
Group 2 – Part 1: Ocular/Uveal Melanoma Monotherapy (n=7)
Group 2 – Part 2: Ocular/Uveal Melanoma Monotherapy (n=20)

Figure 2: IOA-244 was investigated in a First-in-Human (FiH) dose escalation (PART A) to determine the BED. The BED of 80 mg QD was used for the Recommended Phase 2 Dose (RP2D). Once the RP2D was determined, Part B was initiated, consisting of 7 expansion cohorts. The first expansion group was established to investigate the single-agent IOA-244 administration in patients with uveal melanoma, first in 7 patients, followed by an additional 20 patients should a response be observed among the 7 patients.

METHODS

Design:

- 3+3 cohort dose escalation
- By cohort reassessment of PK and PD to inform and confirm the predictive non-clinical PK/PD model
- BED defined as the concentration of IOA-244 at which CD63 is inhibited ≥50% area under the effect (AUE) per 24 hours

Patients Eligibility

- ≥18 years of age with the following:
- A performance status of ≤2 on the Eastern Cooperative Oncology Group (ECOG) scale
- Histological or cytological evidence of a diagnosis of cancer that is advanced and/or metastatic disease for mesothelioma, cutaneous, and uveal melanoma
- Adequate organ functioning

Assessments:

- Toxicities graded according to Common Terminology Criteria for Adverse Events (CTCAE) version 5.0
- Standard laboratory hematology and chemistry
- RECIST 1.1. based evaluation (ORR)
- Benefit/Risk for Recommended Phase 2 Dose (RP2D)

RESULTS

Demography and Baseline Characteristics

Solid Tumor	10 mg	20 mg	40 mg	80 mg	Overall
N	4	4	4	11	23
Age (median)	73.5	52.0	70	56.5	56.5
Sex (m/f)	4/0	1/3	2/2	3/8	10/13
Primary Diagnosis					
Uveal Melanoma	1	2	3	10	16
Mesothelioma	1	1	0	0	2
Melanoma	2	1	1	1	5
Median prior Tx	2 (1-3)	4 (1–6)	2 (1-3)	2 (0-2)	2 (0-6)

Safety

All causality TEAEs by System Organ Class	10 mg (n)	20 mg (n)	40 mg (n)	80 mg (n)	Overall (n)
Evaluable Patients	4	4	4	11	23
Any Grade TEAE (events)	25	26	12	109	172
Any Serious TEAE	2	0	1	1	4
Disease Progression					
Grade 3/4	0	1	0	0	1
Diarrhoea					
Grade 3/4	0	0	0	1	1
Abdominal pain upper					
Grade 3/4	0	0	0	1	1
Investigations					
Grade 3/4	0	0	0	3	3
ALT increase					
Grade 3/4ᵻ	0	0	0	2	2
AST increase					
Grade 3/4ᵻ	0	0	0	2	2
Respiratory					
Grade 5*	1	0	1	0	2
Dyspnoea					
Grade 5*	1	0	0	0	1
Pulmonary embolism					
Grade 5*	0	0	1	0	1

ᵻ ALT/AST increase transient and considered related to liver metastases. Transient and improved on continuous treatment (no dose interruption)
*The CTCAE Grade 5 toxicities observed were associated with tumour progression in patients with mesothelioma and not considered related to treatment

Considered Drug-related Treatment Emergent Adverse Events (TEAEs) Organ System/Preferred Term (MedDRA)	10 mg	20 mg	40 mg	80 mg	Overall
Patients evaluable / dosed	2/4	1/4	2/4	2/11	5/23
GRADE 1 (MILD)					
Any drug-related TEAE (# events)	3	1	5	2	11
Nervous System Disorders	1	0	2	0	4
Dysgeusia	1	0	0	0	1
Headache	0	0	1	0	1
Lethargy	0	0	1	0	1
Parasthesia	0	0	0	1	1
Gastrointestinal Disorders	0	0	2	1	3
Diarrhoea	0	0	2	0	2
Vomiting	0	0	0	1	1
General Disorders	0	1	0	0	1
Asthenia	0	1	0	0	1
Metabolism & Nutrition Disorders	1	0	0	0	1
Decreased Appetite	1	0	0	0	1
Musculoskeletal & Connective Tissue Disorders	0	0	0	1	1
Arthralgia	0	0	0	1	1
Skin & Subcutaneous Tissue Disorders	1	0	0	0	1
Dry skin	1	0	0	0	1
GRADE 2 (MODERATE)					
Any drug-related TEAE (# events)	0	0	0	2	2
Gastrointestinal Disorders	0	0	0	1	1
Diarrhoea	0	0	0	1	1
Eye Disorders	0	0	1	0	1
Uveitis	0	0	1	0	1
GRADE 3/4/5 (SEVERE) Any drug-related TEAE (# events)	0	0	0	0	0

In vivo metabolite profile shows no significant risks

Metabolite	m/z	Cohort 1	Cohort 2	Cohort 3	Cohort 4
IOA-244 (parent)	**527**	**94.03**	**94.83**	**93.70**	**93.18**
IOA-244 +O	543	1.18	0.77	0.71	1.05
IOA-244 -C_2H_2	501	1.20	0.40	0.50	0.34
IOA-244 +O+H_2	545	0.52	0.23	0.25	0.25
IOA-244 +O -H_2	541	2.60	0.78	0.57	1.06
IOA-244 +O	543	ND	0.88	0.60	1.42
IOA-244 +O+H_2	545	0.32	0.12	0.30	0.43
IOA-244 -H_2	525	1.38	2.01	3.36	2.26

Figure 3: Plasma protein binding

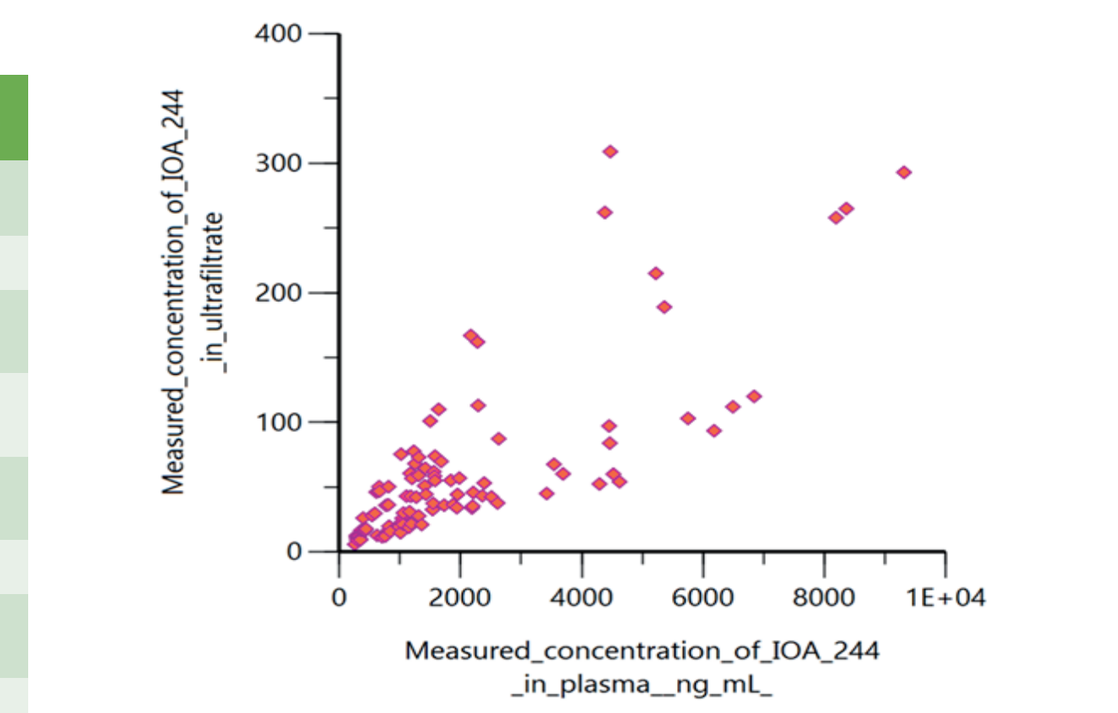
Measured_concentration_of_IOA_244 _in_ultrafiltrate
Measured_concentration_of_IOA_244 _in_plasma__ng_mL_

