PRECLINICAL ACTIVITY OF NOVEL TGF BETA RECEPTOR I KINASE INHIBITORS IOA-359 AND IOA-360 FOR TREATMENT OF ANEMIA IN MDS/AML

N. RAMACHANDRA MS[1], C.T.R. VEGIVINTI MD[1], S. SAHU MS[1], B. RIVERA-PEÑA PHD[1], S. AMINOV[1], A. FROMOWITZ, MD[1], J. ZHANG, MD[1], F. AODENGTUYA[1], K. PRADHAN, PHD[1], R. ZHAO, PHD[1], L. SCHWARTZ, MD[1], V. THIRUVANANTHAPURAM, MS[1], R. SANAWAR, PHD[1], E. RABINOVICH, MD[1], O. AJIBADE, MD[1], A. SHASTRI, MD[1], M. LAHN, MD[2], and A. VERMA, MD[1]

1. Blood Cancer Institute, Department of Oncology, Albert Einstein College of Medicine, Bronx, NY
2. iOnctura SA, Avenue de Secheron 15, 1202 Geneva, Switzerland

INTRODUCTION

Overactivation of the Transforming growth factor beta (TGF-β1) superfamily has been associated with bone marrow failure in MDS. TGF-β1 binds to set of receptors that include the TGF-receptor I kinase (also known as ALK5), that in