IOA-289 - a Novel, Clinical Stage Autotaxin Inhibitor for the Treatment of Fibrotic Lung Disease

Marcel A. Deken[1], Karolina Niewola-Staszkowska[1], Michael M. Lahn[1] , Anne Cheasty[2], Pritom Shah[2], Stuart Farrow[2] , Lars van der Veen & Zoë Johnson[1]

1. iOnctura SA, Avenue de Sécheron 15, 1202 Geneva, Switzerland; 2. Cancer Research UK Therapeutic Discovery Laboratories, Babraham Research Campus, Cambridge, CB22 3AT, UK

Background

Autotaxin (ATX) is a secreted glycoprotein with lysophospholipase D activity that hydrolyzes lysophosphatidylcholine (LPC) to lysophosphatidic acid (LPA). LPA signaling through LPA receptors is implicated in controlling cellular activities including migration, proliferation and survival in fibrotic pathologies, including idiopathic pulmonary fibrosis (IPF) and cancer (Figure 1). Several inhibitors of the ATX/LPA pathway have been developed for treatment of IPF.
IOA-289 is a novel, highly potent autotaxin inhibitor with a unique chemical structure that is in clinical development for the treatment of fibrotic pathologies including cancer. IOA-289 does not bind to the catalytic zinc region of ATX but binds to both the substrate pocket as well as to the LPA carrier channel thereby blocking both functions of ATX. This results in a more favorable safety profile compared to first generation inhibitors. A second ATX inhibitor from a different chemical series is in preclinical development.

Figure 1

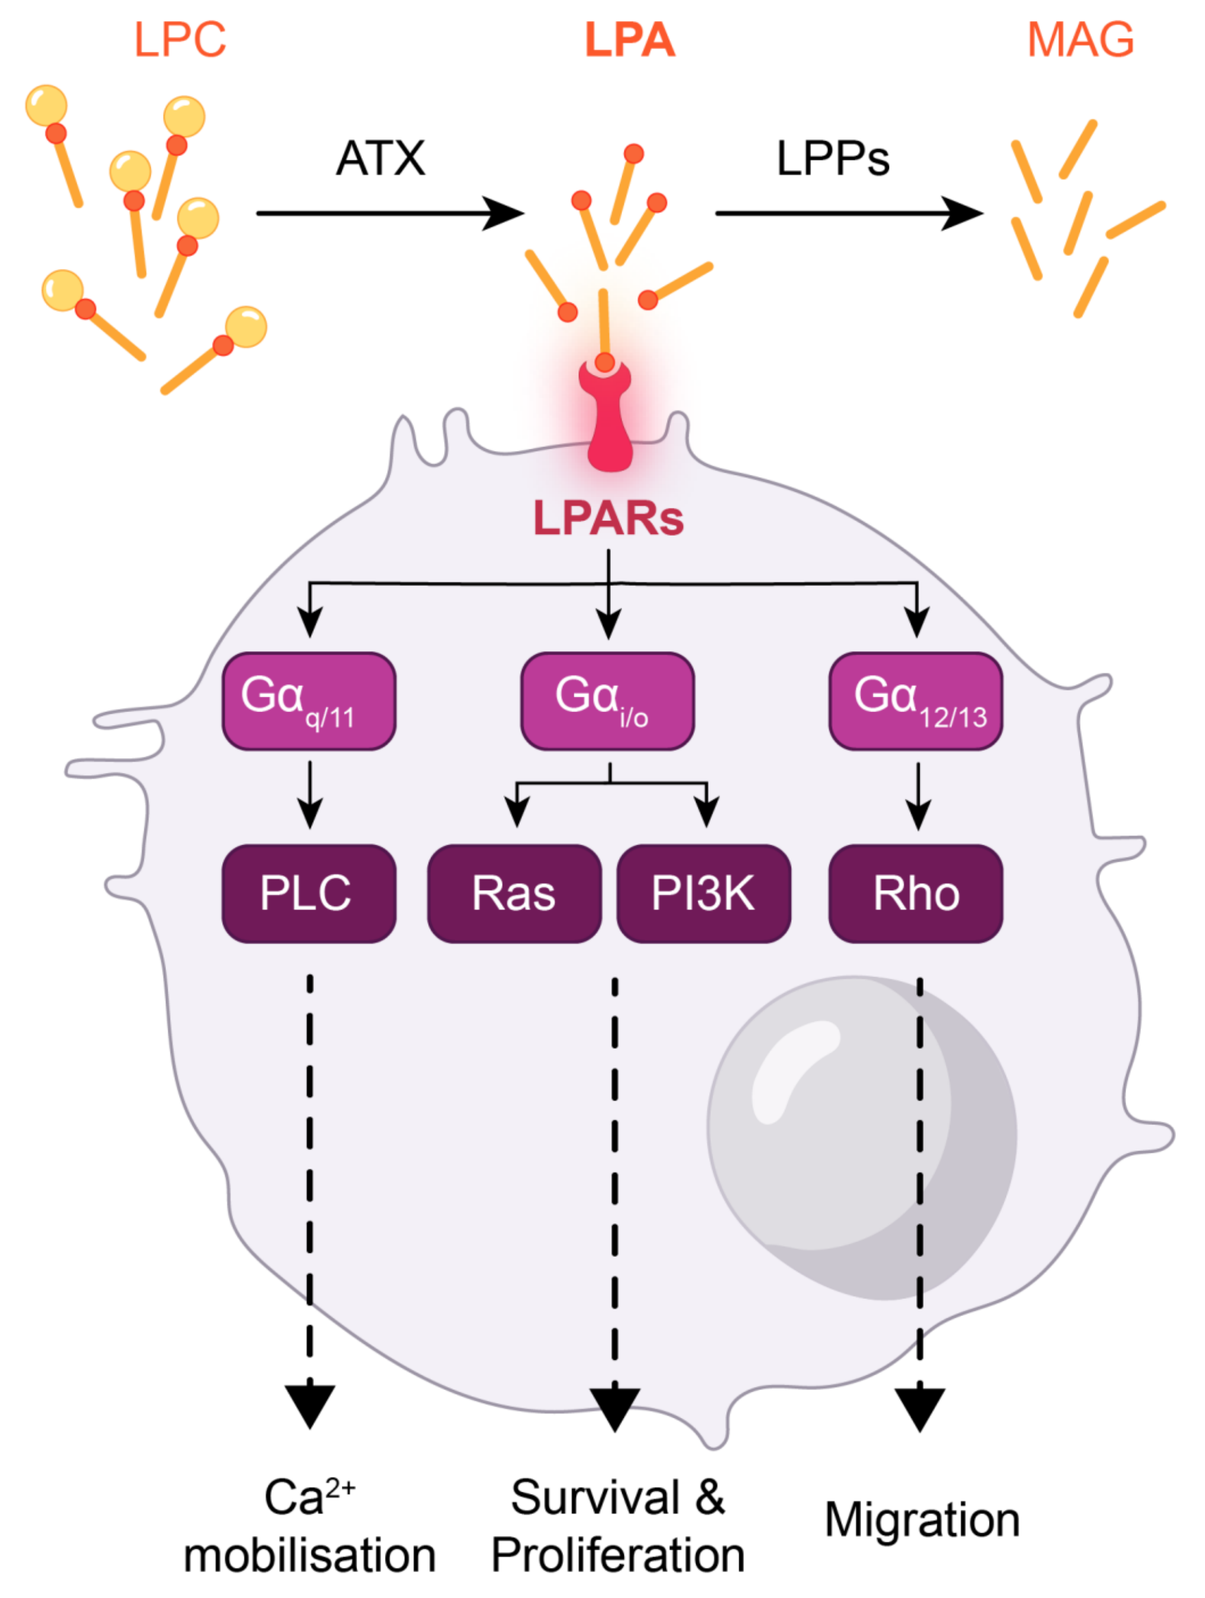
LPC
LPA
MAG
ATX
LPPs
LPARs
Gα q/11
Gα i/o
Gα 12/13
PLC
Ras
PI3K
Rho
Ca2+ mobilisation
Survival & Proliferation
Migration