Figure 3: Free IOA-244 is present in patient approximately 4% of total IOA-244

Figure 4

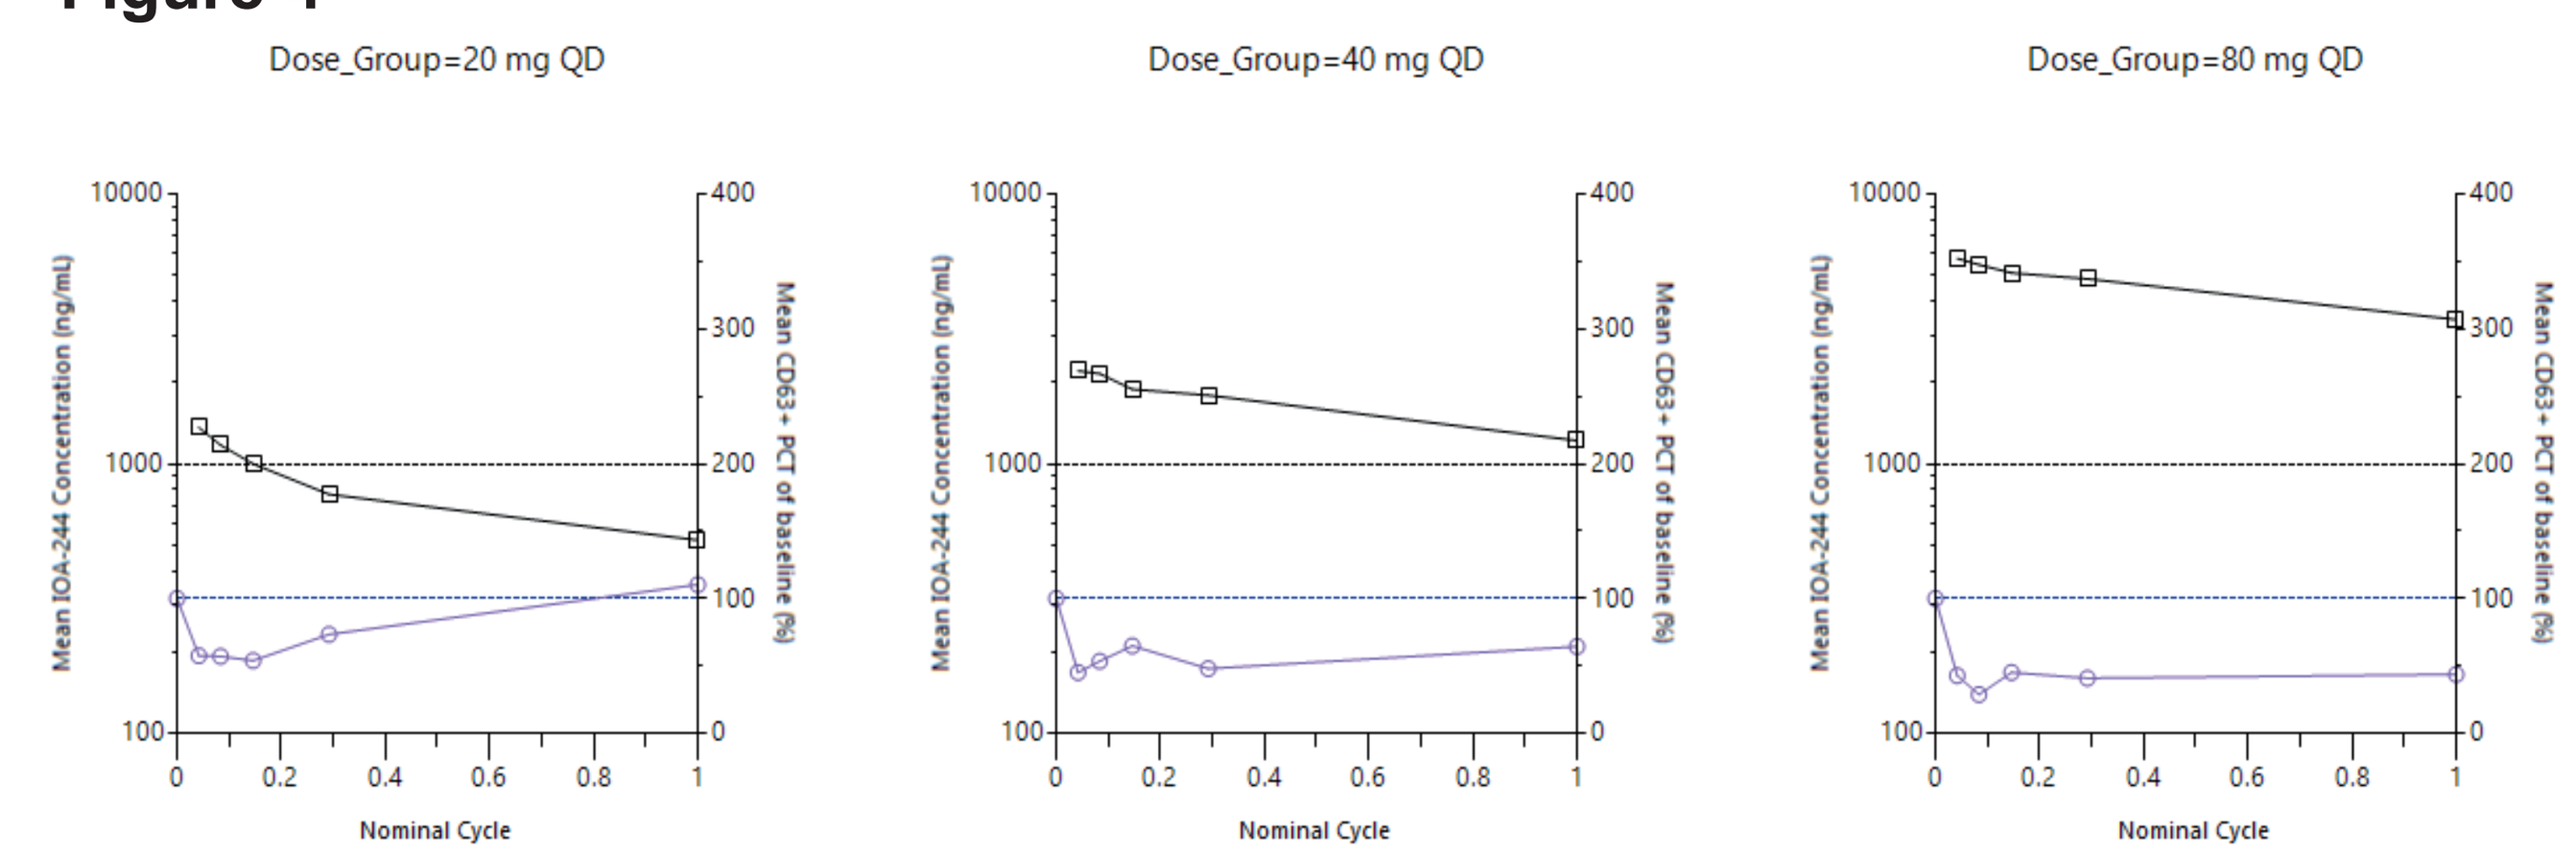
Dose_Group=20 mg QD
Dose_Group=40 mg QD
Dose_Group=80 mg QD
Mean IOA-244 Concentration (ng/mL)
Mean CD63+ PCT of baseline (%)
Nominal Cycle