turn phosphorylates and activates the downstream SMAD2/3 proteins. Activation of SMAD2/3 transcription factors has been shown to occur in MDS and is associated with anemia. Thus, we wanted to evaluate the preclinical efficacy of novel, clinic-ready, TGF-B1receptor I kinase small molecule ALK5 inhibitors IOA-359 and IOA-360 in MDS models.

AIM

Our goal of this study is to evaluate the efficacy of IOA-359 and IOA-360 in inhibiting downstream phosphorylation and activation of the SMAD 2/3. We also wanted to determine cellular activity of these novel ALK5 inhibitors in MDS models.

TGF-β role in Hematopoiesis

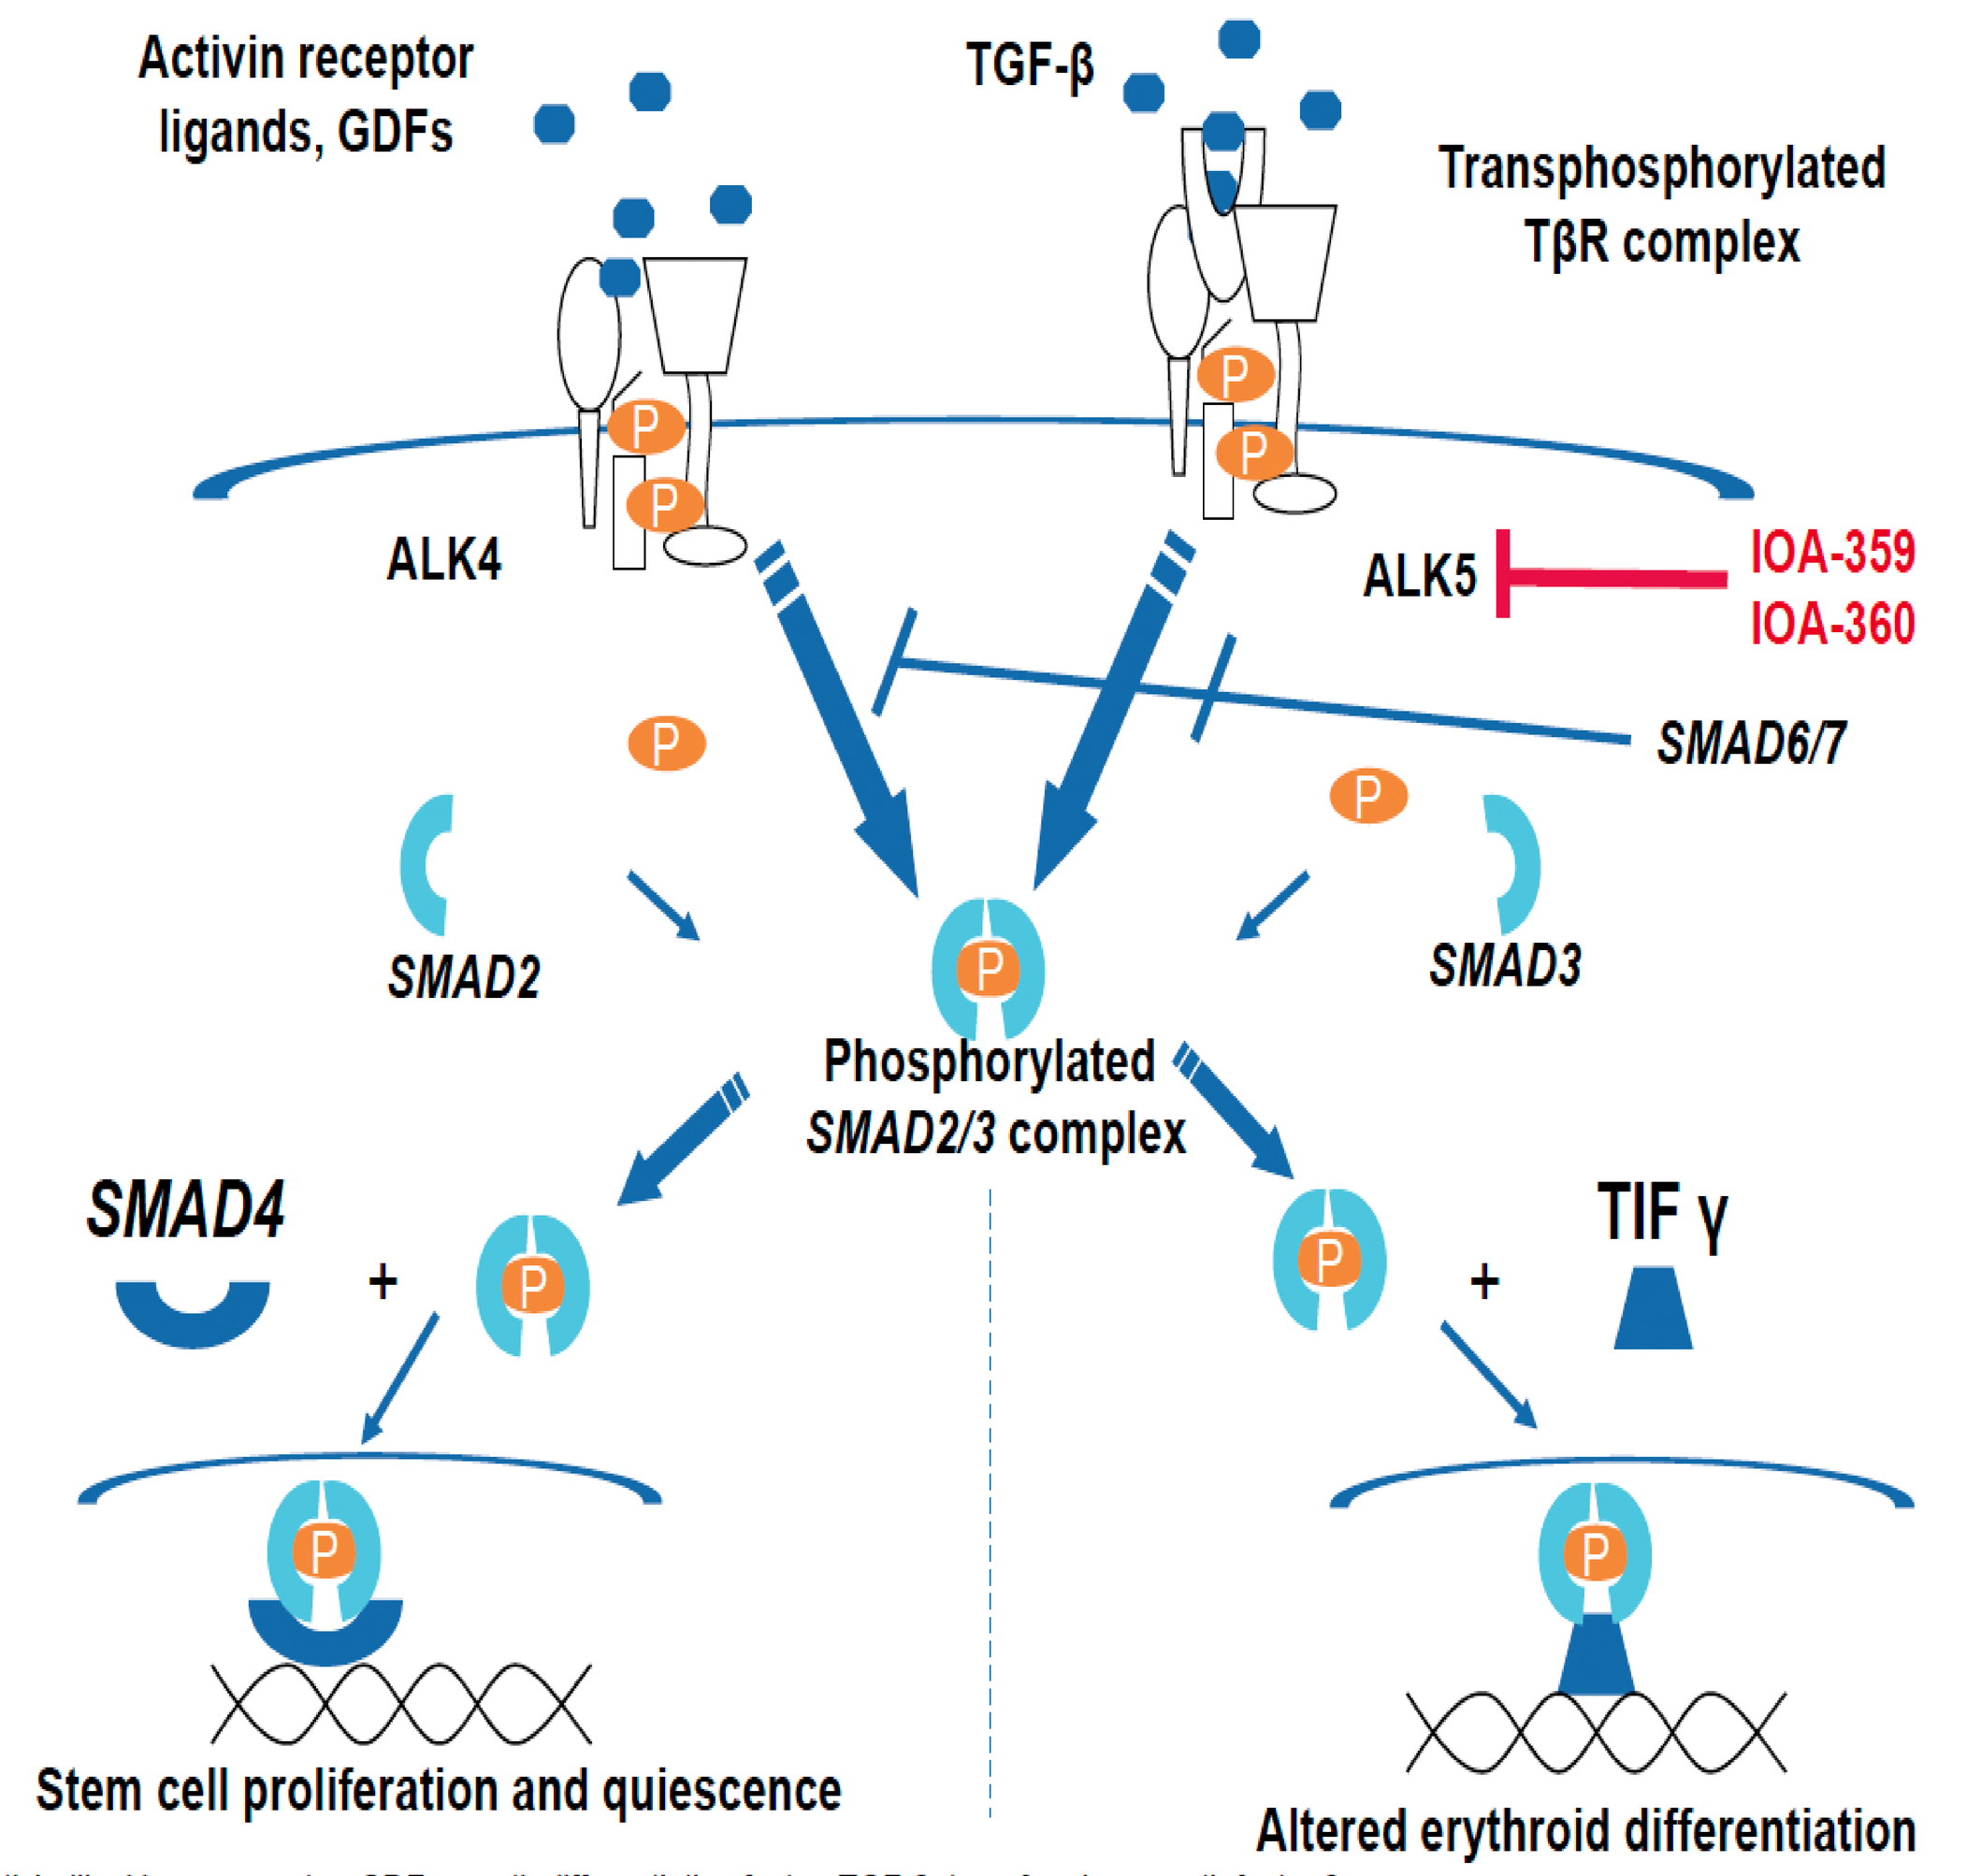
Activin receptor ligands, GDFs
TGF-β
Transphosphorylated TβR complex
ALK4
ALK5
IOA-359
IOA-360
SMAD6/7
SMAD2
SMAD3
Phosphorylated SMAD2/3 complex
SMAD4
TIF γ
Stem cell proliferation and quiescence
Altered erythroid differentiation

