IOA-289, an orally available type IV autotaxin inhibitor, ameliorates steatosis and fibrosis in a progressive preclinical murine NASH model

EASL CONGRESS 2023
Vienna, Austria
21–24 June

UNIVERSITY OF TWENTE.

Marit ten Hove [1], Giusy Di Conza [2], Ruchi Bansal [1*]
[1] Translational Liver Research, Faculty of Science and Technology, University of Twente, the Netherlands
[2] iOnctura, Geneve, Switzerland
[*] Correspondence to r.bansal@utwente.nl

1 Introduction

Non-alcoholic steatohepatitis (NASH) represents a major health burden worldwide with no FDA-approved therapies available. Lysophosphatidic acid (LPA), produced by autotaxin (ATX), is correlated with the progression of NASH, and regulates lipid homeostasis, and induces liver inflammation and fibrosis via 6 different G protein-coupled LPA-receptors (LPAR1-6).

Antibody-mediated neutralization of ATX in a high-fat diet (HFD) mouse model showed protection against non-alcoholic fatty liver disease by decreasing hepatic steatosis and fibrosis (Qiu et al., 2022 *CMGH*).

Recently, we have shown that inhibiting ATX using a type-IV inhibitor possess better potential in ameliorating NASH via intraperitoneal administration than a type-I inhibitor (Booijink et al., 2022 *EMBO*).