Figure 4: At Cycle1 Day1/Day2: Above 40 mg QD, IOA-244 reaches maximum inhibition of CD63 expression (Basophil Activation Test; values are mean for 4 pts)

Figure 5

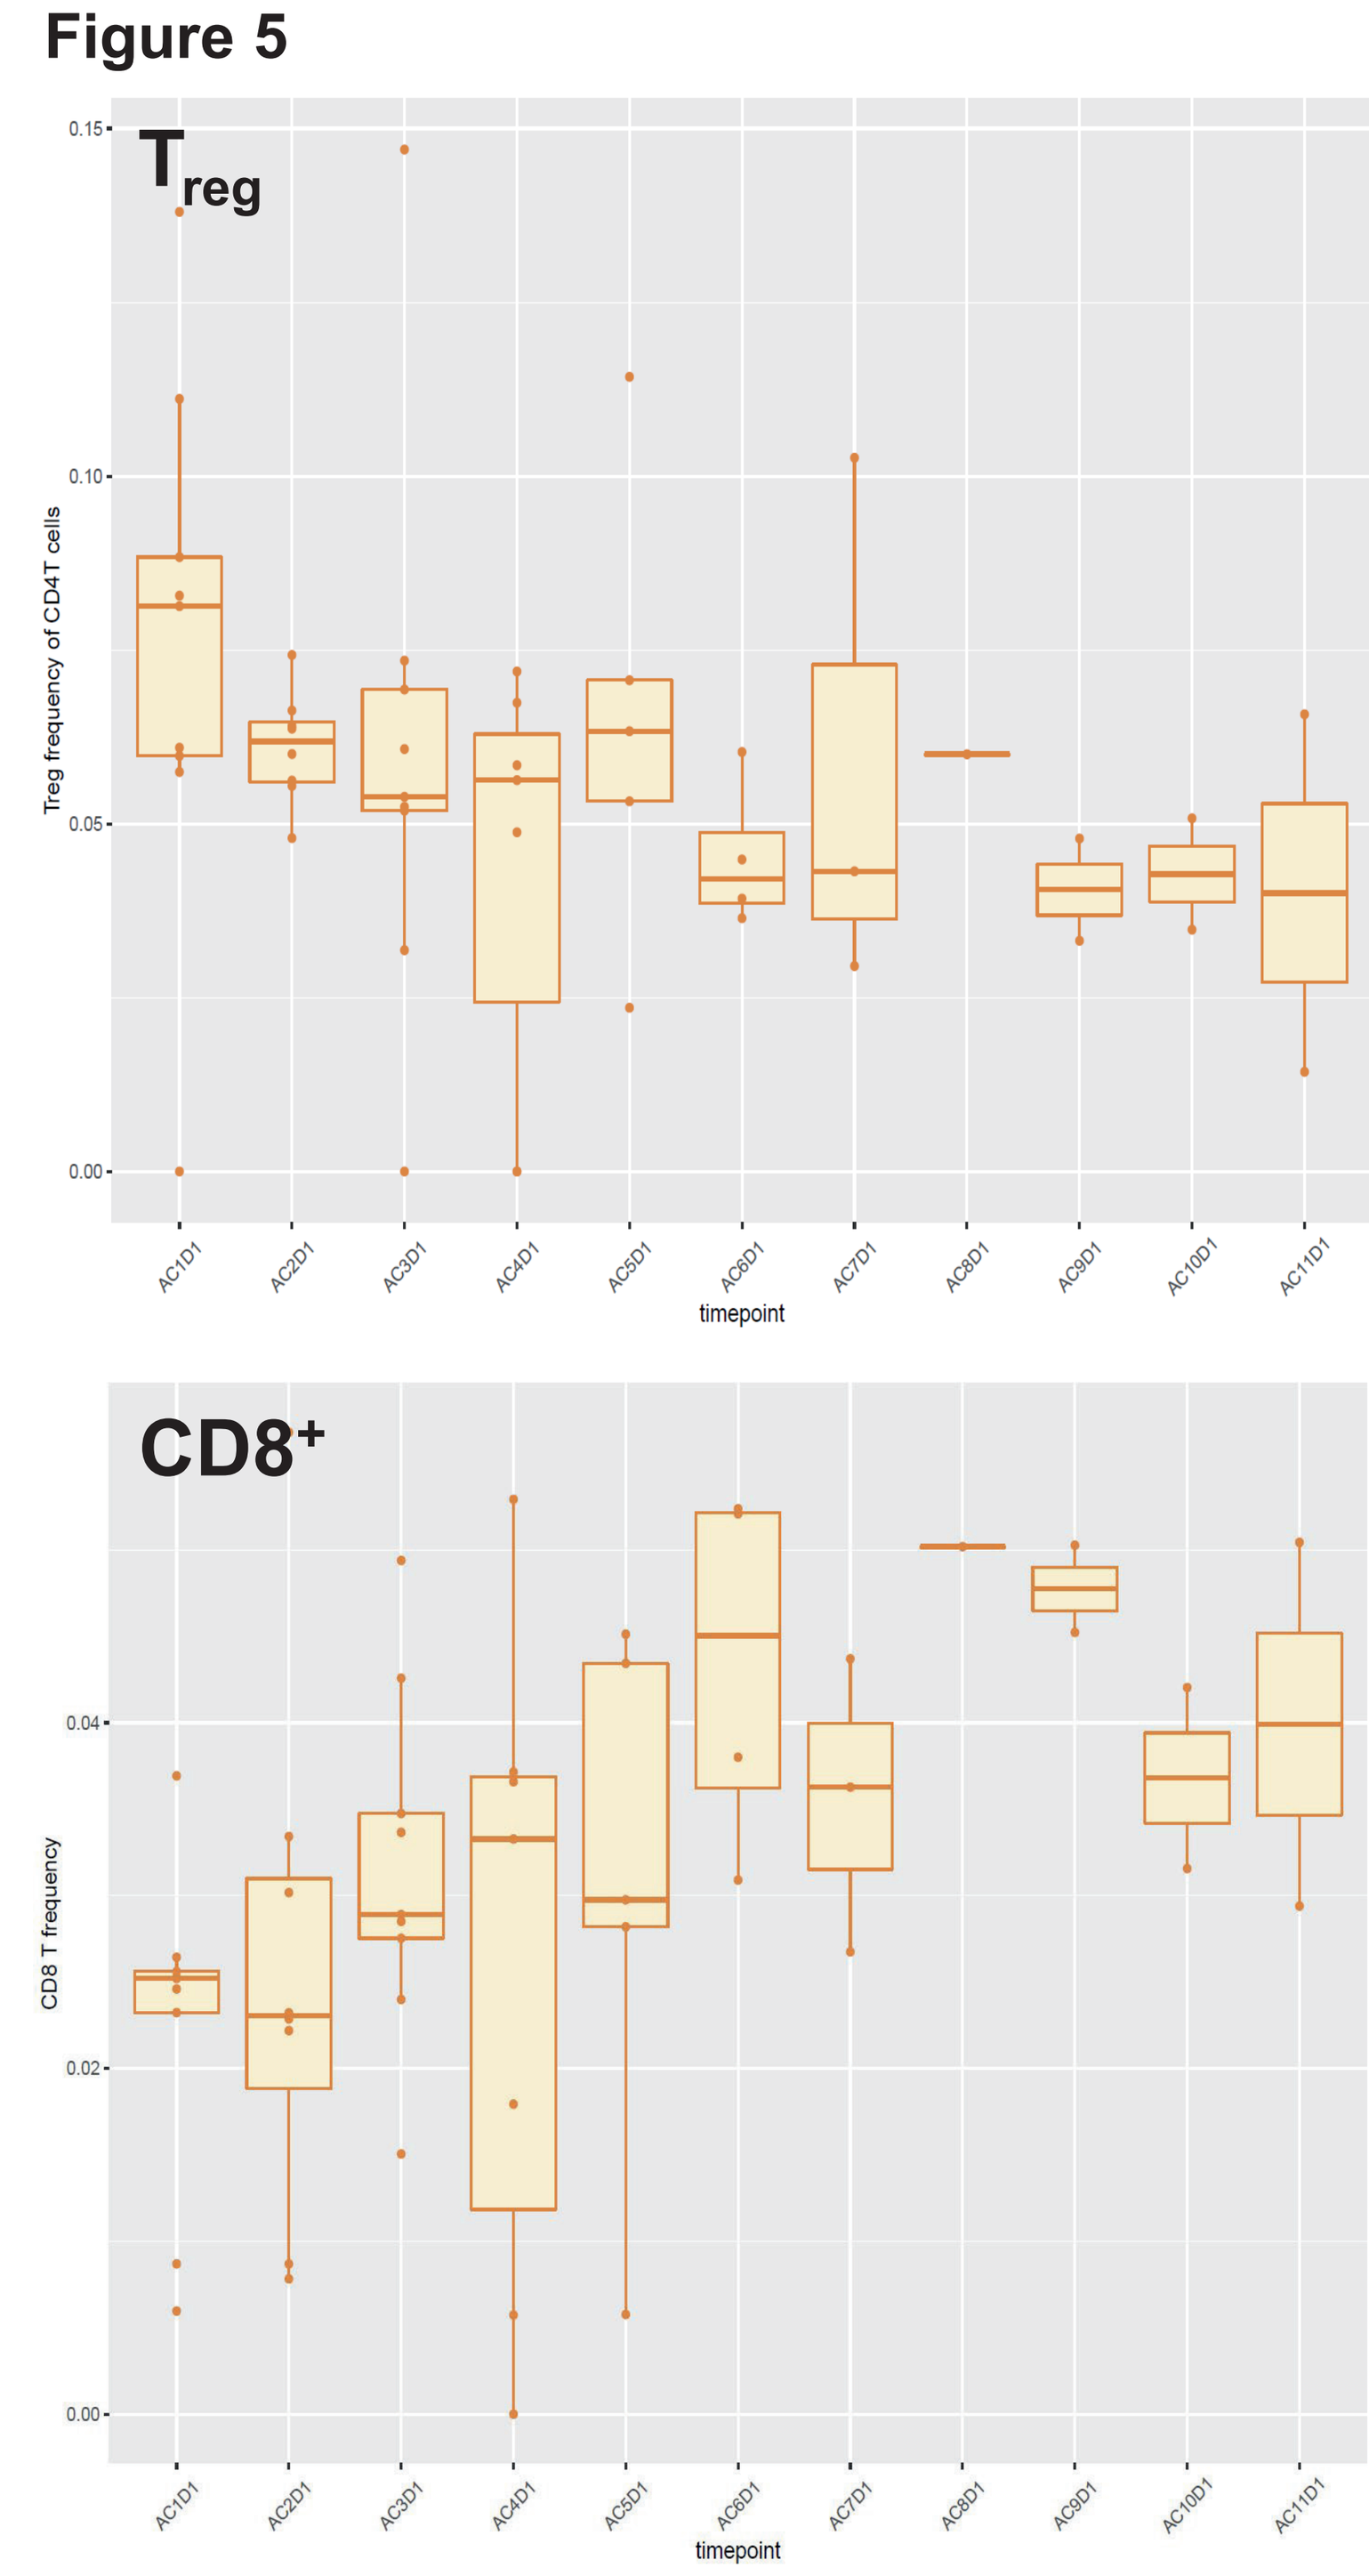
Treg
Treg frequency of CD4T cells
CD8+
CD8 T frequency
timepoint
AC1D1
AC2D1
AC3D1
AC4D1
AC5D1
AC6D1
AC7D1
AC8D1
AC9D1
AC10D1
AC11D1

Figure 5: Evaluation of T cell subsets in the peripheral blood using CyTOF (X-axis shows the blood draws at each monthly visit and Y-axis indicates the frequency of gated events for T_{reg} and CD8⁺ T cells for each evaluable patient)