ALK, activin-like kinase receptor; GDF, growth differentiation factor; TGF-β, transforming growth factor β.

RESULTS

Fig 1

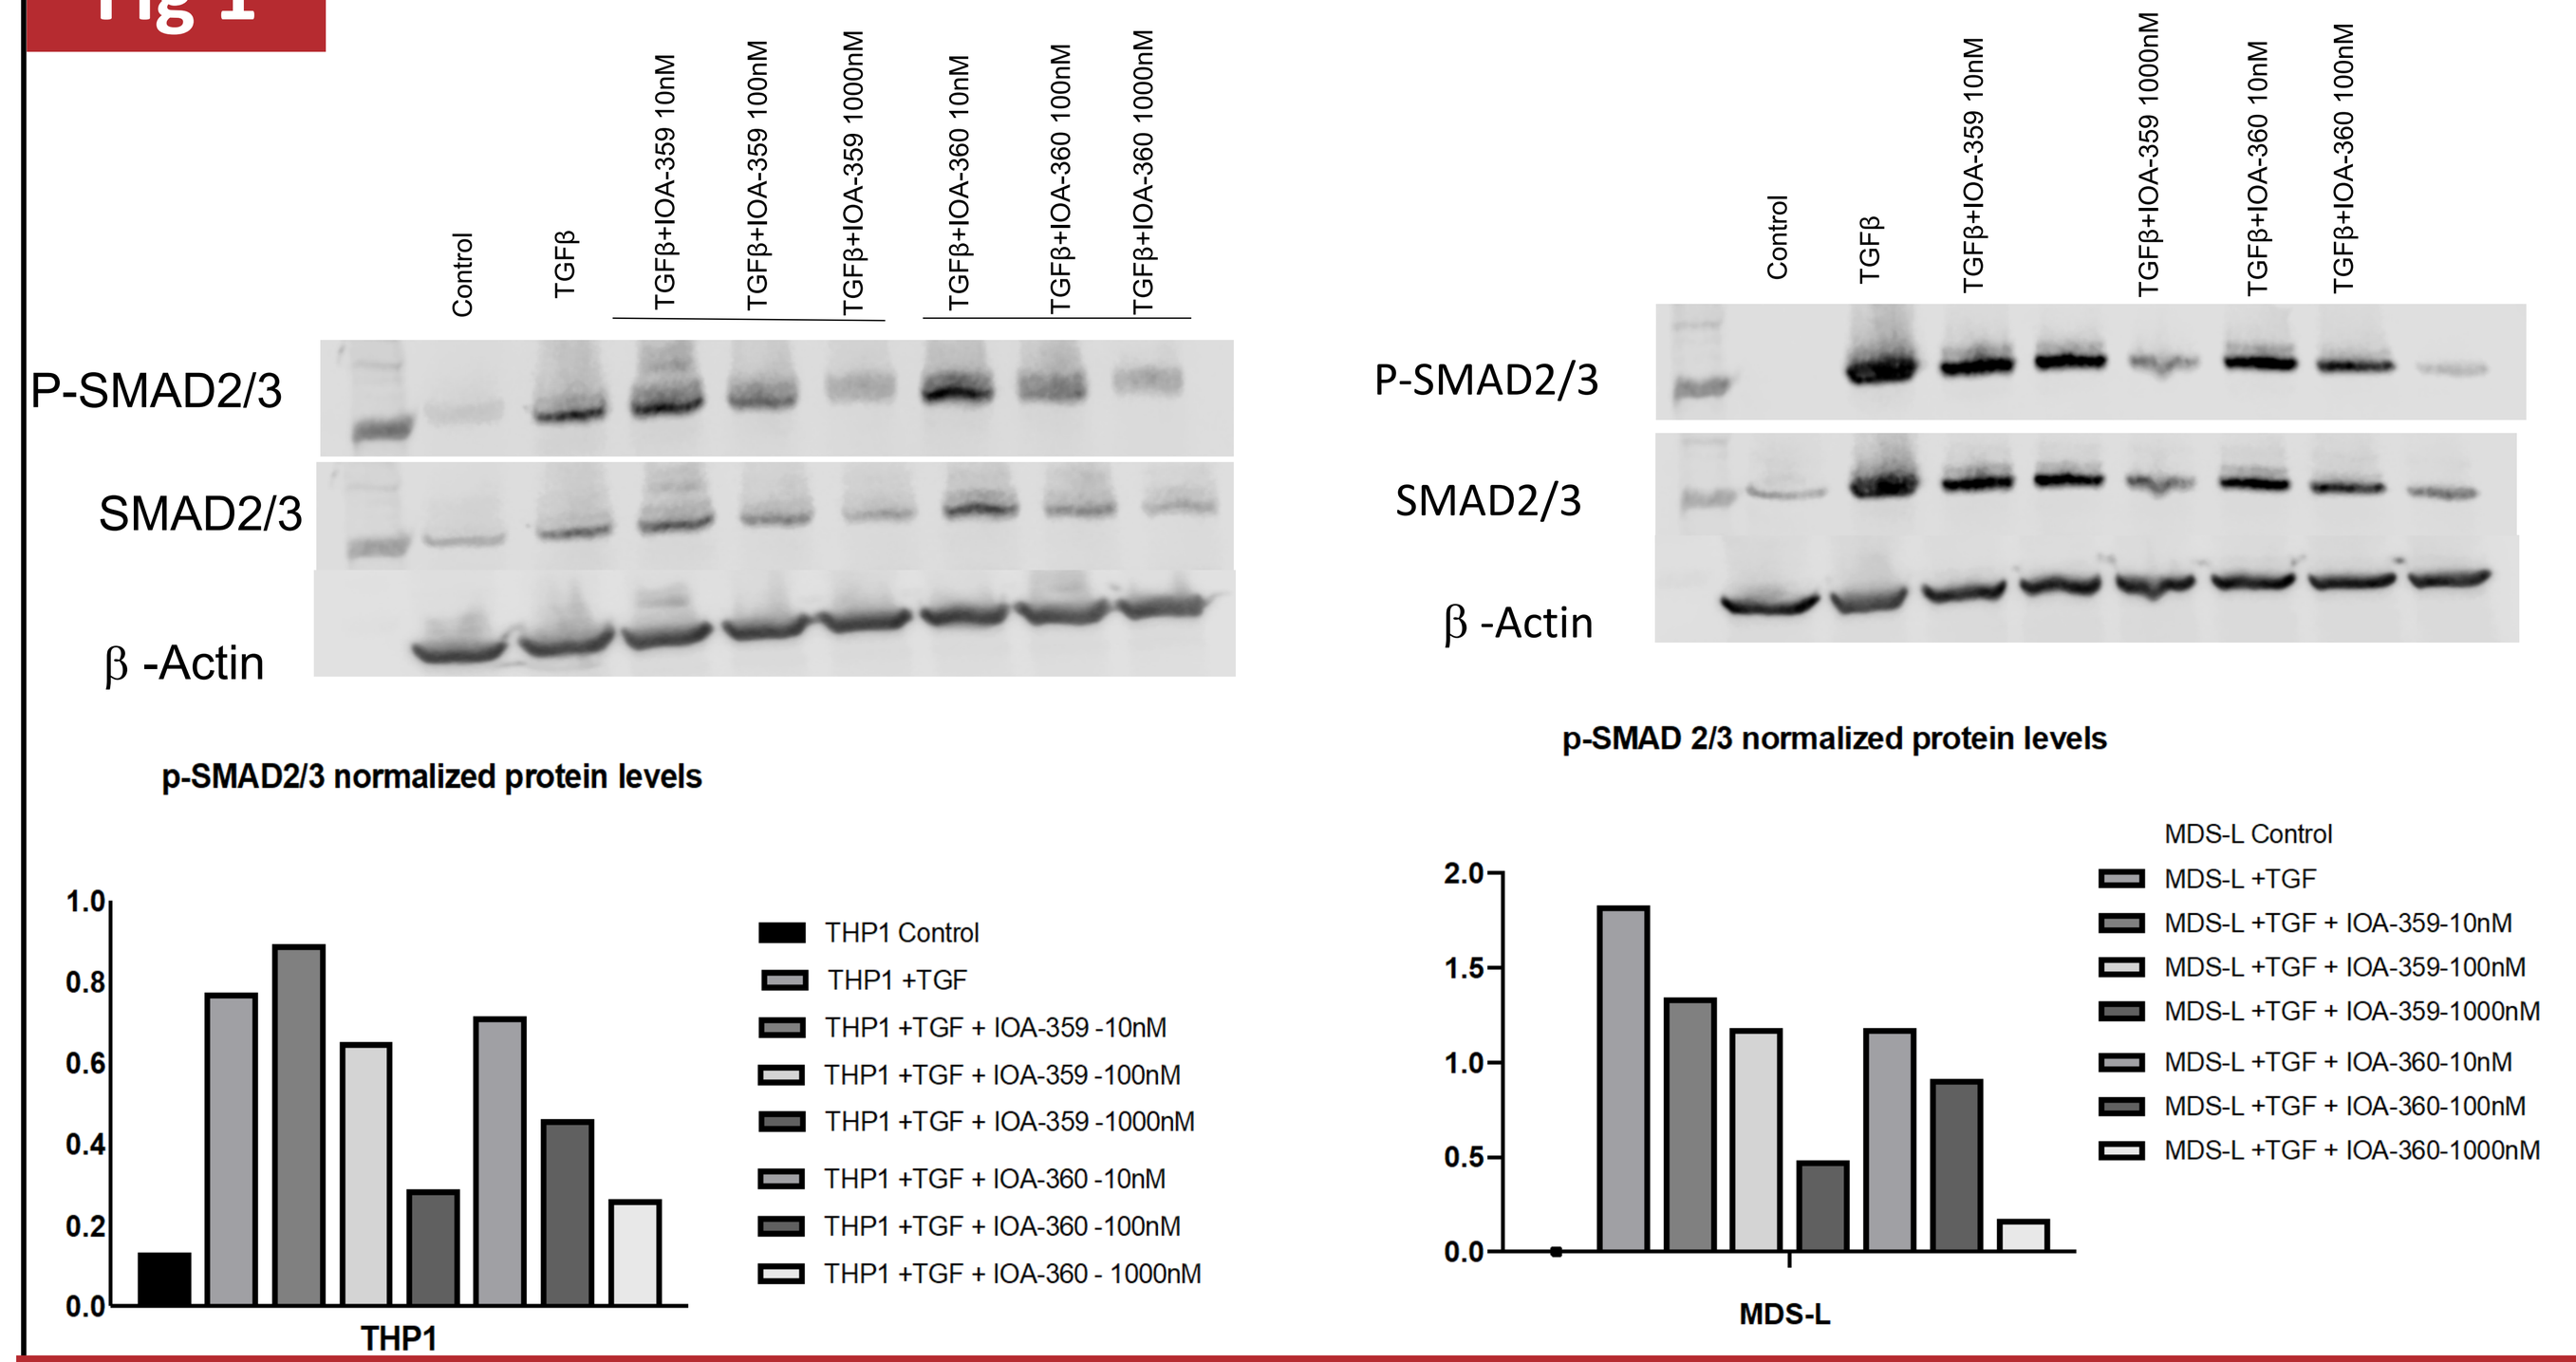
Control
TGFβ
TGFβ+IOA-359 10nM
TGFβ+IOA-359 100nM
TGFβ+IOA-359 1000nM
TGFβ+IOA-360 10nM
TGFβ+IOA-360 100nM
TGFβ+IOA-360 1000nM
P-SMAD2/3
SMAD2/3
β -Actin
p-SMAD2/3 normalized protein levels
THP1
THP1 Control
THP1 +TGF
THP1 +TGF + IOA-359 -10nM
THP1 +TGF + IOA-359 -100nM
THP1 +TGF + IOA-359 -1000nM
THP1 +TGF + IOA-360 -10nM
THP1 +TGF + IOA-360 -100nM
THP1 +TGF + IOA-360 - 1000nM
p-SMAD 2/3 normalized protein levels
MDS-L
MDS-L Control
MDS-L +TGF
MDS-L +TGF + IOA-359-10nM
MDS-L +TGF + IOA-359-100nM
MDS-L +TGF + IOA-359-1000nM
MDS-L +TGF + IOA-360-10nM
MDS-L +TGF + IOA-360-100nM
MDS-L +TGF + IOA-360-1000nM

Fig 1: IOA-359 and IOA-360 can inhibit SMAD2/3 phosphorylation in THP1 and MDS-L cells upon TGF stimulation: Immunoblotting of phospho-SMAD2/3 and SMAD2/3 in (A) THP1 and (B) MDS-L cells that were treated with IOA-359 and IOA-360, respectively, at multiple concentrations (10nM, 100nM and 1000nM), followed by 30 minutes stimulation of TGF-β (20 ng/mL). Quantification of anti-psmad2/3, normalized to β-actin, by densitometry analysis, is shown as bar graphs.