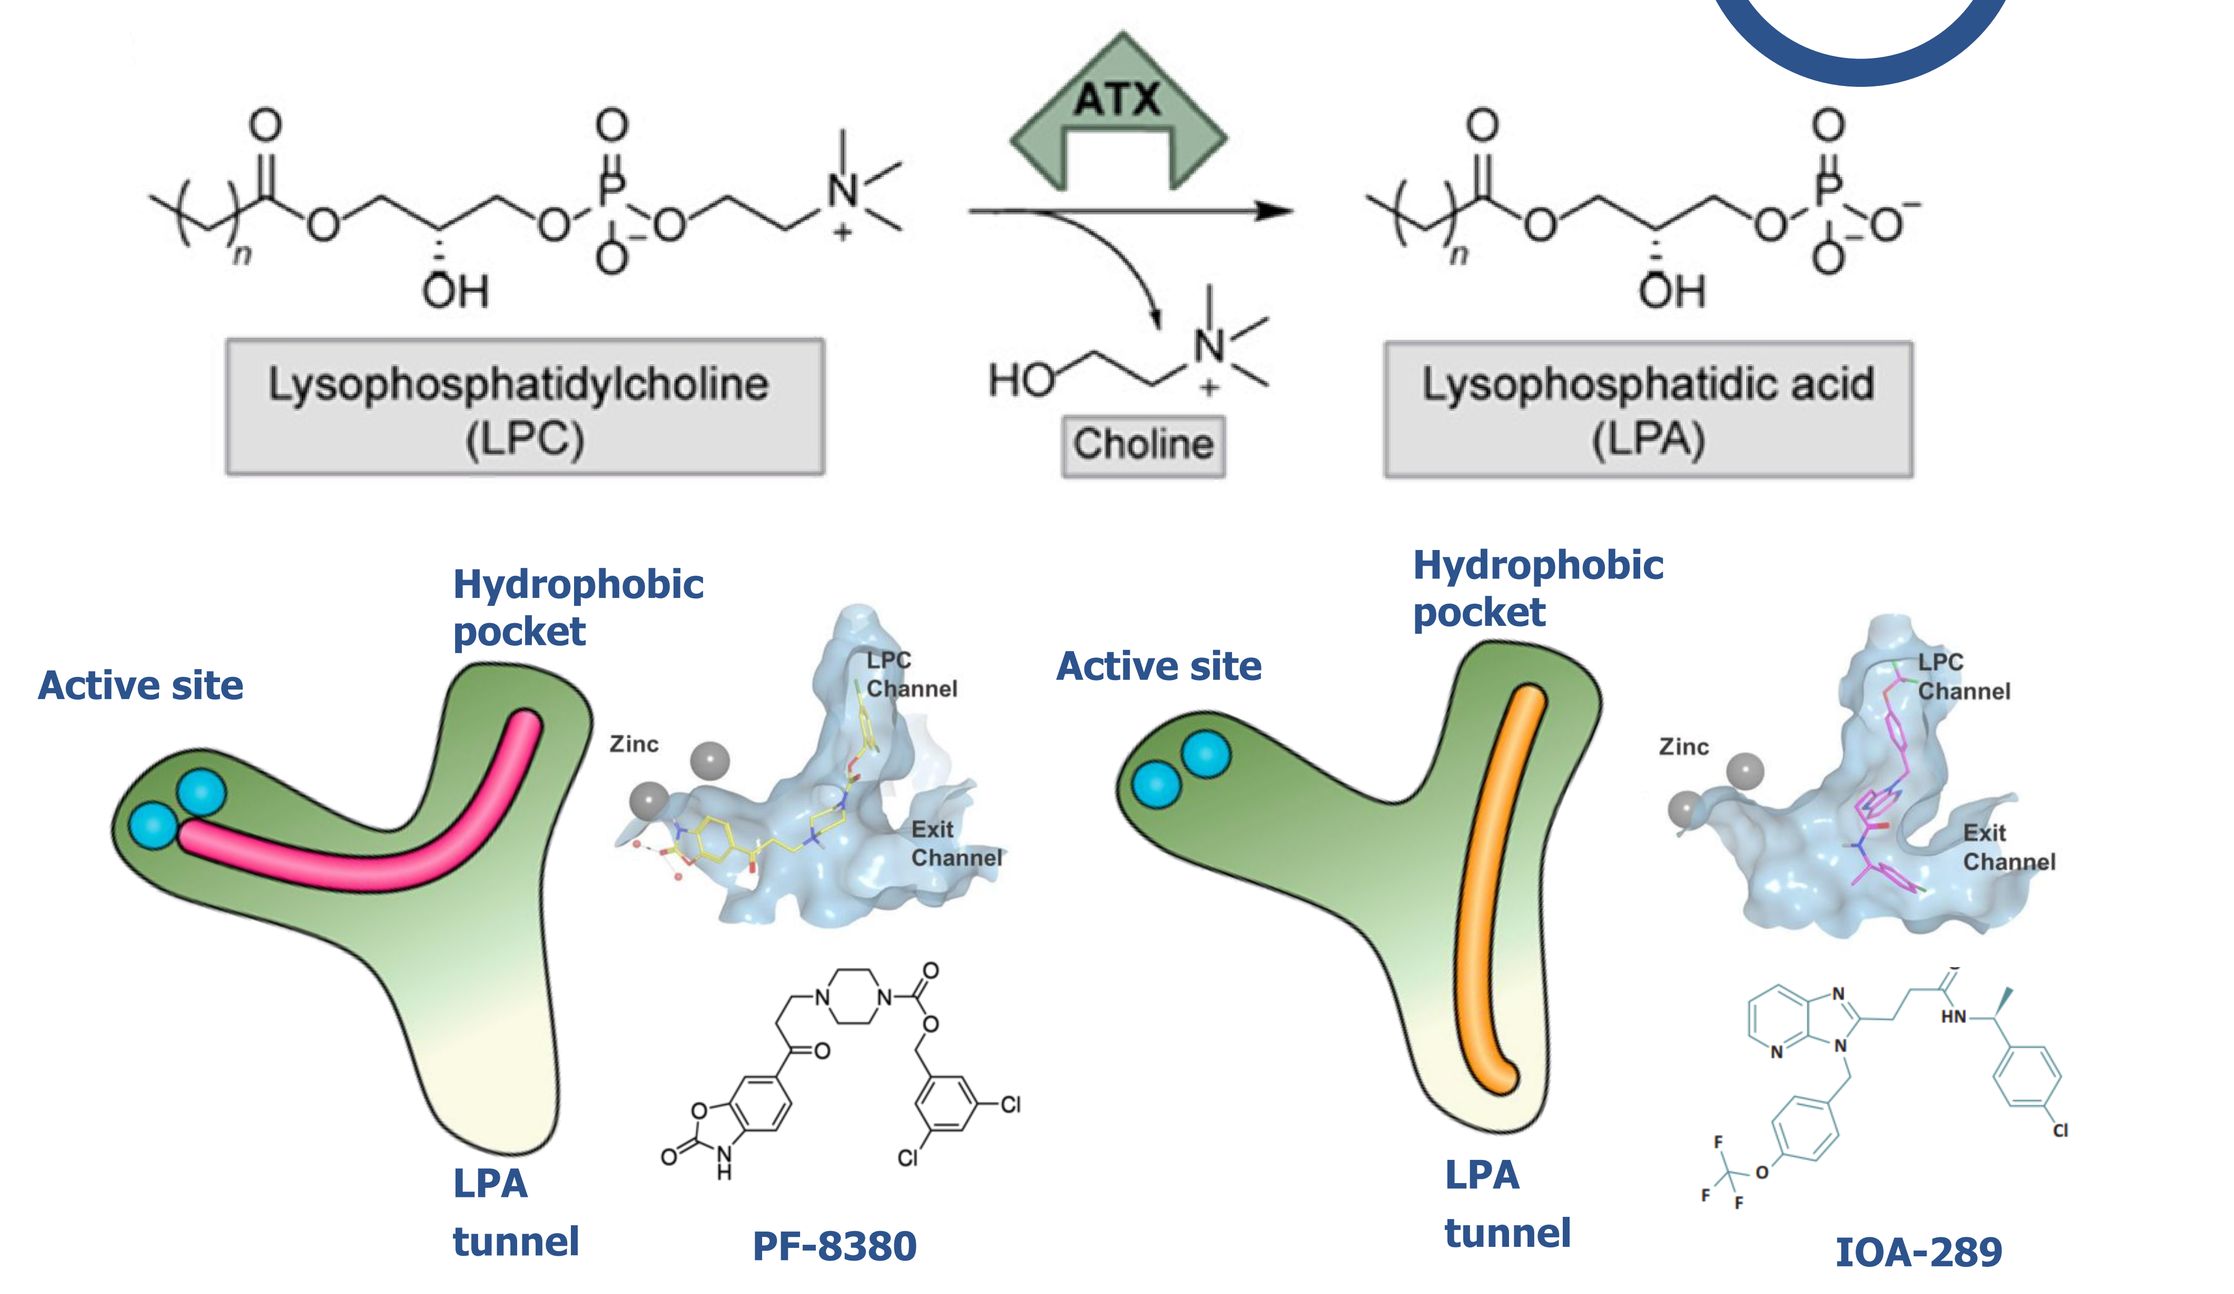
ATX
Lysophosphatidylcholine (LPC)
Choline
Lysophosphatidic acid (LPA)
Active site
Hydrophobic pocket
LPA tunnel
Zinc
LPC Channel
Exit Channel
PF-8380
IOA-289