Figure 6

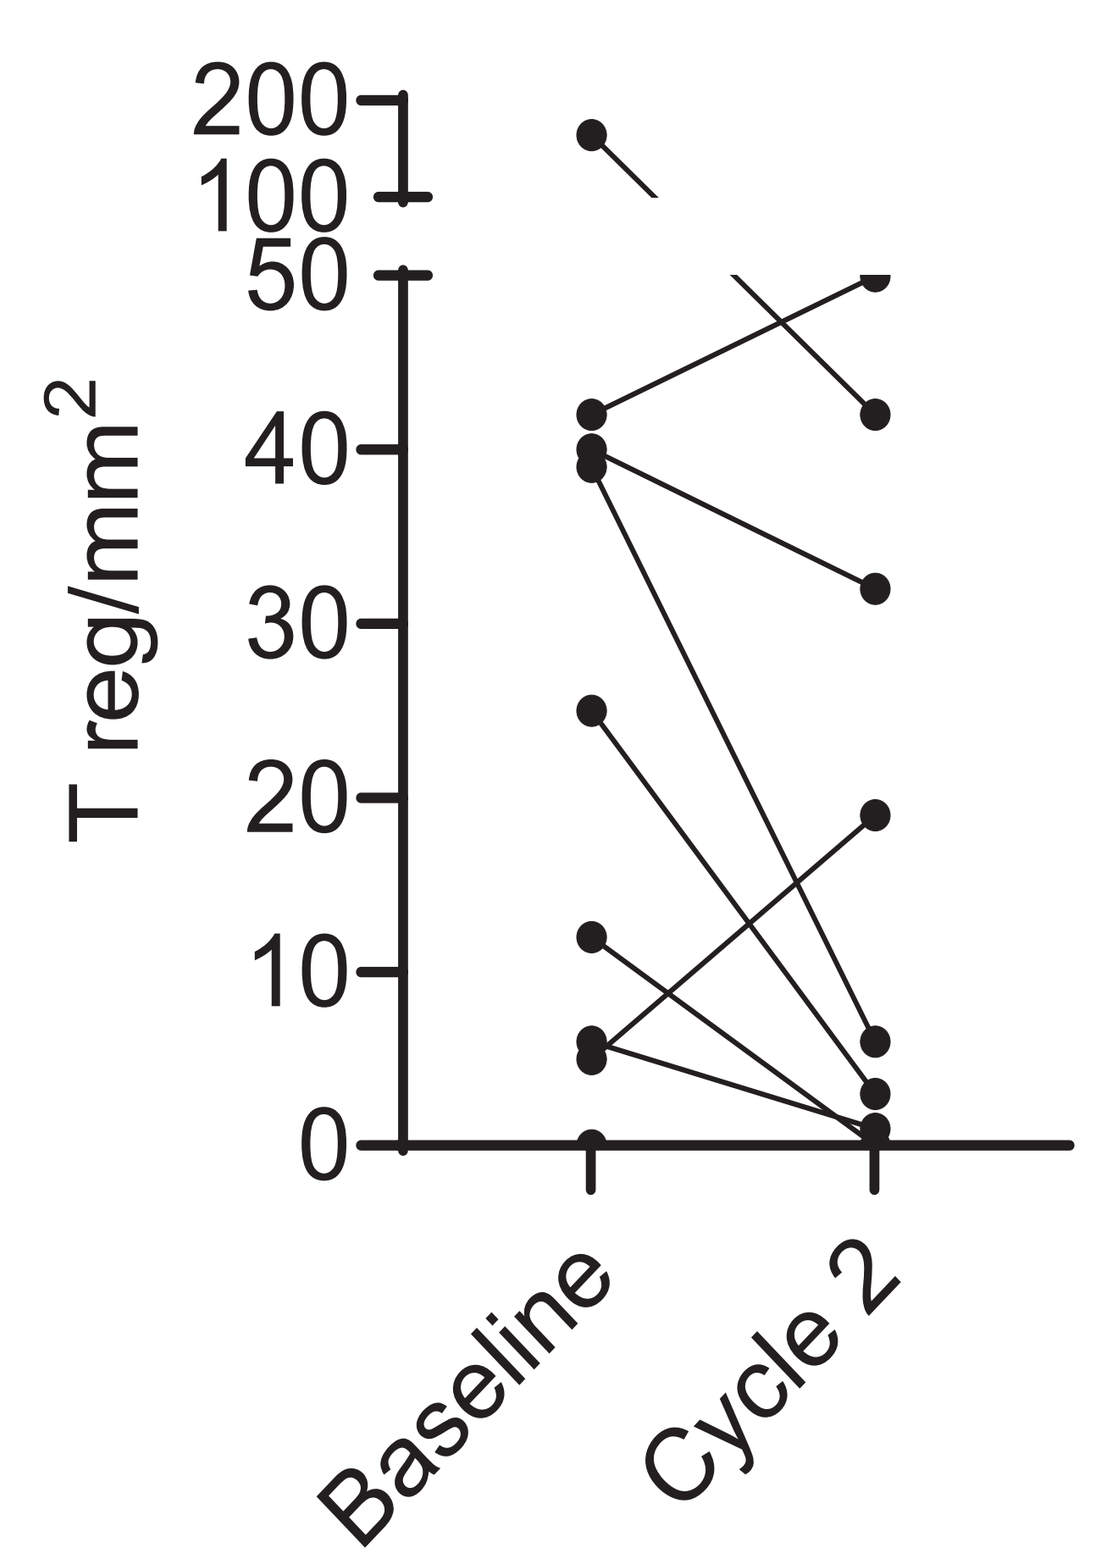
T reg/mm2
Baseline
Cycle 2

Figure 6: Multiplex IHC assessing T_{reg} cells in pre- and post-treatment biopsies of evaluable patients with cutaneous and uveal melanoma (n=9). Paired biopsies obtained at Week 5 post-treatment. 9 of 16 patients were evaluable for this exploratory examination. T_{reg} were reduced in 6/9 pts (67%).