Methods

- *In vitro* activity on biomarkers of fibrosis was assessed using the BioMAP® screen, profiling with 3 different human primary cell systems.
- *In vivo* efficacy was studied in the bleomycin-induced pulmonary fibrosis (BLM) model in C57BL/6 mice with prophylactic treatment of compounds for the study duration. For histopathological evaluation, lung tissue was stained according to Crossman´s Trichrome and assessed using the Ashcroft score for fibrosis.
- LPA levels in plasma and bronchoalveolar lavage fluid (BALF) were quantified by LC-MS/MS.

IOA-289 is a potent ATX inhibitor

Characteristics of IOA-289.

Parameter	IOA-289	GLPG1690
IC_{50} ATX	10 nM	39 nM
IC_{50} LPA 18:2 (human plasma)	34 nM	98 nM
MW (Da)	502	588
Caco2 permeability / efflux ratio	$25 \cdot 10^{-6} cms^{-1}$ / 1.5	$6.7 \cdot 10^{-6} cms^{-1}$ / 4.1
Oral bioavailability (m/r)	41% / 37%	29% / 37%
CEREP safety screen (safety factor for 44 off-targets)	> 100	

IOA-289 is a potent modulator of fibrosis

The BioMAP® Fibrosis panel consists of systems that models fibrotic tissues and disease by co-culturing stimulated human primary cells; lung epithelial cells and fibroblasts, lung fibroblasts only or renal epithelial cells with lung fibroblasts. IOA-289 and GLPG1690 were tested at concentrations of 6, 1.2 and 0.24 µM. At 6 µM IOA-289 decreased the activity of fibrosis relevant factors, including sIL-6, MCP-1, αSMA, collagen-III, and sVEGF. In contract to IOA-289, GLPG1690 did not show any significant effect in this assay in the pulmonary and general fibrosis system (Figure 2).