Type I
- Targets the catalytic site and the pocket of ATX
- Hampers LPC or LPA binding
- Mimicking LPC substrate
- Competitive inhibitors
- PF-8380 (IC_{50}: 2.8 nM)

Type IV
- Occupy the tunnel and the pocket of ATX
- Hampers LPC or LPA binding
- Counteracting modulatory action of LPA
- Potent competitive inhibitors
- IOA-289 (IC_{50}: 10 nM)

Part of image adapted from Clark et al., 2022 – *J. Med. Chem.*

2 Aim

Investigate the novel and orally administrable type IV ATX inhibitor IOA-289 (provided by iOnctura) for the treatment of NASH to attenuate hepatic steatosis and fibrosis.

3 Method

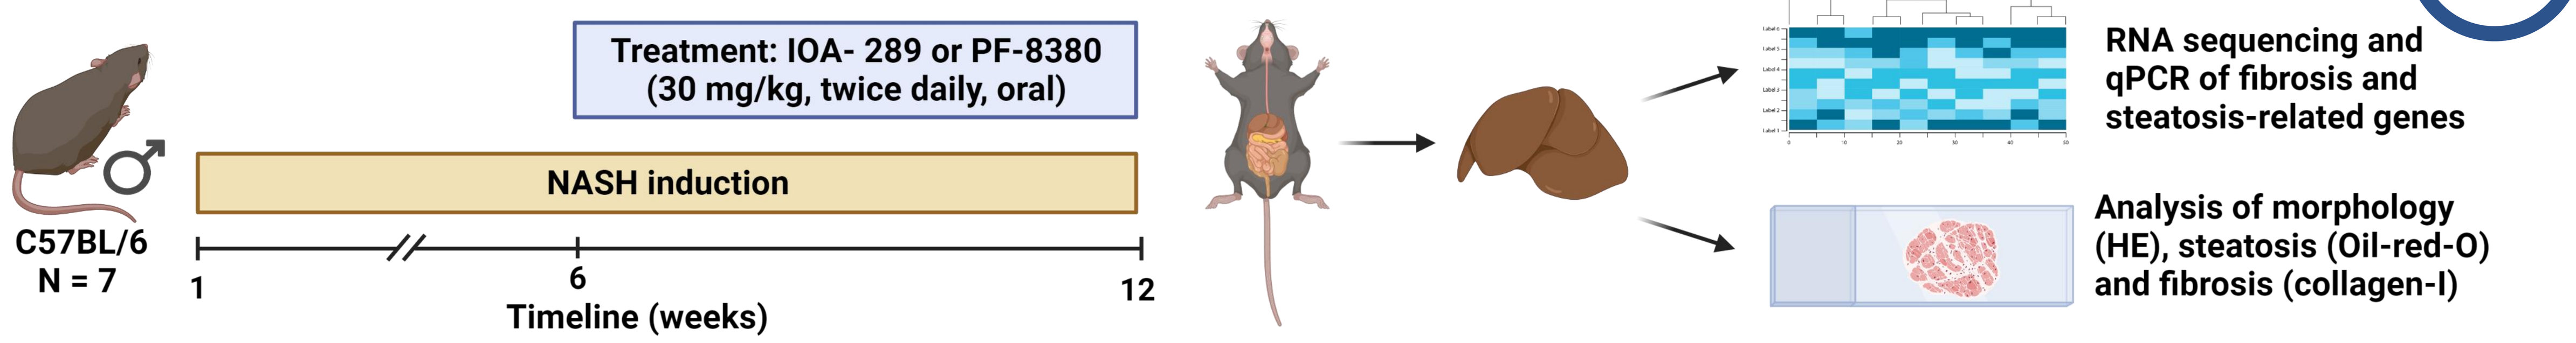
Treatment: IOA- 289 or PF-8380
(30 mg/kg, twice daily, oral)
NASH induction
C57BL/6
N = 7
1
6
12
Timeline (weeks)
RNA sequencing and qPCR of fibrosis and steatosis-related genes
Analysis of morphology (HE), steatosis (Oil-red-O) and fibrosis (collagen-I)

Diet- and chemical-induced western diet fructose (WDF) NASH mouse model:

Western Diet – *ad libitum*
- 21.1% fat
- 34.0% sucrose
- 1.25% cholesterol

Fructose supplemented water – *ad libitum*
- 23.1 g/L fructose
- 18.9 g/L glucose

Carbon tetrachloride (CCl4) – once a week via i.p. administration
- 0.05 – 0.2 mL/kg (week 1-4)
- 0.2 mL/kg (week 5-12)

4 Results

ATX inhibition attenuates hepatic lipid accumulation by decreased *de novo* lipogenesis