Anti-tumor Activity

Figure 7

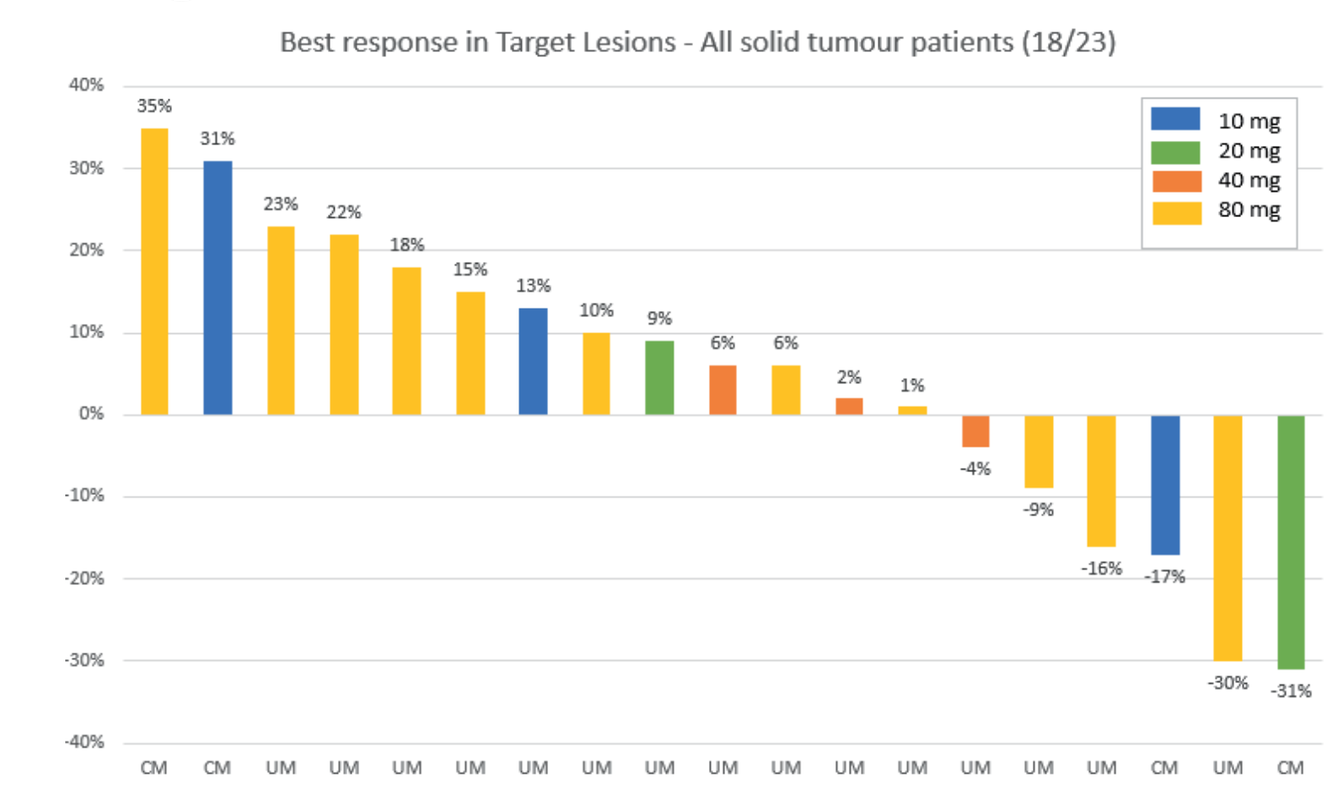
Best response in Target Lesions - All solid tumour patients (18/23)
10 mg
20 mg
40 mg
80 mg

Figure 7: Waterfall plot of best RECIST 1.1 measured response. PR 2/18 (11%) in RECIST 1.1. evaluable cutaneous and uveal melanoma

Figure 8

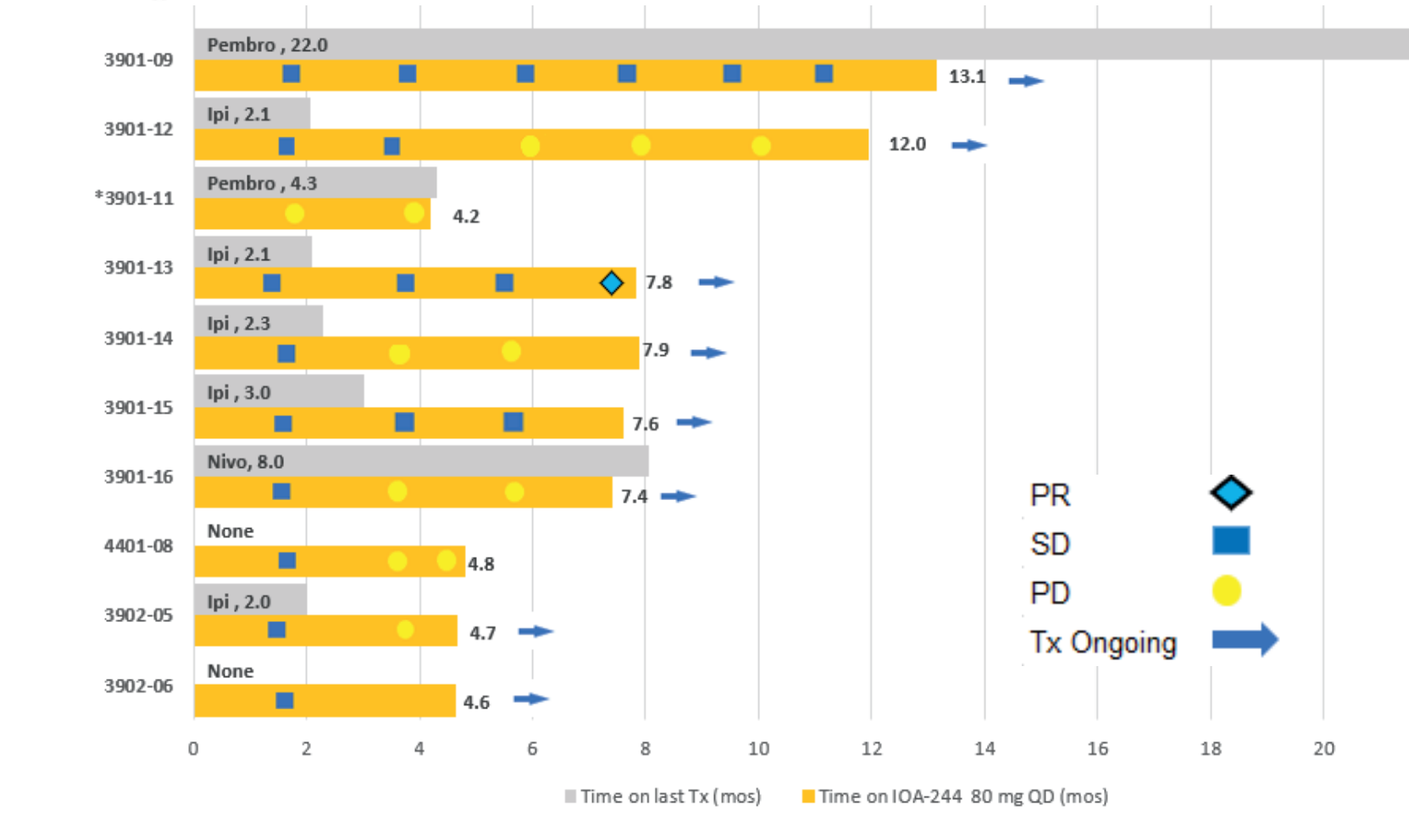
PR
SD
PD
Tx Ongoing
Time on last Tx (mos)
Time on IOA-244 80 mg QD (mos)

Figure 8: Swimmer Plot for patients with uveal melanoma at IOA-244 at 80 mg QD (n=10) (X-axis: months; Y-axis: patient IDs). Patients with the last prior therapy in grey. Numbers at of each bar is month of last assessment. 4/10 pts (40%) are on IOA-244 after RECIST 1.1-based PD (Treatment Beyond Progression).