Figure 2

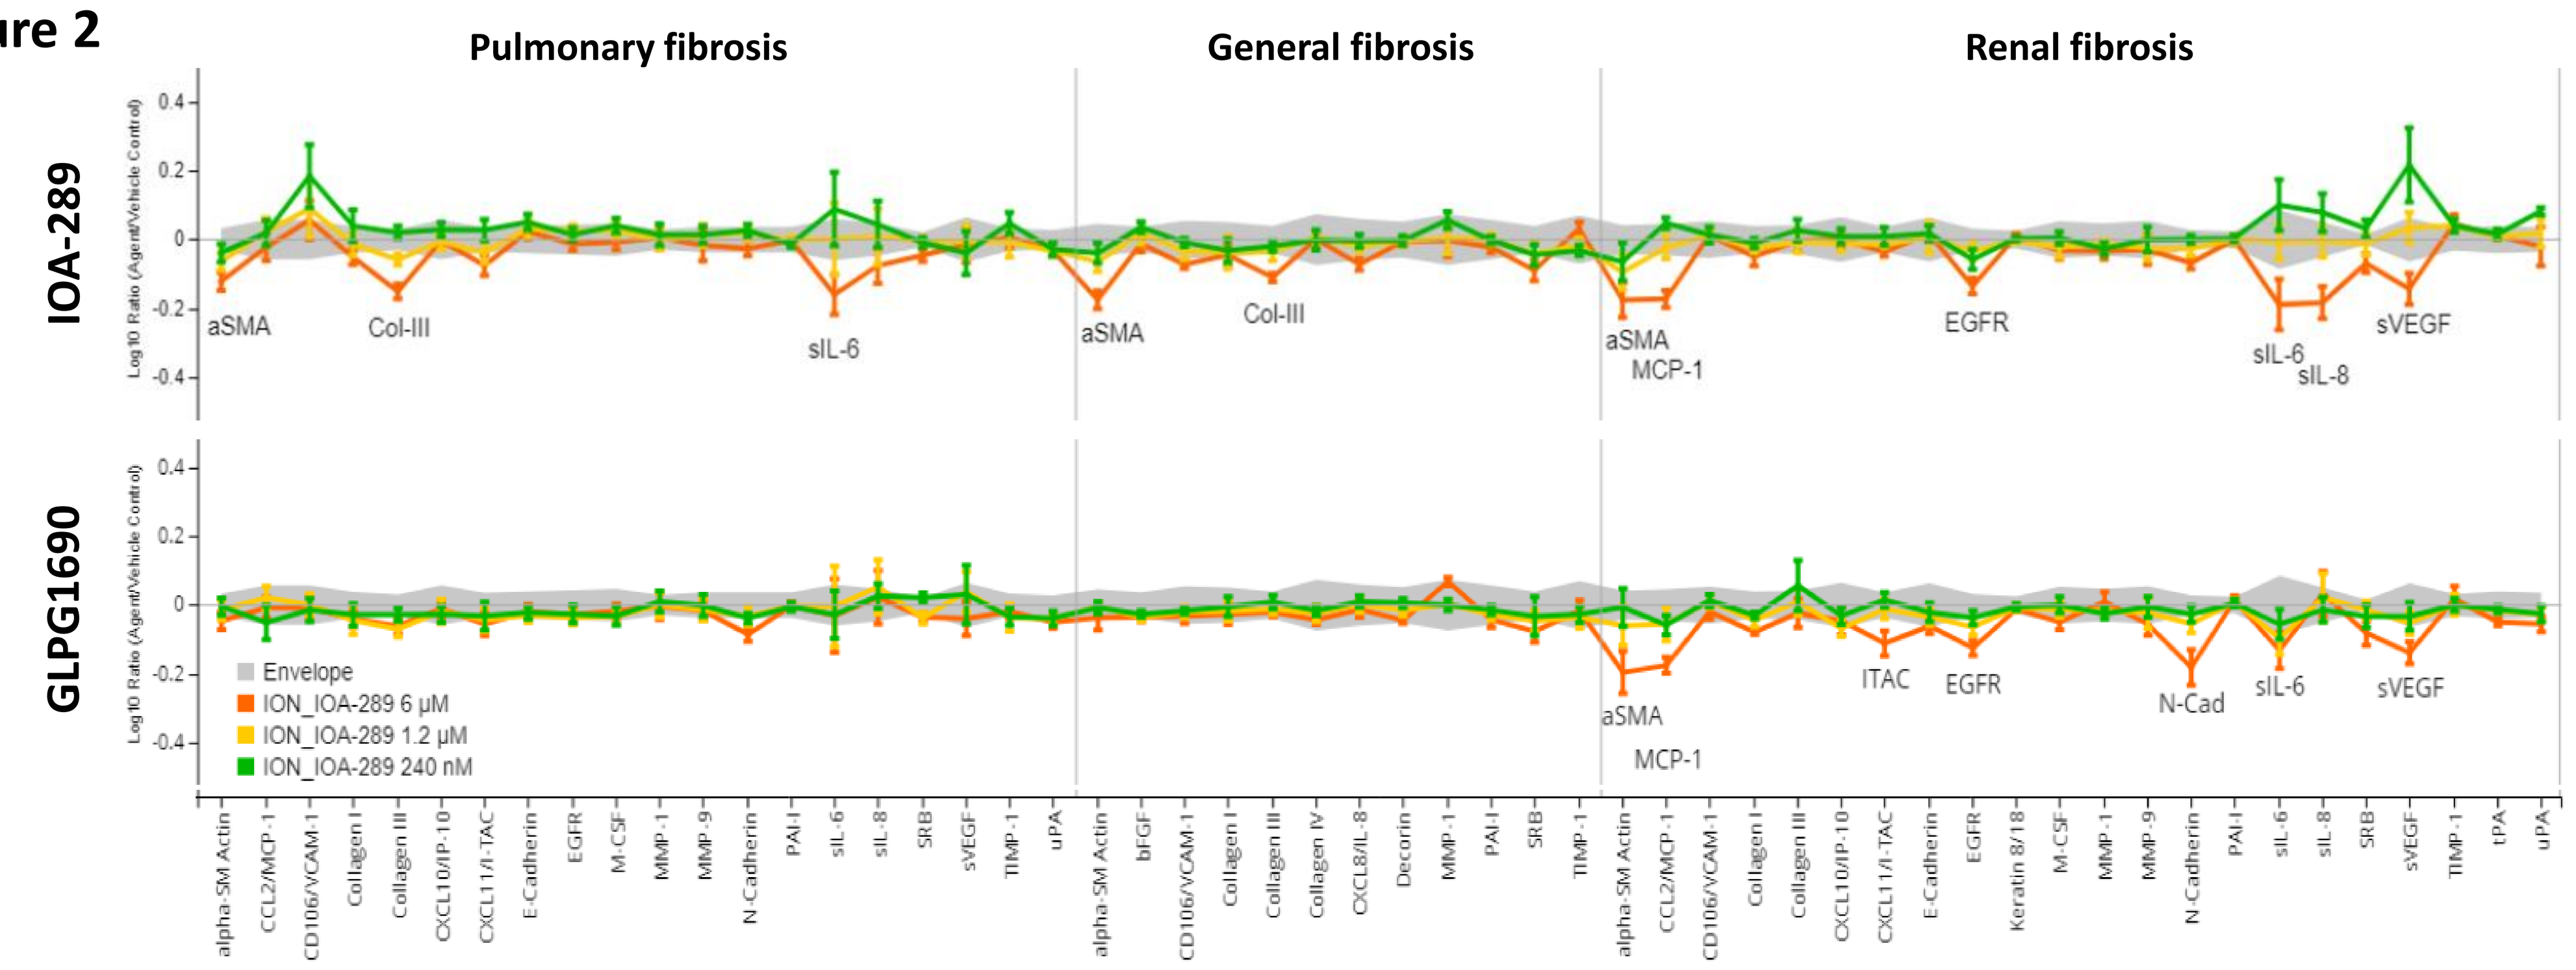
Pulmonary fibrosis
General fibrosis
Renal fibrosis
IOA-289
GLPG1690
Log10 Ratio (Agent/Vehicle Control)
aSMA
Col-III
sIL-6
MCP-1
EGFR
sIL-8
sVEGF
ITAC
N-Cad
Envelope
ION_IOA-289 6 µM
ION_IOA-289 1.2 µM
ION_IOA-289 240 nM

IOA-289 is efficacious in bleomycin lung fibrosis mouse model

In vivo efficacy testing of IOA-289 at 10 mg/kg p.o. BID in the intranasal bleomycin (BLM) induced lung fibrosis model showed marked reduction of efficacy read-outs by Ashcroft score Figure 3A) and collagen content (Figure 3B), equal or better than standard-of-care nintedanib dosed at 60 mg/kg BID p.o.