Fig 2

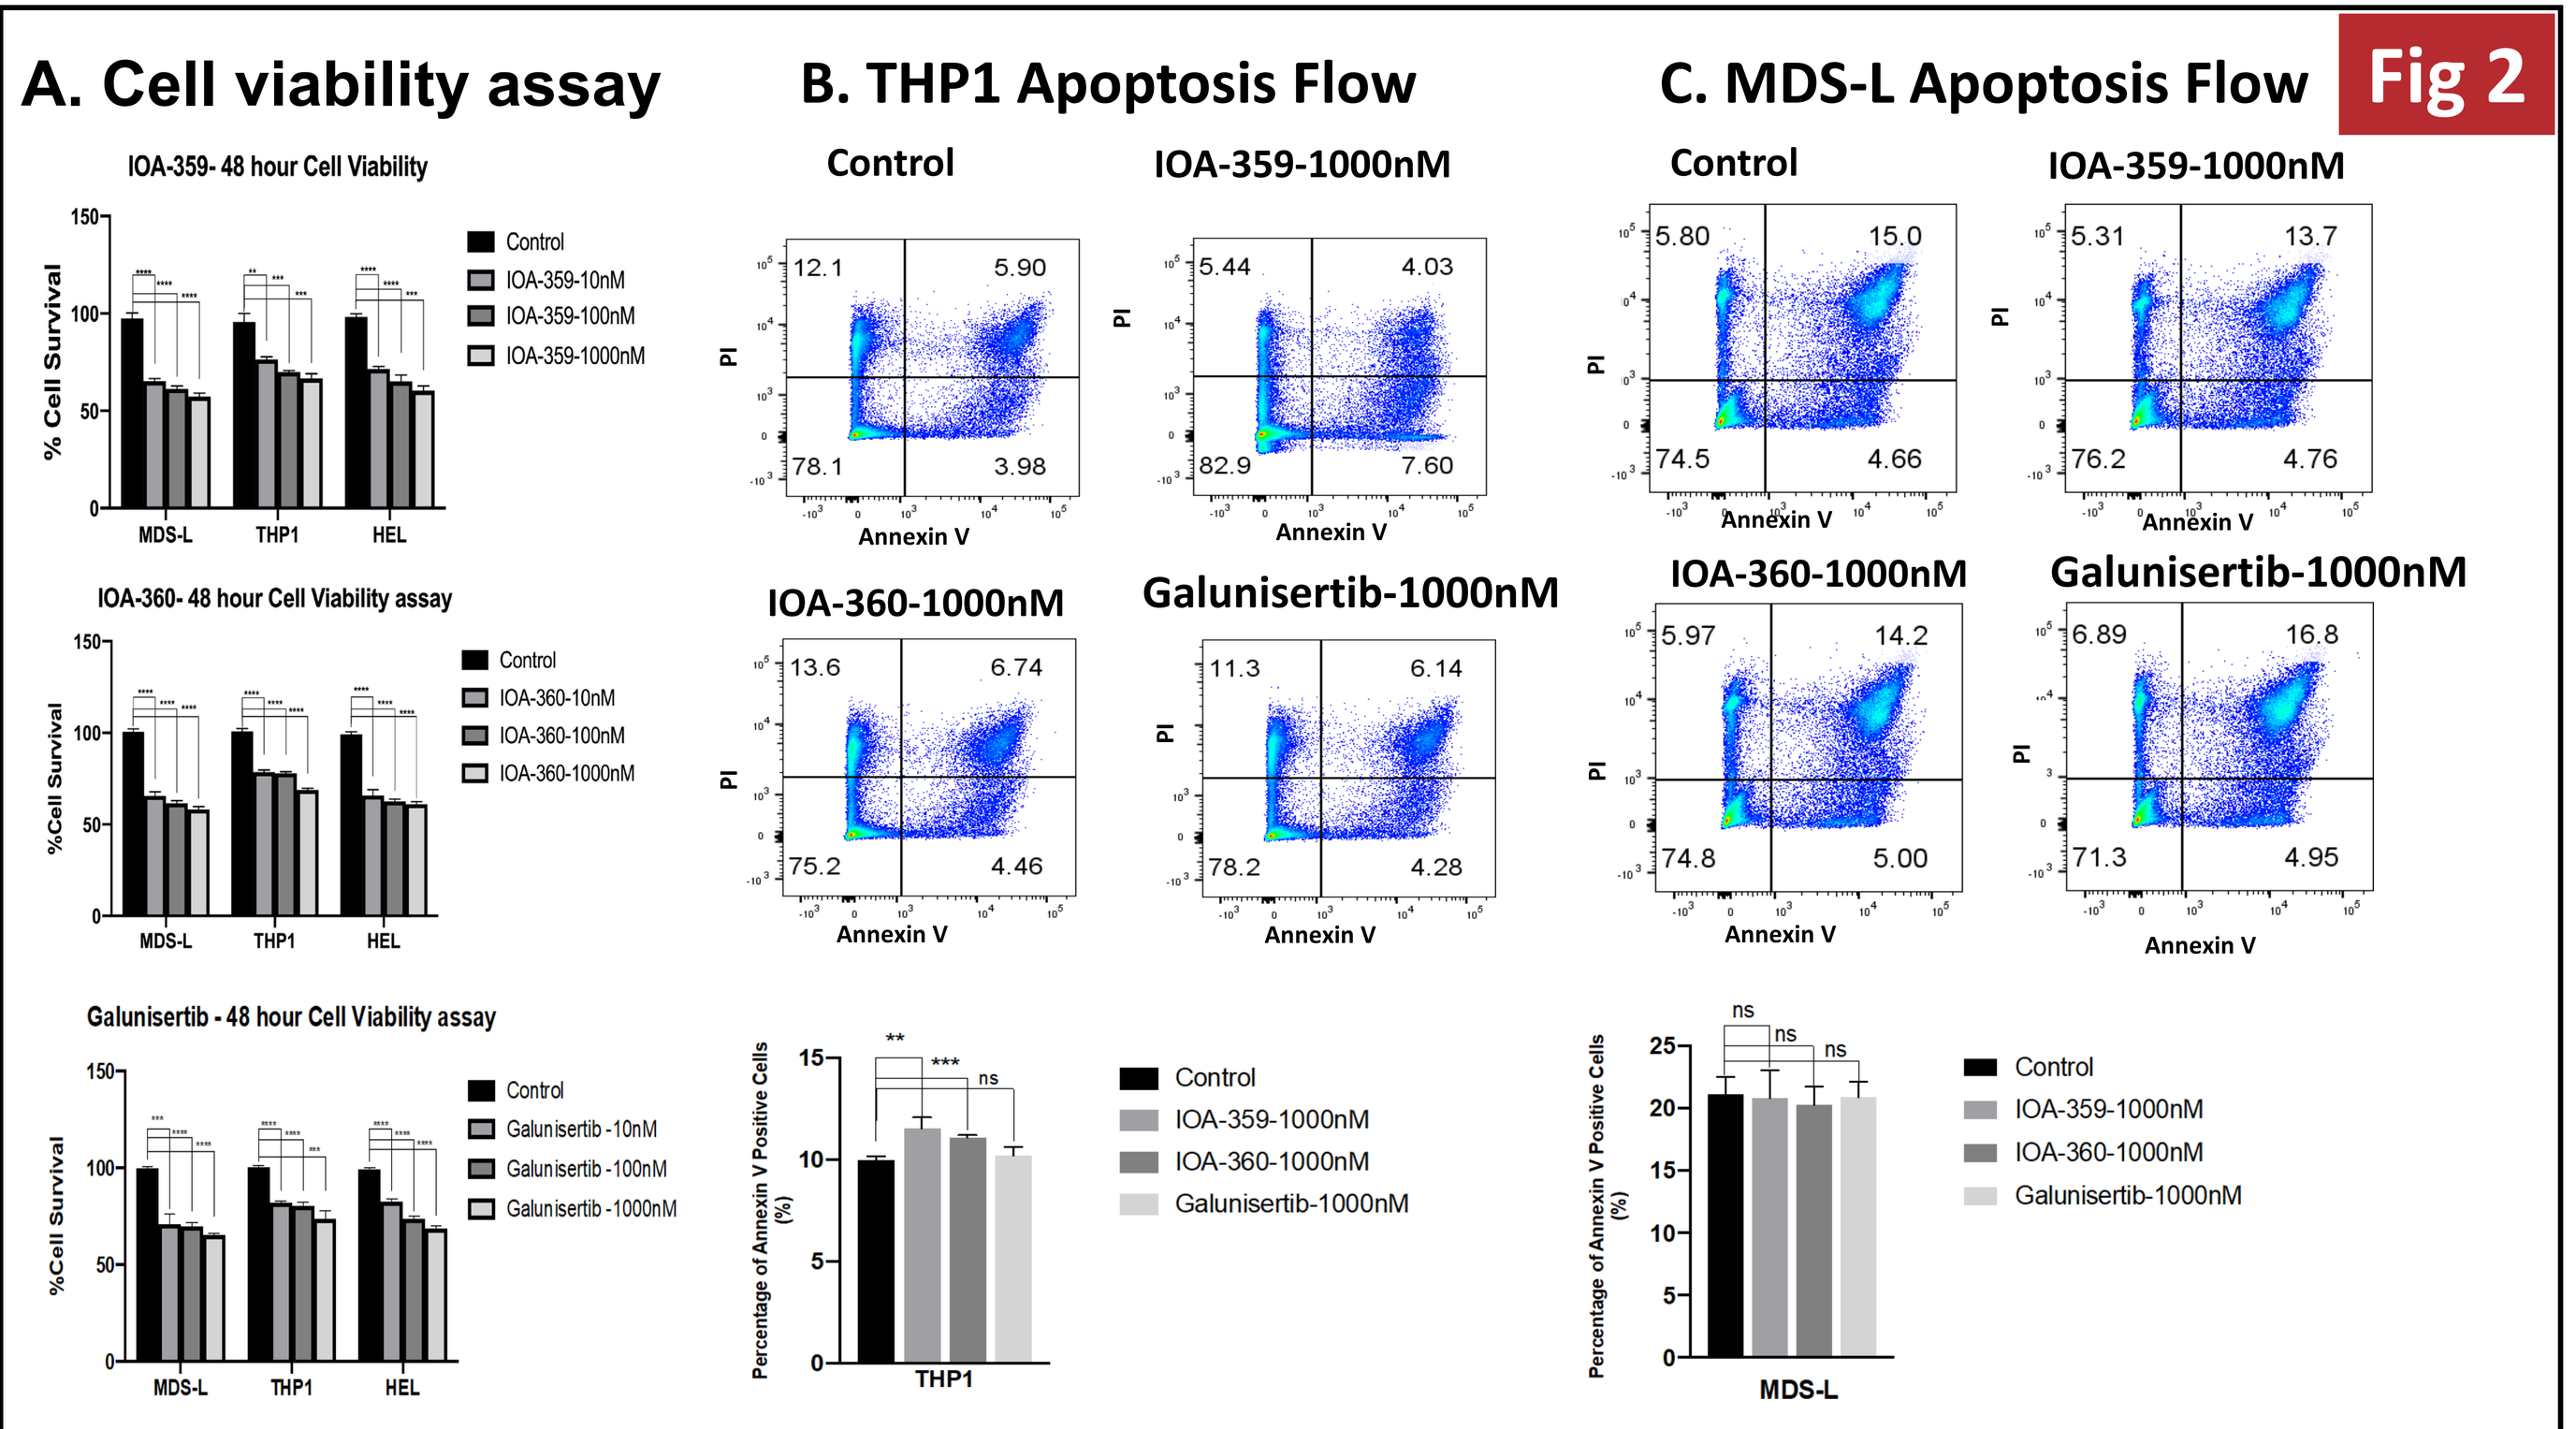
A. Cell viability assay
B. THP1 Apoptosis Flow
C. MDS-L Apoptosis Flow
IOA-359- 48 hour Cell Viability
IOA-360- 48 hour Cell Viability assay
Galunisertib - 48 hour Cell Viability assay
% Cell Survival
MDS-L
THP1
HEL
Control
IOA-359-1000nM
IOA-360-1000nM
Galunisertib-1000nM
Annexin V
PI
Percentage of Annexin V Positive Cells (%)