Figure 9

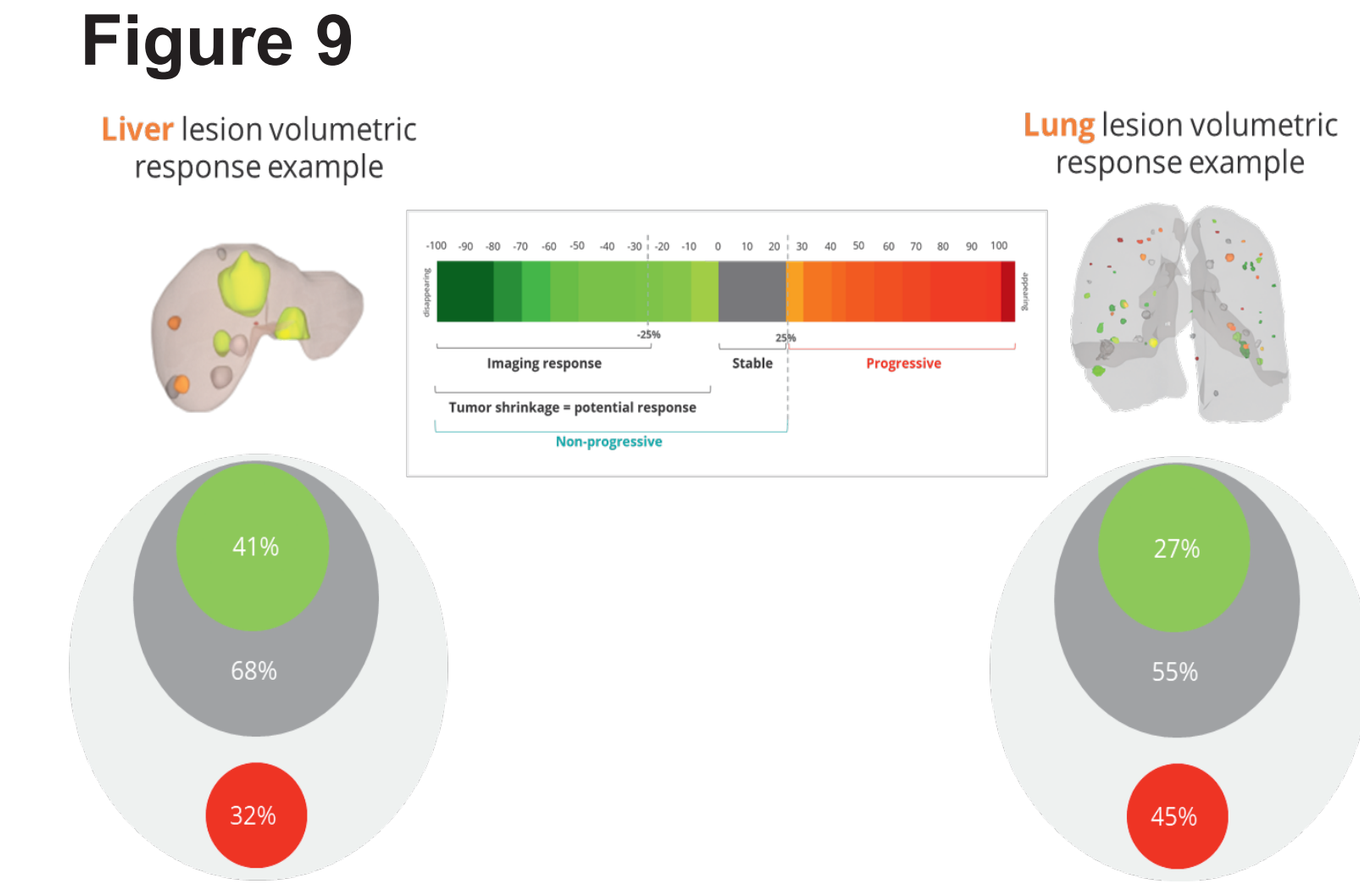
Liver lesion volumetric response example
Lung lesion volumetric response example
Imaging response
Stable
Progressive
Tumor shrinkage = potential response
Non-progressive

Figure 9: Per lesion volumetric radiomics analysis of subset of 7 Uveal Melanoma patients comparing CT scans acquired at baseline (T0) and first follow up (T1). Representative image for Liver and Lung evaluated in this assessment. Color scale indicate imaging responses

Shape features at baseline	$\frac{Volume\ at\ T1}{Volume\ at\ T0}$		$Volume\ at\ T1 - Volume\ at\ T0$	
Organ	Lung	Liver	Lung	Liver
Surface	0.088	0.000	0.058	**0.625**
MinorAxisLength	0.087	0.004	0.131	**0.612**
Compactness	0.093	0.001	0.057	**0.513**
MajorAxisLength	0.129	0.000	0.193	**0.428**
MaxDiameter_3_D	0.116	0.001	0.156	**0.426**

Mixed-effect correlation between volume evolution and lesions shapes features: moderate correlations (0.4-0.7) between volume variation and shape features at baseline for the lesions in the liver

More details on the radiomics analysis can be found in poster 3068: LESION SPECIFIC RADIOMICS ANALYSIS SHOWS PROMISING RESULTS FOR EARLY-STAGE EFFICACY ASSESSMENT OF IOA-244 IN UVEAL MELANOMA

Figure 10

Treatment Duration in Uveal Melanoma Patients (n=10) at 80 mg QD:

Uveal Melanoma 80 mg QD	Mean (median) treatment duration (as of 13th May 2022) (months)	Range of treatment (months)
Part A/B n=3/7	**7.4 (7.5)** [Part A: 9.8 (12.0)] [Part B: 6.4 (7.4)]	**4.2 – 13.1** [Part A: 4.2 – 13.1] [Part B: 4.6 – 7.9]