Figure 3

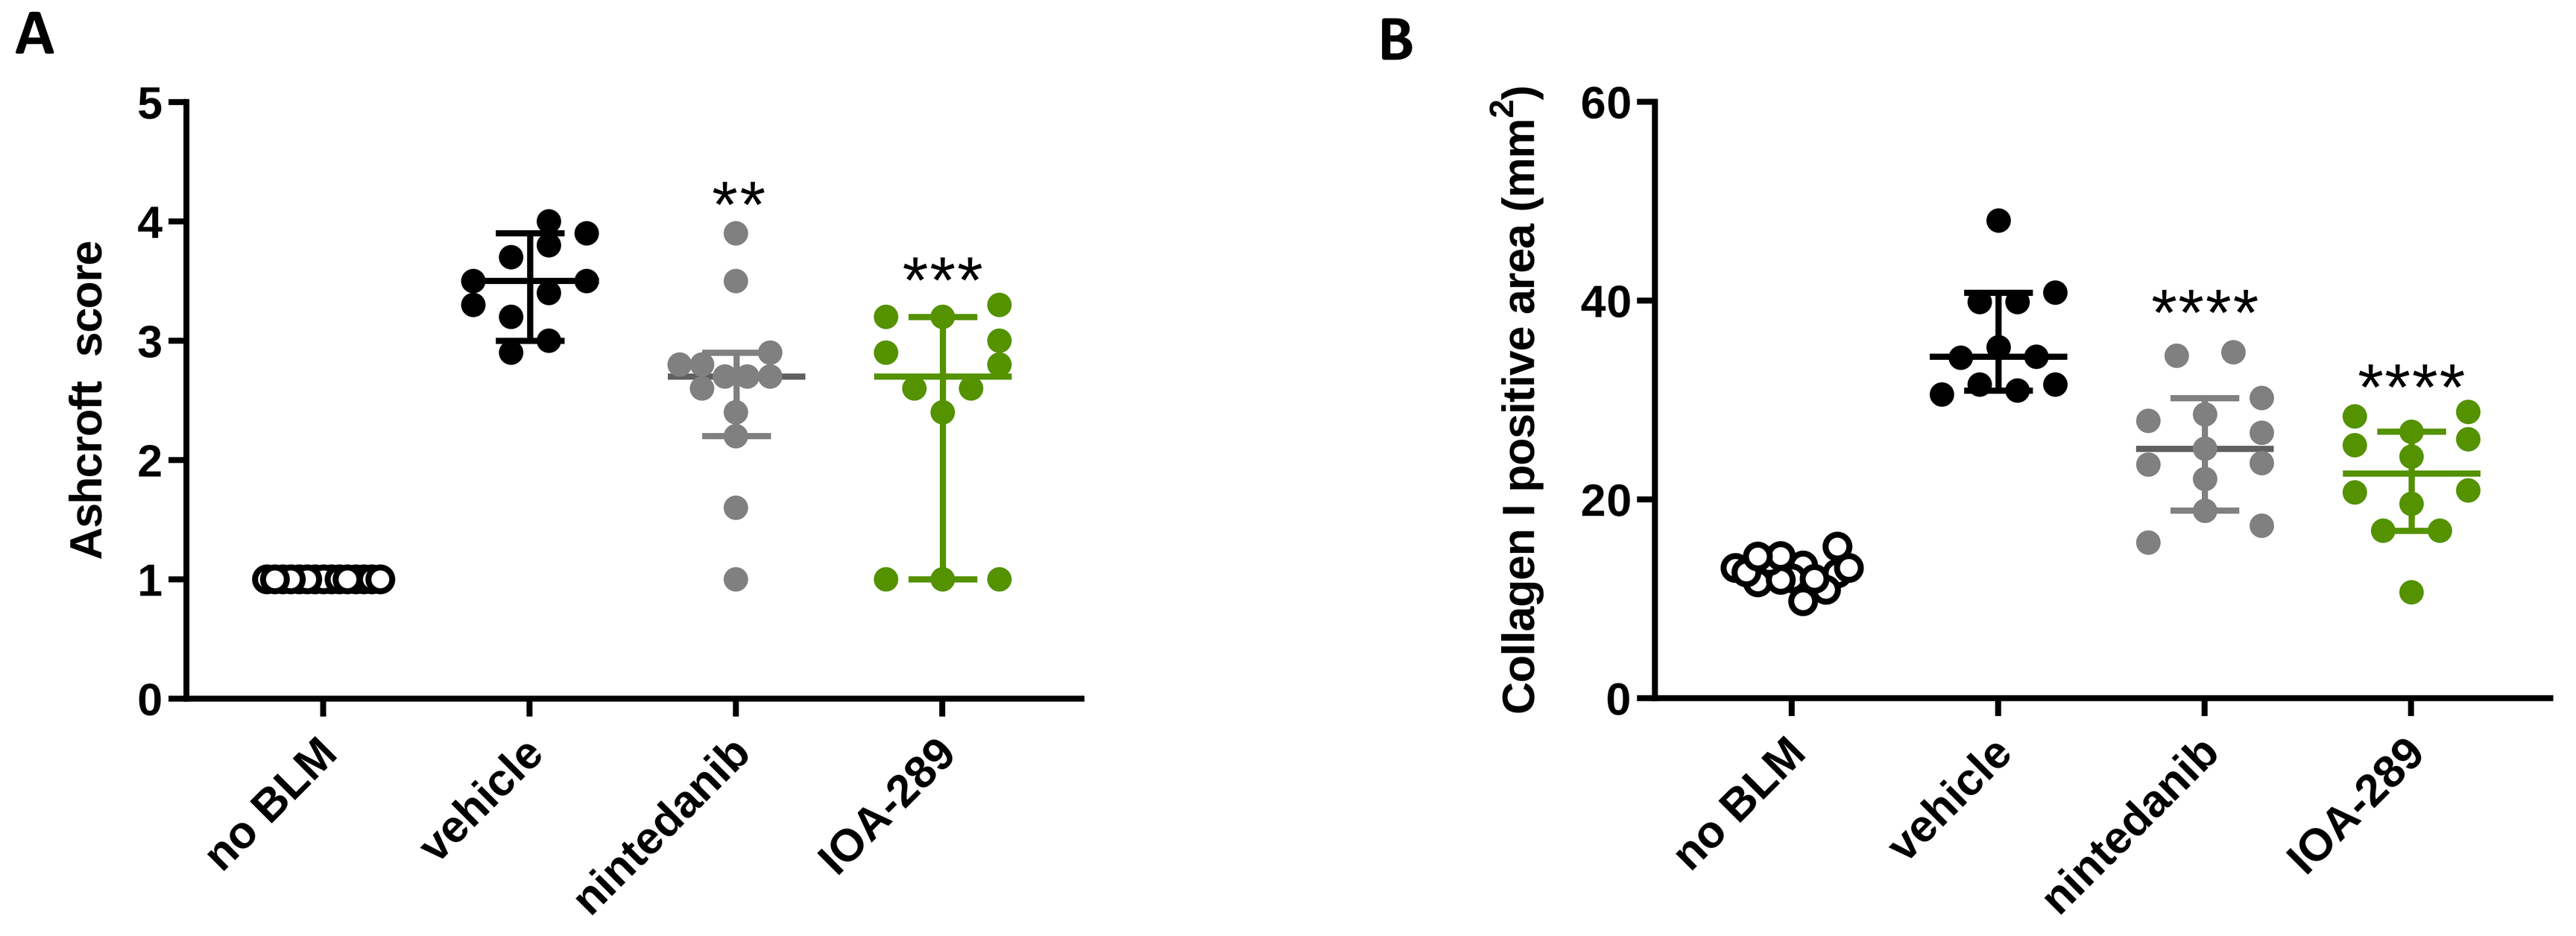
A
B
Ashcroft score
Collagen I positive area (mm2)
no BLM
vehicle
nintedanib
IOA-289
**