Fig 2: IOA-359 and IOA-360 ALK5 inhibitors do not increase leukemic cell proliferation – (A) MDS-L, THP1 and HEL cell lines were treated with IOA-359, IOA-360 and Galunisertib at multiple concentrations (10nM, 100nM and 1000nM), respectively, for 48 hours. Cell viability was assessed at 48 hours with Cell-Titer Blue assay. **(B)** THP1 and **(C)** MDS-L cells treated with IOA-359, IOA-360 and Galunisertib, (1000 nM) respectively, for 72 hours. Cells were harvested and stained with Annexin V and PI and analyzed by FACS. Representative bar plots are also shown.

Fig 3

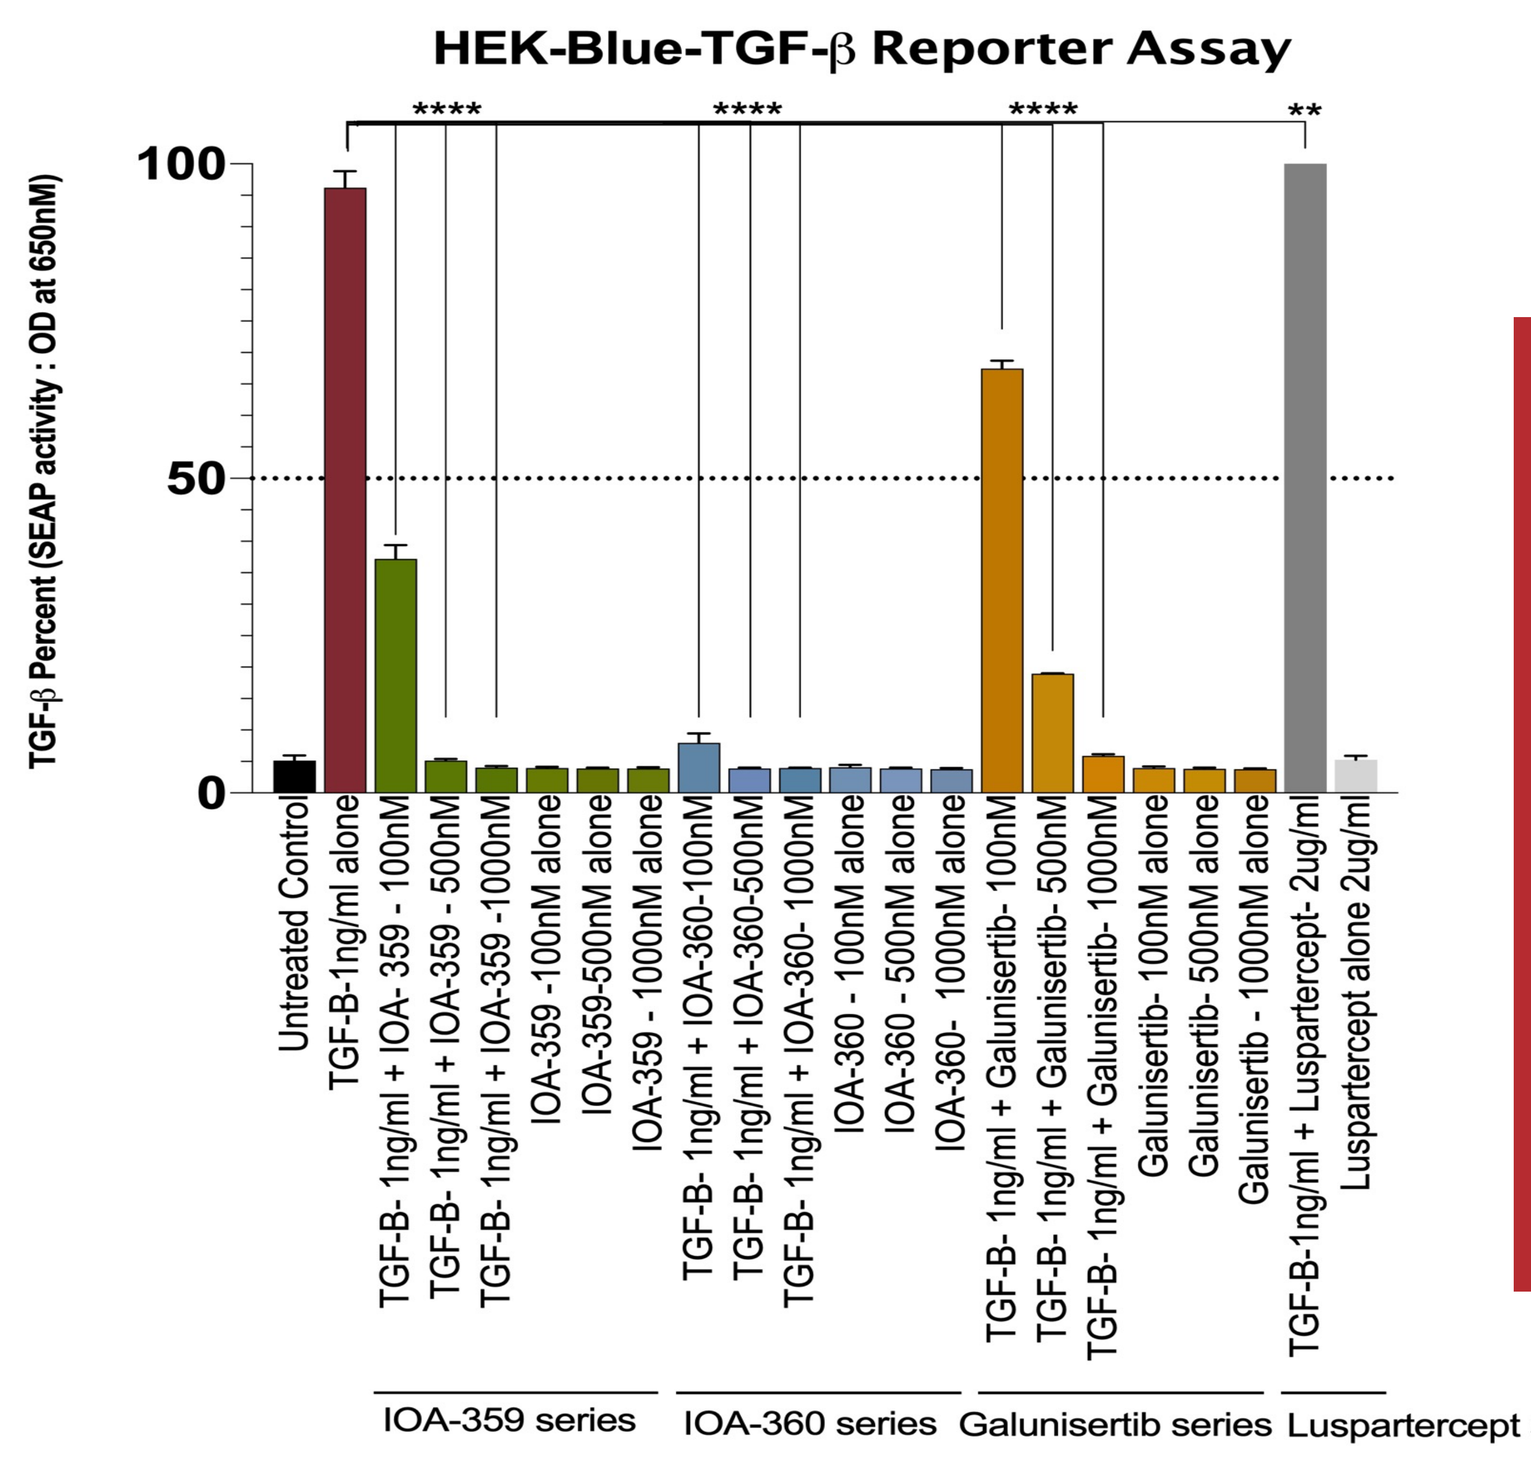
HEK-Blue-TGF-β Reporter Assay
TGF-β Percent (SEAP activity : OD at 650nM)
Untreated Control
TGF-B-1ng/ml alone
TGF-B- 1ng/ml + IOA-359 - 100nM
TGF-B- 1ng/ml + IOA-359 - 500nM
TGF-B- 1ng/ml + IOA-359 -1000nM
IOA-359 -100nM alone
IOA-359-500nM alone
IOA-359 - 1000nM alone
TGF-B- 1ng/ml + IOA-360-100nM
TGF-B- 1ng/ml + IOA-360-500nM
TGF-B- 1ng/ml + IOA-360- 1000nM
IOA-360 - 100nM alone
IOA-360 - 500nM alone
IOA-360- 1000nM alone
TGF-B- 1ng/ml + Galunisertib- 100nM
TGF-B- 1ng/ml + Galunisertib- 500nM
TGF-B- 1ng/ml + Galunisertib- 1000nM
Galunisertib- 100nM alone
Galunisertib- 500nM alone
Galunisertib - 1000nM alone
TGF-B-1ng/ml + Luspartecept- 2ug/ml
Luspartecept alone 2ug/ml
IOA-359 series
IOA-360 series
Galunisertib series
Luspartercept series