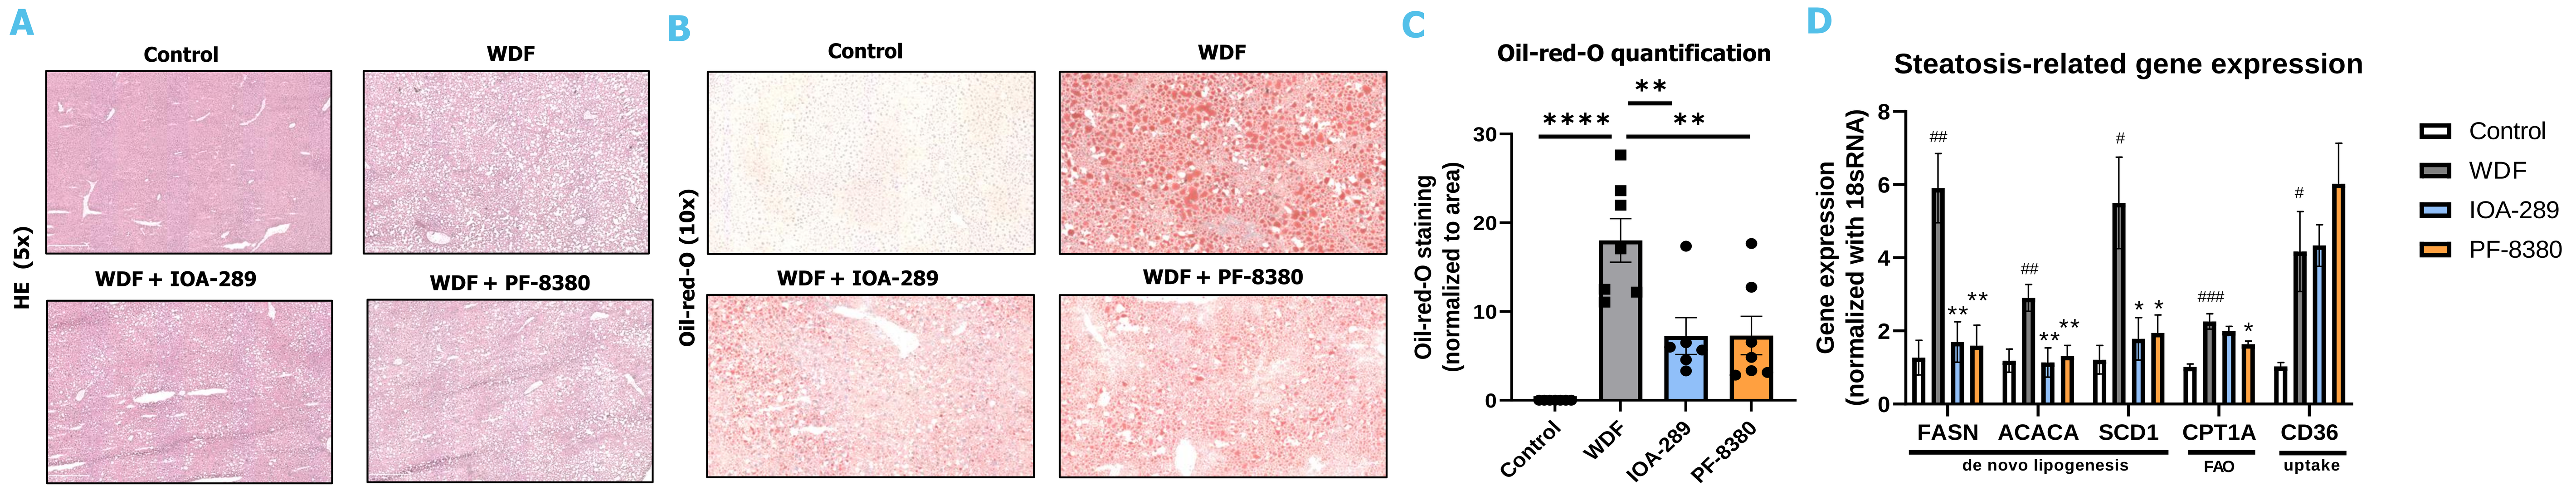
A
HE (5x)
Control
WDF
WDF + IOA-289
WDF + PF-8380
B
Oil-red-O (10x)
Control
WDF
WDF + IOA-289
WDF + PF-8380
C
Oil-red-O quantification
Oil-red-O staining (normalized to area)
Control
WDF
IOA-289
PF-8380
D
Steatosis-related gene expression
Gene expression (normalized with 18sRNA)
FASN
ACACA
SCD1
CPT1A
CD36
de novo lipogenesis
FAO
uptake
Control
WDF
IOA-289
PF-8380

(A) Representative images of the liver sections stained with haematoxylin and eosin (HE). **(B)** Representative images of the liver sections stained with Oil-red-O and **(C)** quantitative image analysis of Oil-red-O stained liver sections shows significant decrease in lipid accumulation after ATX inhibition. **(D)** Graphs showing the expression of steatosis-related genes in liver tissue of control mice and WDF mice with and without ATX inhibitors. The results show significant downregulation of de novo lipogenesis related genes. Multiple comparisons between different groups were performed by ANOVA with Bonferroni post-hoc test. *$p < 0.05$, **$p < 0.01$, ***$p < 0.001$, ****$p < 0.0001$. All together these results indicate that ATX inhibition (both type I and type IV) decrease the hepatic lipid accumulation via decreased *de novo* lipogenesis in a WDF NASH model.