In an independent experiment, IOA-289 was tested head-to-head with GLPG1690 both at 10 mg/kg p.o. BID. IOA-289 showed a greater reduction in fibrosis score compared with GLPG1690 (Figure 4A). LPA levels in plasma were significantly reduced by both treatments and IOA-289 also markedly reduced LPA levels in BALF (Figure 4B and C).

Figure 4

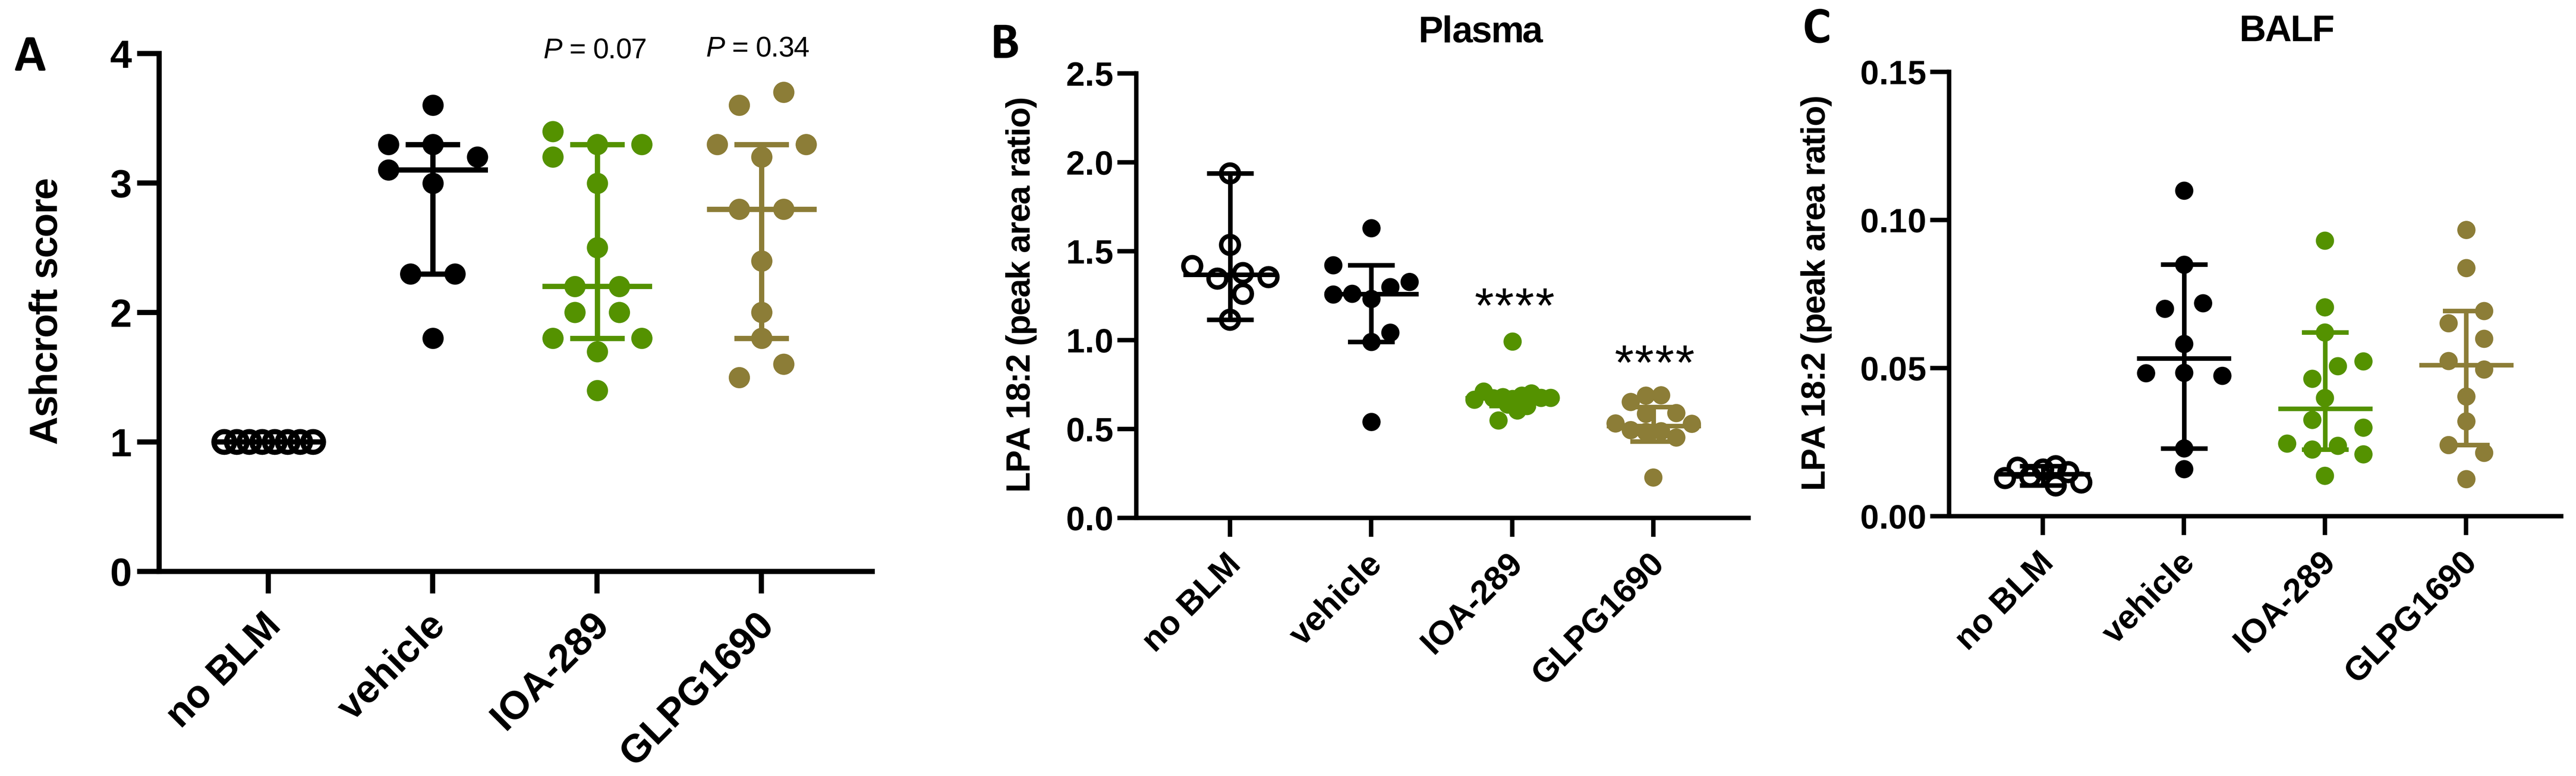
A
B
C
Plasma
BALF
Ashcroft score
LPA 18:2 (peak area ratio)
P = 0.07
P = 0.34