Fig 3 - IOA-359 and IOA-360 inhibit TGF-β mediated gene expression. (A) HEK-293-Blue-TGF-β reporter cell line was used to detect TGF-β ligand activity and monitor the activation of the TGF-β /SMAD pathway. HEK-293-Blue-TGF-β reporter cells were treated with IOA-360, IOA-359 and Galunisertib for 24 hours at multiple concentrations (100nM, 500nM and 1000nM), respectively, and co-stimulated with TGF-β ligand (1 ng/mL). HEK-293-Blue-TGF-β reporter cells treated with IOA-360, IOA-359 and Galunisertib, without TGF-β ligand stimulation, were used as control. HEK-293-Blue-TGF-β reporter cells treated with Luspartecept (2 ug/mL) with or without TGF-β ligand (1 ng/mL) stimulation were used as control.

Fig 4

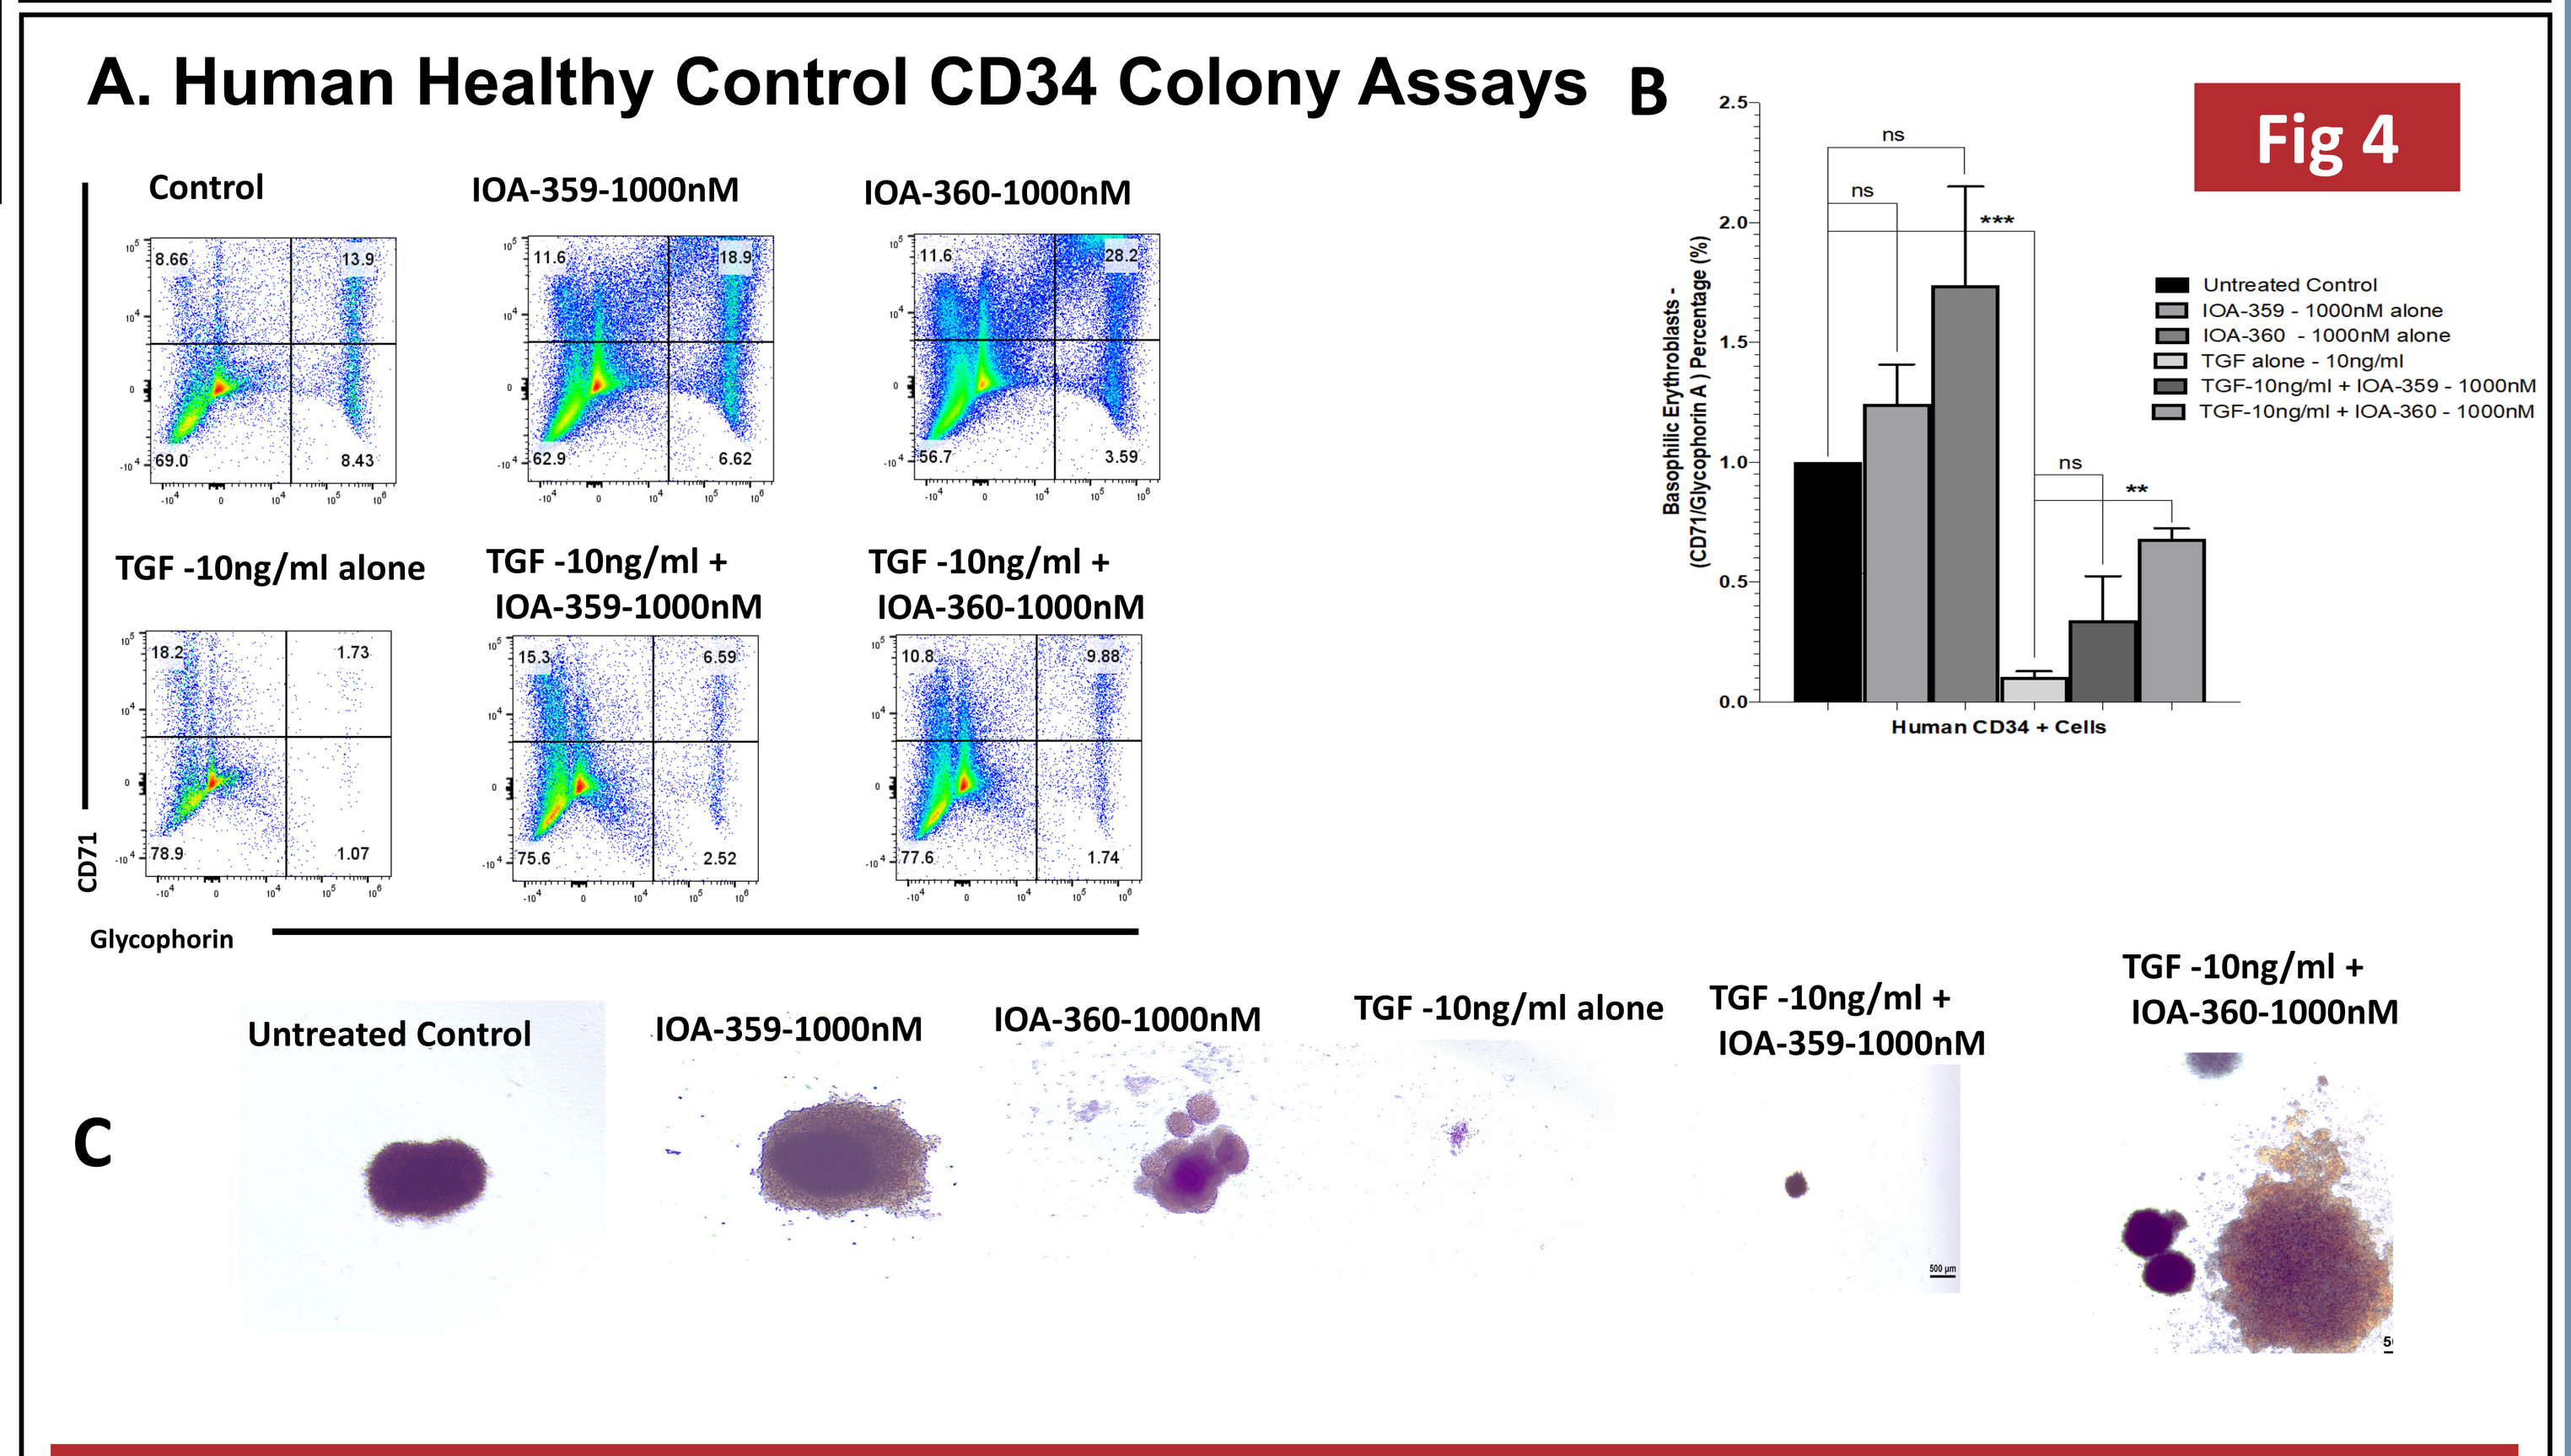
A. Human Healthy Control CD34 Colony Assays
B
C
Control
IOA-359-1000nM
IOA-360-1000nM
TGF -10ng/ml alone
TGF -10ng/ml + IOA-359-1000nM
TGF -10ng/ml + IOA-360-1000nM
CD71
Glycophorin
Untreated Control
Basophilic Erythroblasts - (CD71/Glycophorin A) Percentage (%)
Human CD34 + Cells
Untreated Control
IOA-359 - 1000nM alone
IOA-360 - 1000nM alone
TGF alone - 10ng/ml
TGF-10ng/ml + IOA-359 - 1000nM
TGF-10ng/ml + IOA-360 - 1000nM

Fig 4: IOA-359 and IOA-360 can reverse growth inhibitory effects of TGF-β in healthy human CD34 cells – (A) Healthy Human CD34+ cells were grown on methylcellulose to determine the effects of drugs IOA-359 and IOA-360 respectively and /or TGF-β stimulation. Cells were harvested after 14 days for erythroid differentiation by FACS analysis. TGF-β inhibited erythroid differentiation. An increase in mature erythroid and basophilic progenitors was observed in TGF-β stimulated cells treated with IOA-359 and IOA-360.**(B)** A representative flow bar plot shows percentage of Basophilic erythroblasts (CD71/Glycophorin A) after 14 days. (C) A representative picture of colonies treated with drugs TGF-β and IOA-359 and IOA-360 were larger in size when compared to TGF-β alone.