Type IV ATX inhibition decreases activation of fibroblasts resulting in decreased extracellular matrix deposition

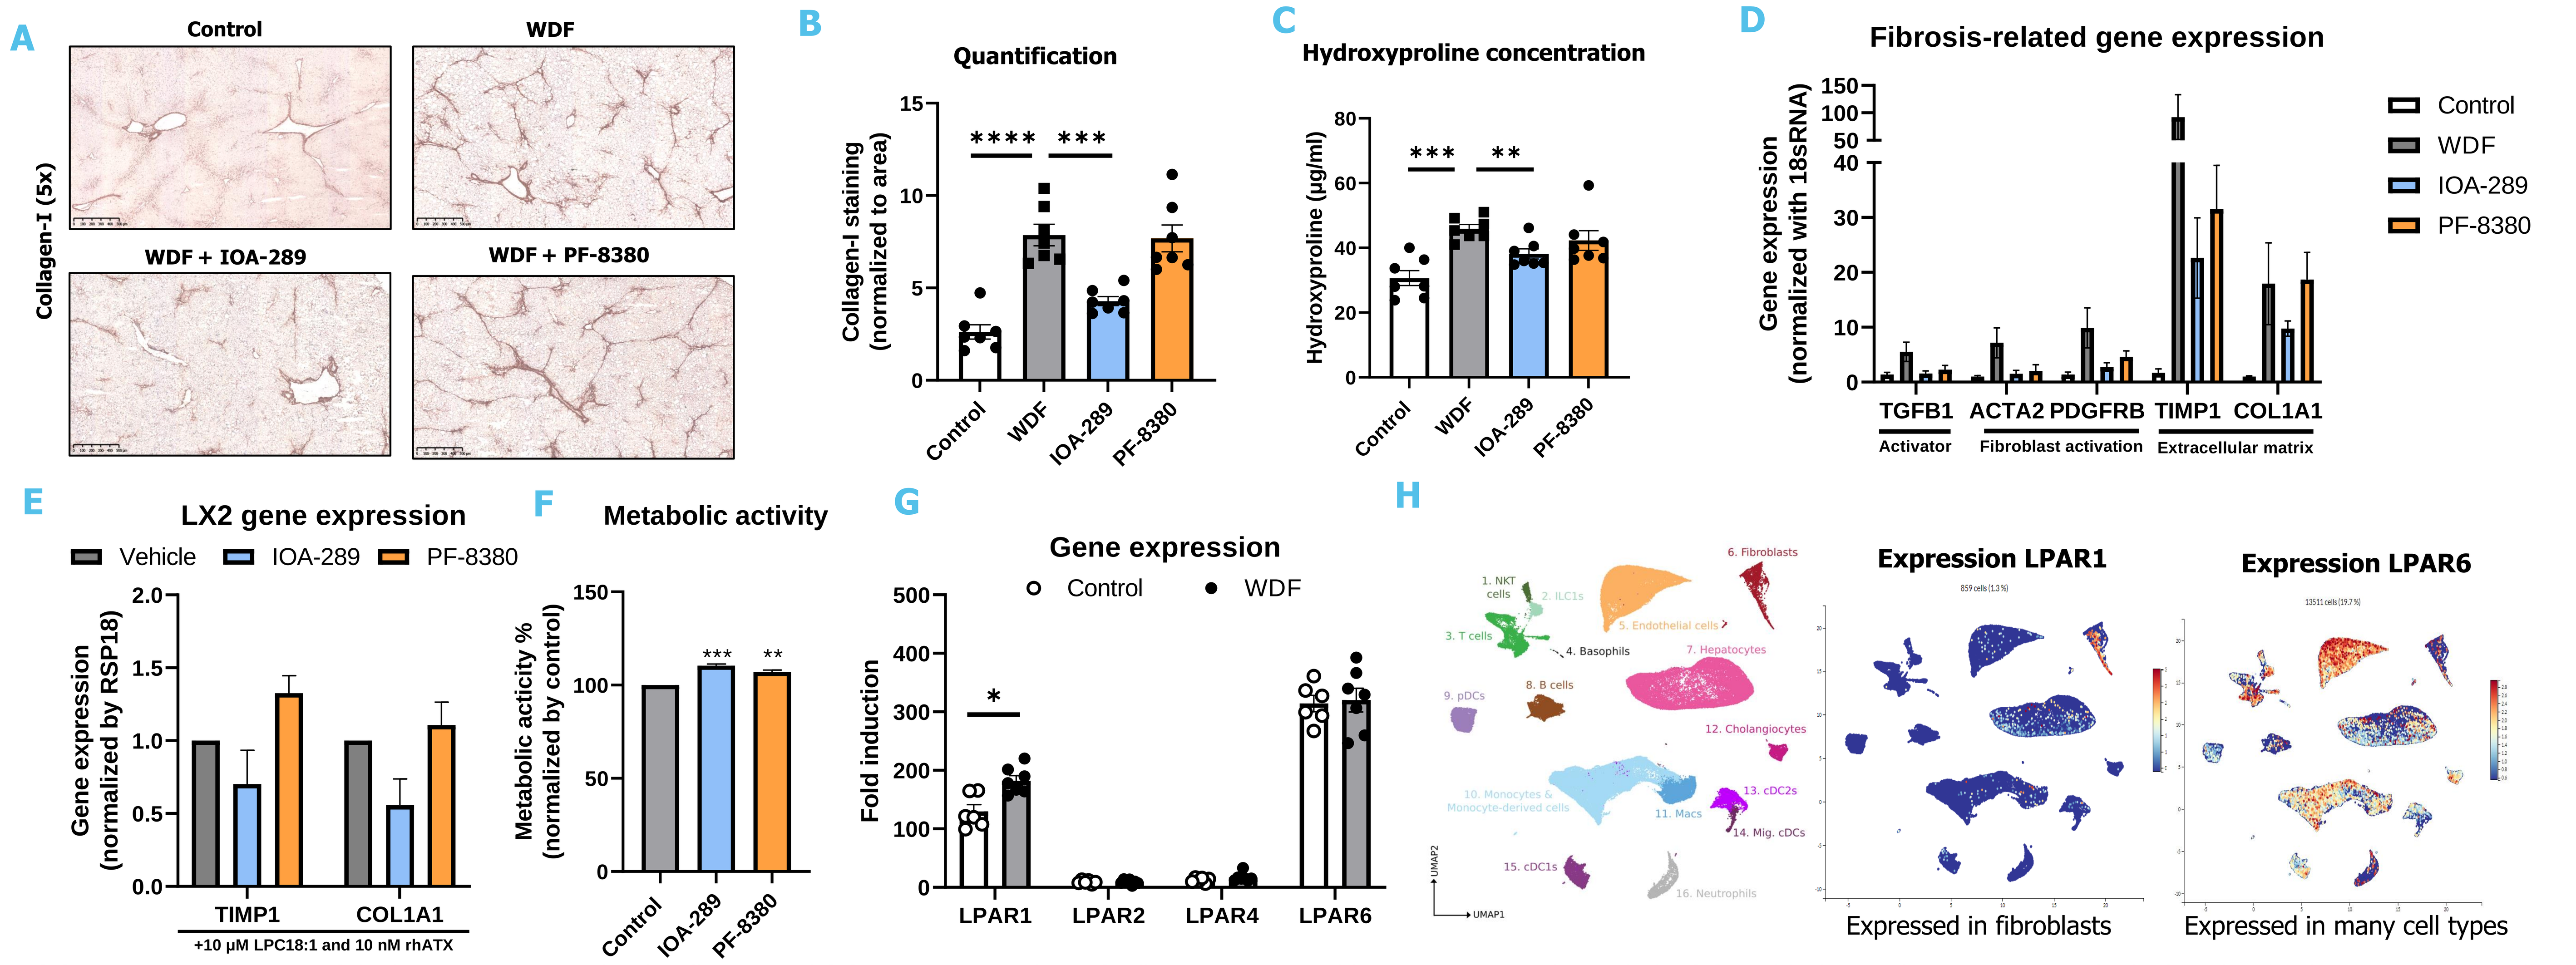
A
Collagen-I (5x)
Control
WDF
WDF + IOA-289
WDF + PF-8380
B
Quantification
Collagen-I staining (normalized to area)
C
Hydroxyproline concentration
Hydroxyproline (µg/ml)
D
Fibrosis-related gene expression
Gene expression (normalized with 18sRNA)
TGFB1
ACTA2
PDGFRB
TIMP1
COL1A1
Activator
Fibroblast activation
Extracellular matrix
Control
WDF
IOA-289
PF-8380
E
LX2 gene expression
Vehicle
IOA-289
PF-8380
Gene expression (normalized by RSP18)
TIMP1
COL1A1
+10 µM LPC18:1 and 10 nM rhATX
F
Metabolic activity
Metabolic acticity % (normalized by control)
G
Gene expression
Control
WDF
Fold induction
LPAR1
LPAR2
LPAR4
LPAR6
H
1. NKT cells
2. ILC1s
3. T cells
4. Basophils
5. Endothelial cells
6. Fibroblasts
7. Hepatocytes
8. B cells
9. pDCs
10. Monocytes & Monocyte-derived cells
11. Macs
12. Cholangiocytes
13. cDC2s
14. Mig. cDCs
15. cDC1s
16. Neutrophils
UMAP1
UMAP2
Expression LPAR1
Expression LPAR6
Expressed in fibroblasts
Expressed in many cell types