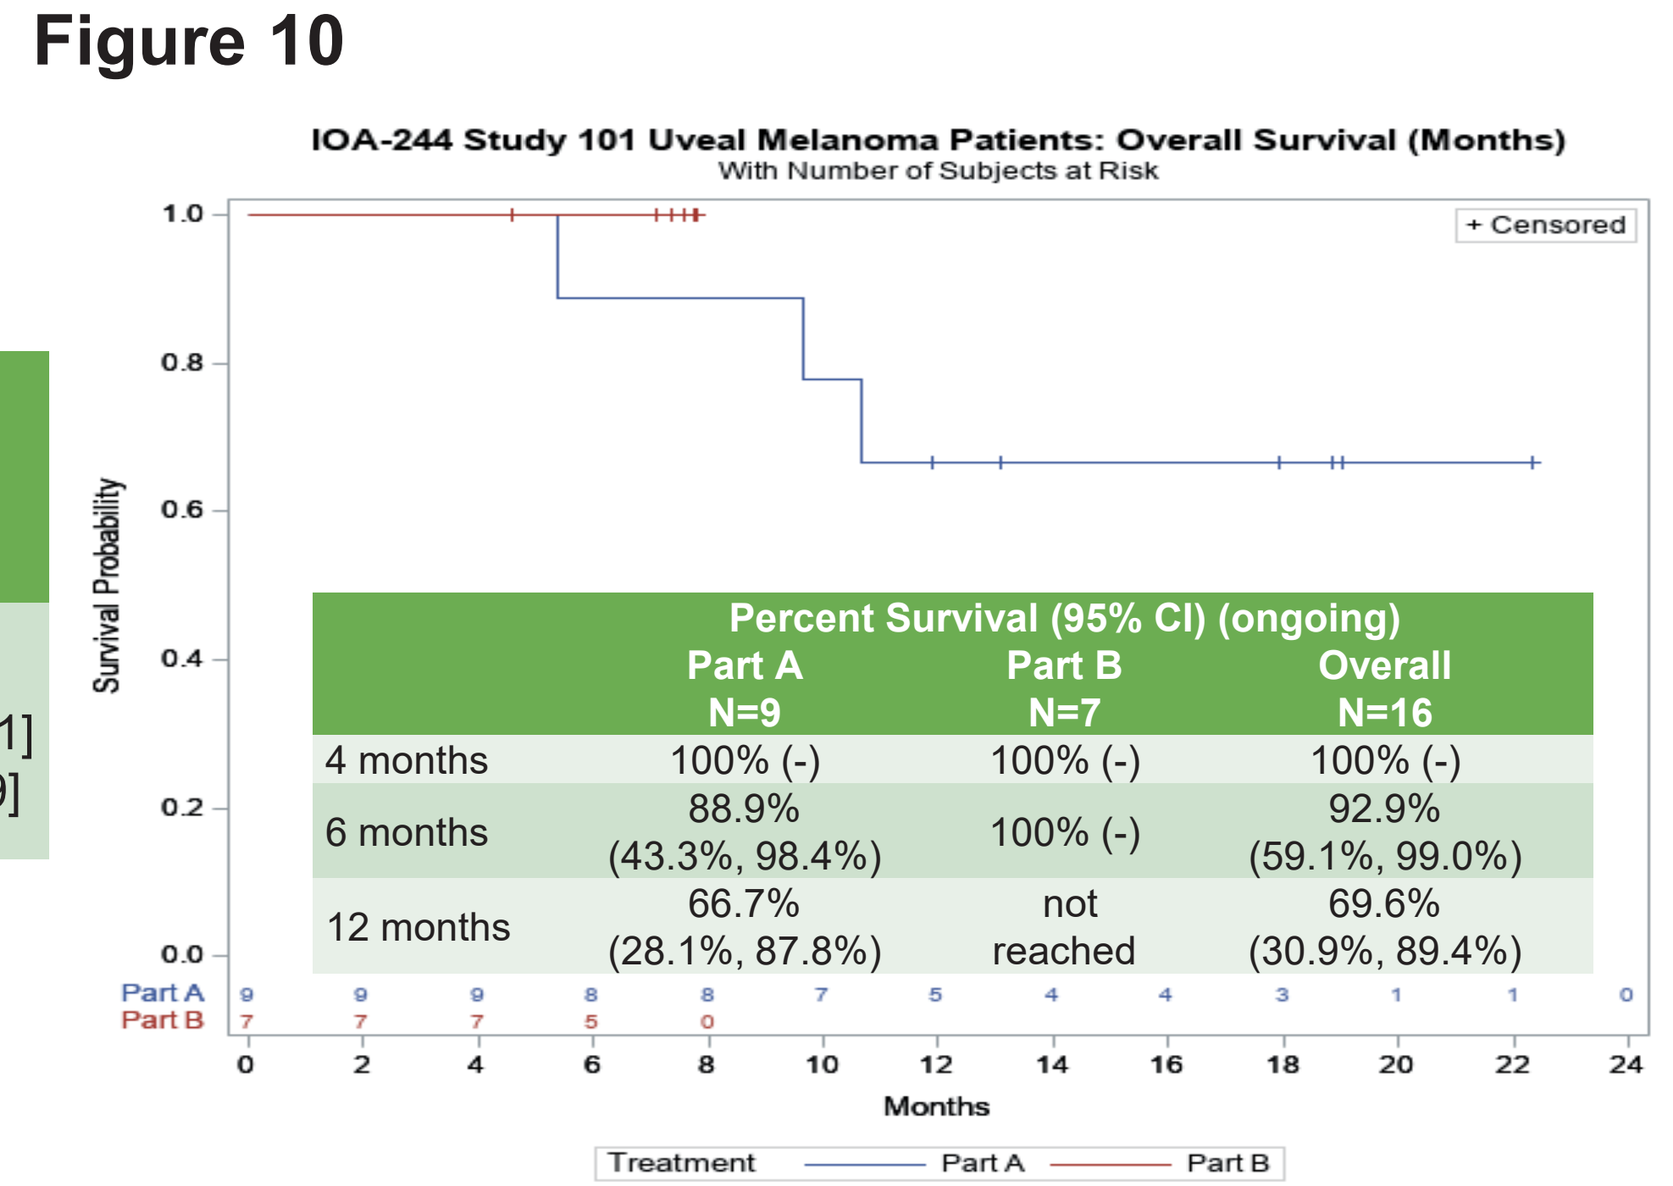
IOA-244 Study 101 Uveal Melanoma Patients: Overall Survival (Months)
With Number of Subjects at Risk
+ Censored
Survival Probability
Months
Treatment
Part A
Part B

	Percent Survival (95% CI) (ongoing) Part A N=9	Part B N=7	Overall N=16
4 months	100% (-)	100% (-)	100% (-)
6 months	88.9% (43.3%, 98.4%)	100% (-)	92.9% (59.1%, 99.0%)
12 months	66.7% (28.1%, 87.8%)	not reached	69.6% (30.9%, 89.4%)

Figure 10: Overall survival in metastatic uveal melanoma in Part A (n=9) and subsequent Part B (n=7)

CONCLUSIONS

- Favorable safety, Absorption, Distribution, Metabolism, Excretion and PK profile of IOA-244 allows long-term administration without dose-reduction or omissions
- T_{regs} are reduced by IOA-244 in peripheral blood and tumor tissue
- Clinical benefit as determined by overall survival is still evolving in >2nd line uveal melanoma

Acknowledgment

The authors thank the patients and their families, all study personal at the clinical investigation centers, the LabCorp Drug Development team, Indivumed team and the Veramed Statistical Consultancy team

ASCO | Annual Meeting - Jun 5-6 2022 | Developmental Therapeutics-Molecularly Targeted Agents and Tumor Biology

Presenter´s email: a.digiacomo@ao-siena.toscana.it

References
Haselmayer et al. 2014 May 22;5:233. doi: 10.3389/fimmu.2014.00233. eCollection 2014
Ali, K., et al., Essential role for the p110delta phosphoinositide 3-kinase in the allergic response. Nature, 2004. 431(7011): p. 1007-11
Di Giacomo et al. Annals of Oncology (2021) 32 (suppl_7): S1428-S1457. 10.1016/annonc/annonc787

NCT04328844 - Sponsored by iOnctura SA
Study Ongoing and Data Not Fully Validated