no BLM
vehicle
IOA-289
GLPG1690

IOA-289 PK and PD

Male CD1 mice dosed with 3, 10 or 30 mg/kg of IOA-289 p.o. showed dose-dependent reduction of circulating LPA C18:2 with an ED_{50} value at 1 h post-dose of around 3 mg/kg (Figure 5A and B).

Figure 5

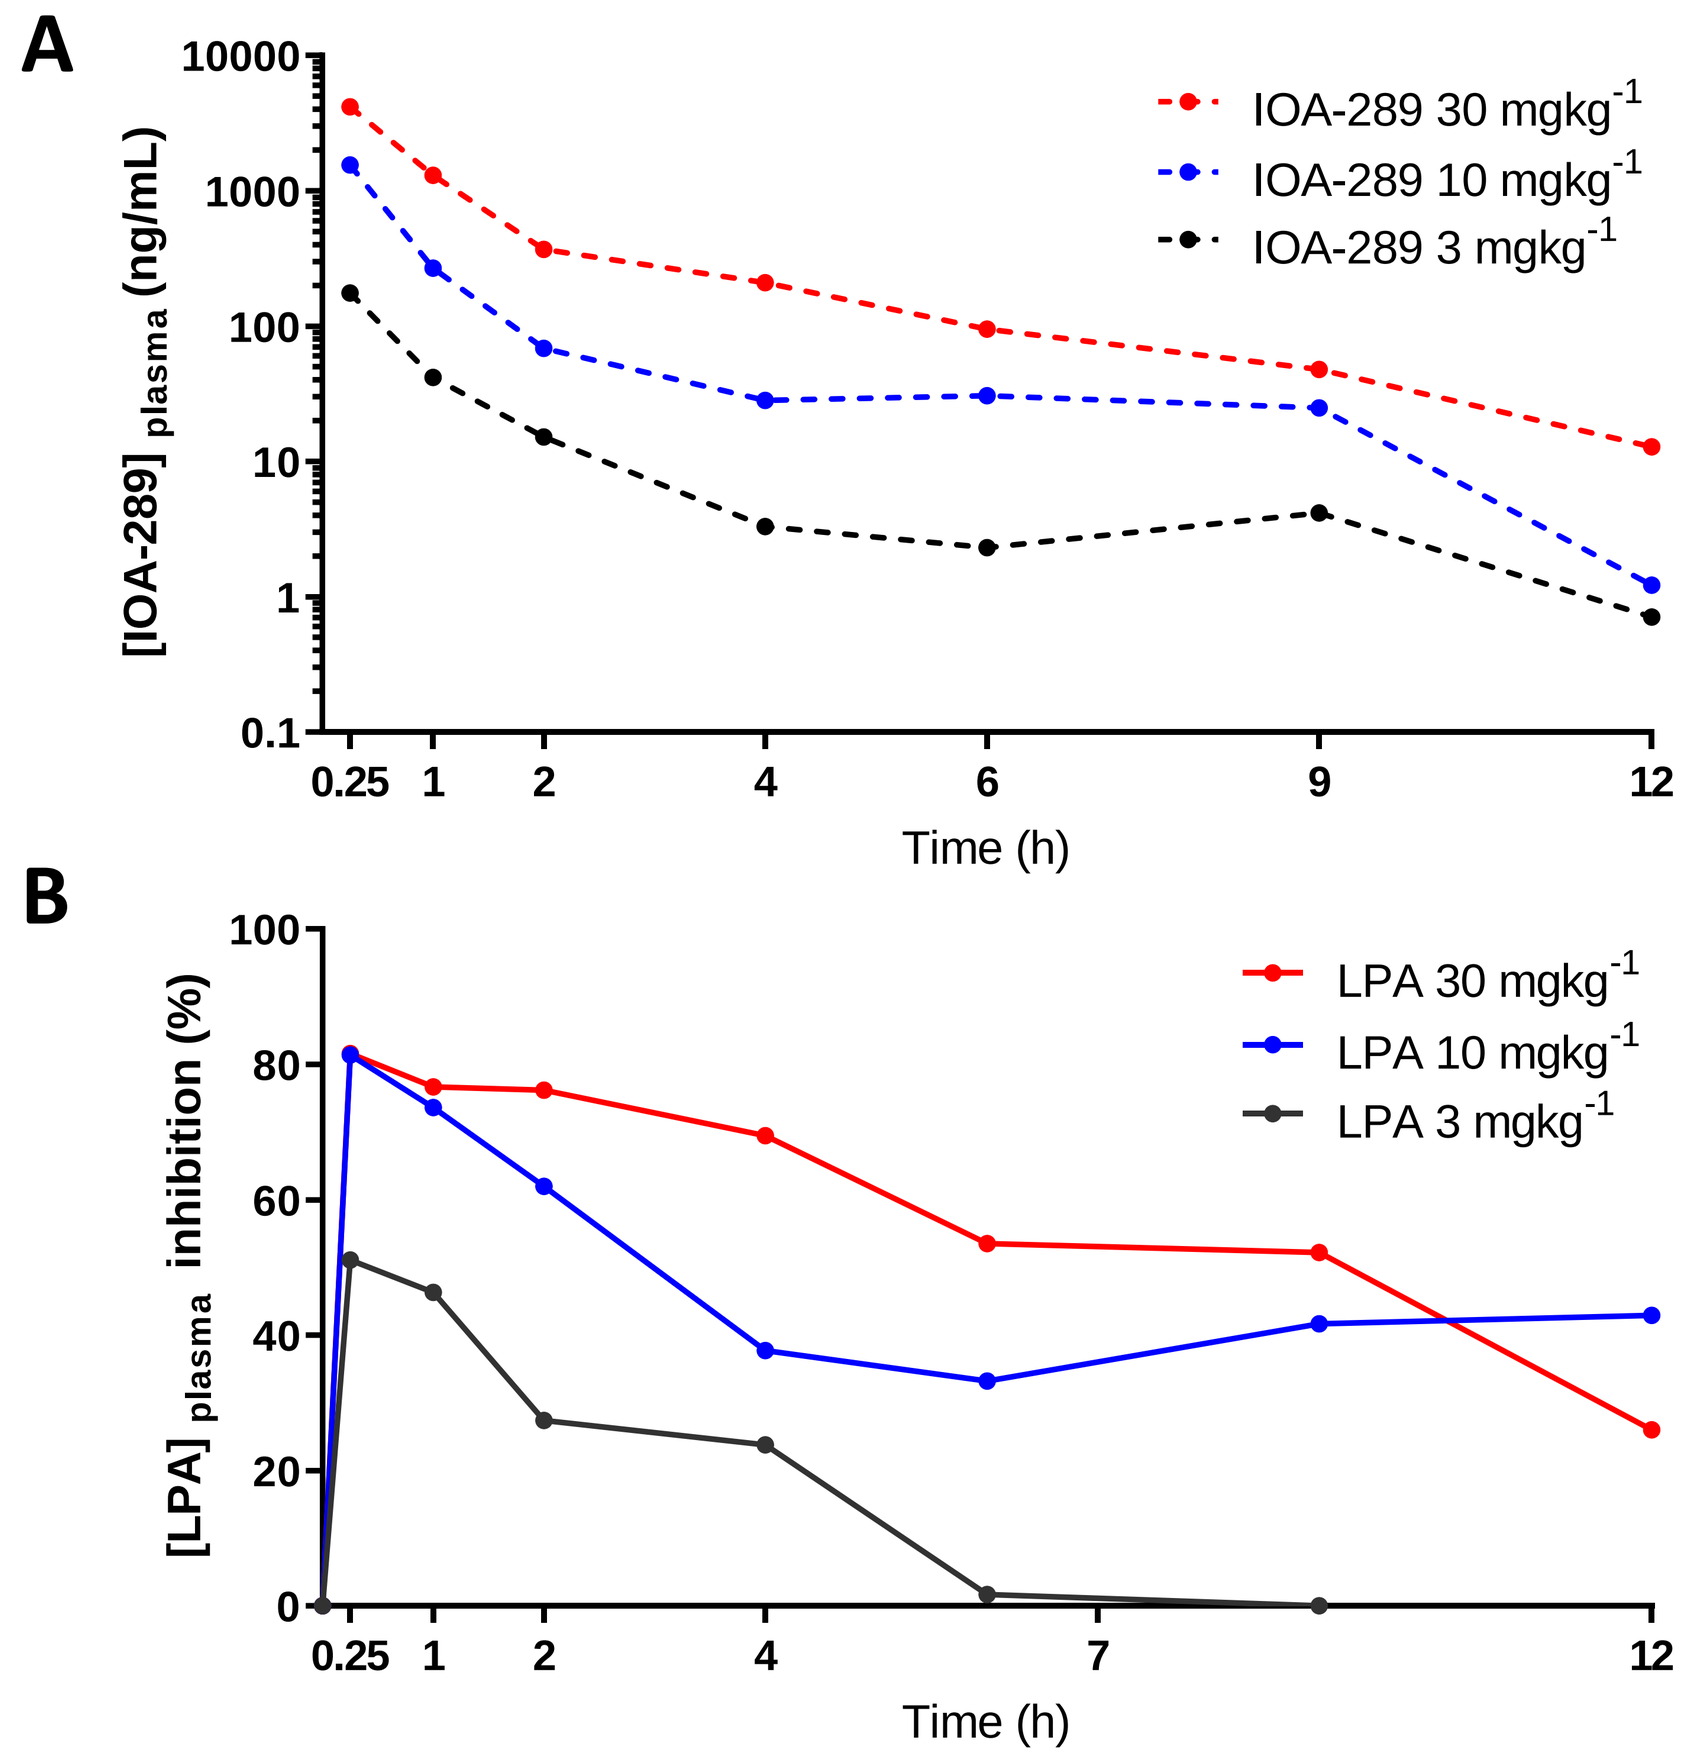
A
B
[IOA-289] plasma (ng/mL)
IOA-289 30 mgkg-1
IOA-289 10 mgkg-1
IOA-289 3 mgkg-1
[LPA] plasma inhibition (%)
LPA 30 mgkg-1
LPA 10 mgkg-1
LPA 3 mgkg-1
Time (h)

Status: Phase I clinical trial

Following successful completion of GLP toxicology studies, IOA-289 has entered a first-in-human clinical trial for safety and tolerability in healthy volunteers. The study consists of a randomized, double-blind, placebo-controlled, single ascending oral dose administration of IOA-289 to healthy male subjects in a fasted state. Subjects will be randomized to a single oral dose of IOA-289 or matching placebo capsules (Figure 6). Pharmacokinetic parameters and reduction of LPA levels will be determined.