(A) Representative images of the liver sections stained with collagen-I and **(B)** quantitative analysis of collagen-I IHC-stained liver sections, together with the **(C)** total hydroxyproline concentration (µg/mL) analysed in liver tissue indicate the reduction of fibrotic extracellular matrix upon treatment with type IV ATX inhibition (IOA-289), but not with type I ATX inhibition (PF-8380). **(D)** Graphs showing the expression of fibrosis-related genes in liver tissue of control mice and WDF mice with and without ATX inhibition. The results show downregulation trend in HSCs activation and extracellular matrix deposition with IOA-289 treatment. **(E)** Gene expression of major fibrosis markers in human hepatic stellate cells (HSCs, LX2) stimulated with 10 µM LPC18:1 and 10 nM recombinant human ATX, upon treatment with IOA-289 and PF-8380 showed the same trend as in the WDF NASH mouse model, while **(F)** there is no indication of toxic effects indicating a direct effect of IOA-289 on (activated) HSCs. Multiple comparisons between different groups were performed by ANOVA with Bonferroni post-hoc test. *$p < 0.05$, **$p < 0.01$, ***$p < 0.001$, ****$p < 0.0001$. **(G)** The RNA expression of the different LPA receptors retrieved from bulk RNA sequencing of liver tissue from WDF mouse model (n=6-7), showing only a significant increase of *Lpar1* in WDF NASH mice compared to control. Comparison between groups were performed by t-test. *$p < 0.05$. **(H)** Using the Mouse NAFLD Atlas – All Liver Cells UMAPs clusters, provided by https://livercellatlas.org/, looking into single cell expression of *Lpar1* and *Lpar6* in the mouse NAFLD atlas visualized in all liver cell UMAPs clusters. In these clusters it is shown that *Lpar1* is predominantly expressed by fibroblasts, while *Lpar6* is expressed by multiple different cell types, indicating that the activation of fibroblast is (partly) induced via LPA-LPAR1 signaling. All together these results indicate that treatment IOA-289, a type IV ATX inhibitor, decreases the activation of fibroblasts resulting in deceased extracellular matrix deposition in a WDF NASH model.

5 Conclusions

Our results demonstrate that inhibition of the LPA-ATX pathway, particularly using an orally administrable type IV inhibitor, represents a potential therapeutic target in NASH, by attenuating steatosis and fibrosis, with possible clinical implications.

6 Acknowledgements

TLR Translational Liver Research
iOnctura
MCBP