Figure 6

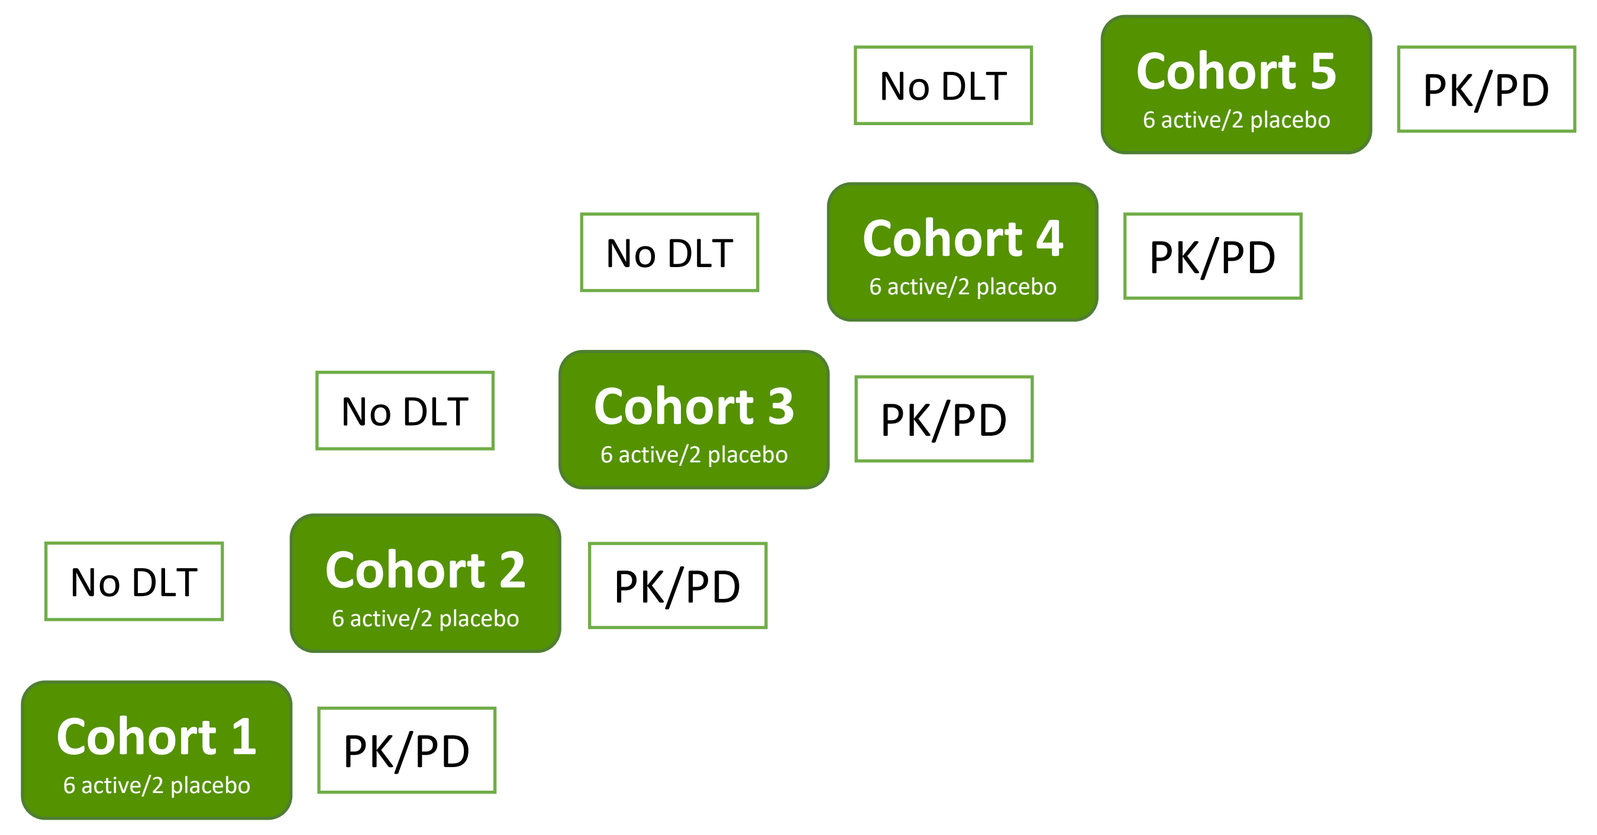
No DLT
Cohort 5
6 active/2 placebo
PK/PD
No DLT
Cohort 4
6 active/2 placebo
PK/PD
No DLT
Cohort 3
6 active/2 placebo
PK/PD
No DLT
Cohort 2
6 active/2 placebo
PK/PD
Cohort 1
6 active/2 placebo
PK/PD

Conclusion

- IOA-289 is a highly potent and selective inhibitor of ATX.
- Inhibition of ATX by IOA-289 showed superior efficacy to standard-of-care and GLPG1690 in the BLM model.
- IOA-289 has an acceptable safety and PK profile and non-clinical safety studies did not raise any safety issues.
- Based on an extensive preclinical package supporting proof of concept in oncology, IOA-289 is entering into clinical trials in the first half of 2021 for fibrotic cancer indications.
- IOA-289 has great potential to be further developed for fibrotic pathologies, including idiopathic pulmonary fibrosis (IPF) and other organ specific fibrosis indications.

Conflict of interest statement:
MD, KNS, ML, LvdV and ZJ are employees and shareholders of iOnctura

m.deken@ionctura.com / z.johnson@ionctura.com

ATS 2021|Virtual Meeting | May 14-19, 2021