Fig 5A

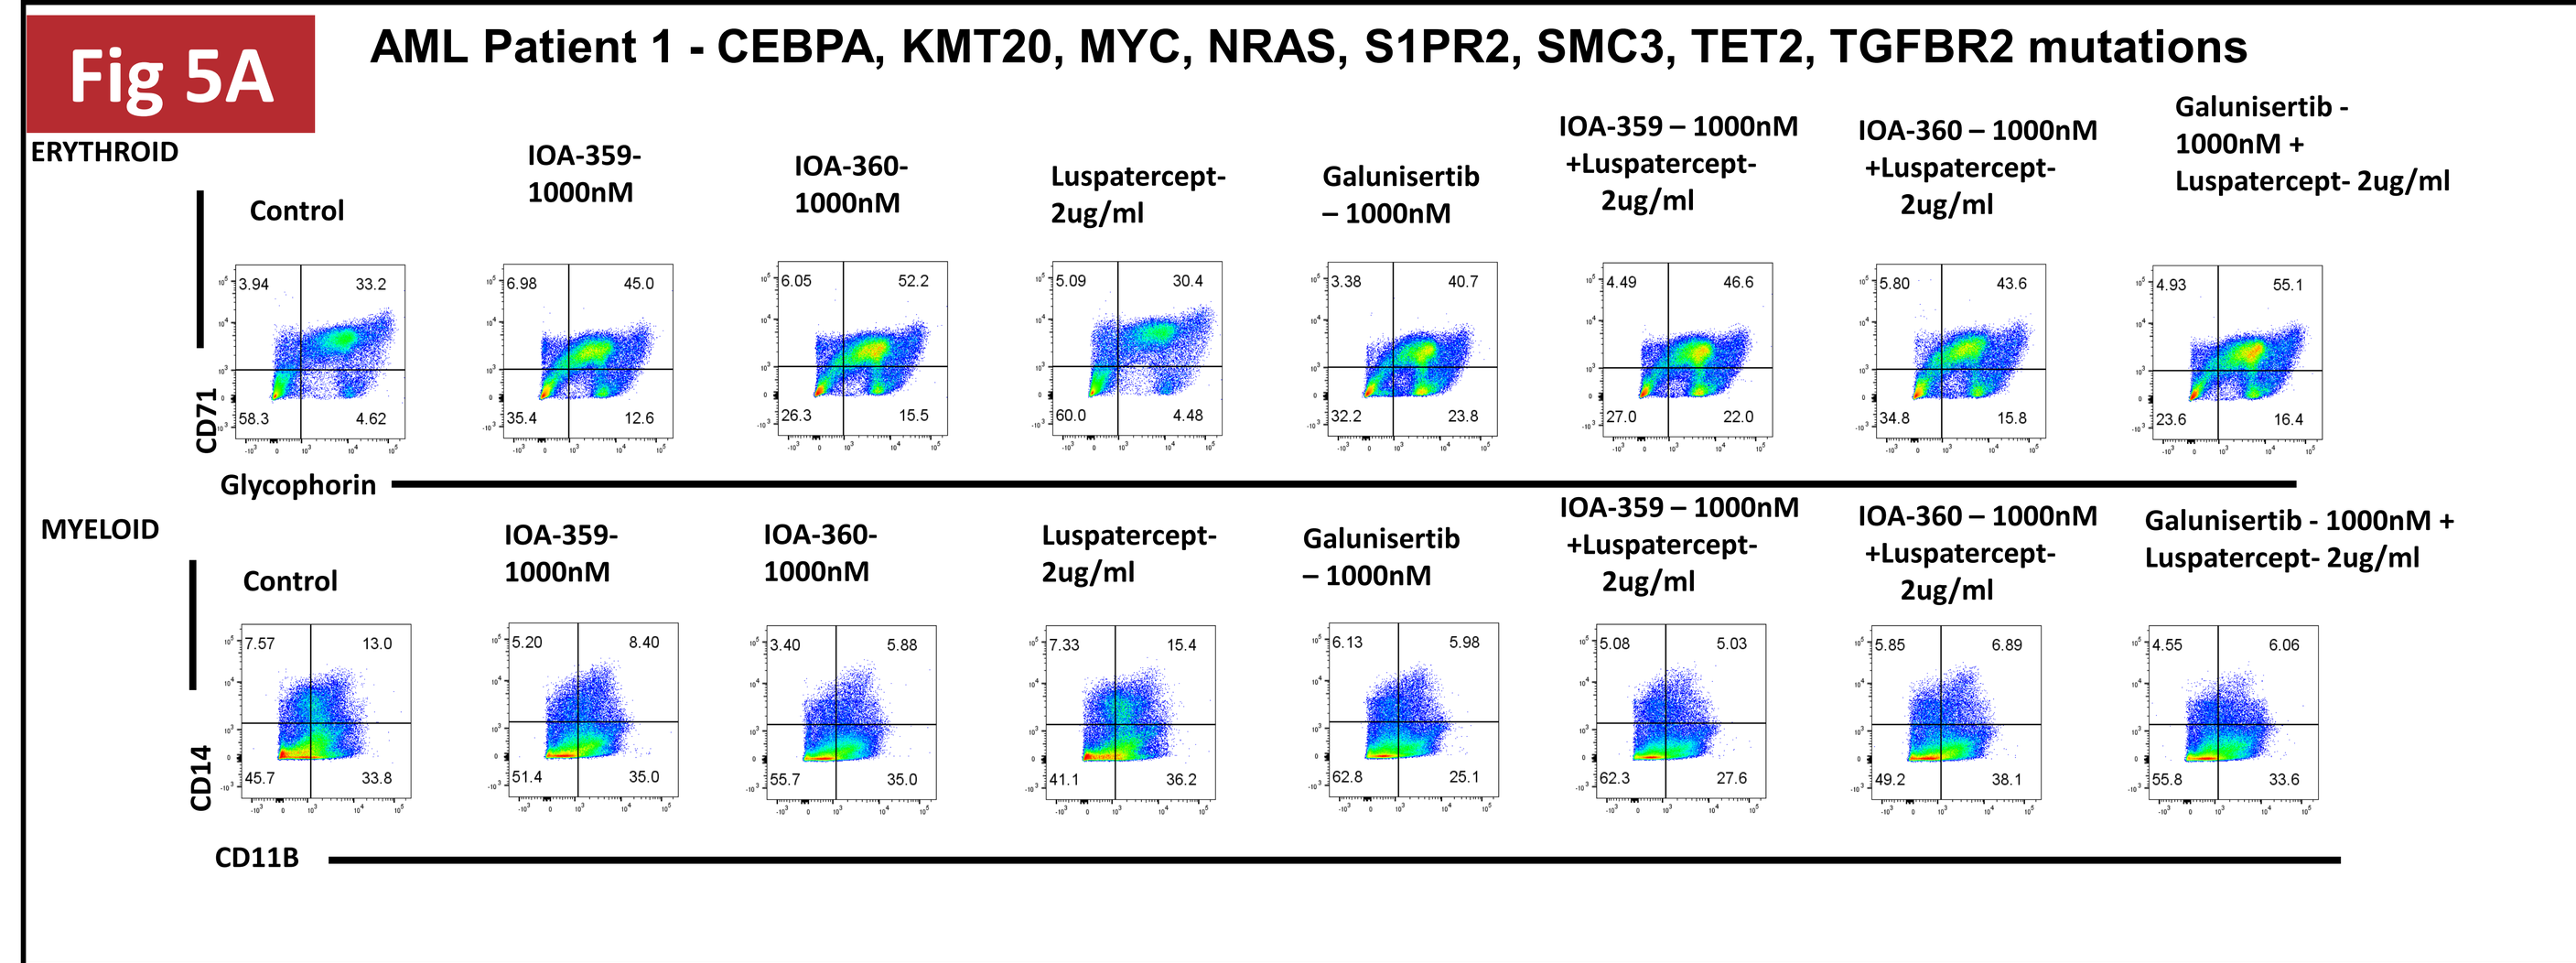
AML Patient 1 - CEBPA, KMT20, MYC, NRAS, S1PR2, SMC3, TET2, TGFBR2 mutations
ERYTHROID
MYELOID
Control
IOA-359-1000nM
IOA-360-1000nM
Luspatercept-2ug/ml
Galunisertib – 1000nM
IOA-359 – 1000nM +Luspatercept-2ug/ml
IOA-360 – 1000nM +Luspatercept-2ug/ml
Galunisertib - 1000nM + Luspatercept- 2ug/ml
CD71
Glycophorin
CD14
CD11B

Fig 5B

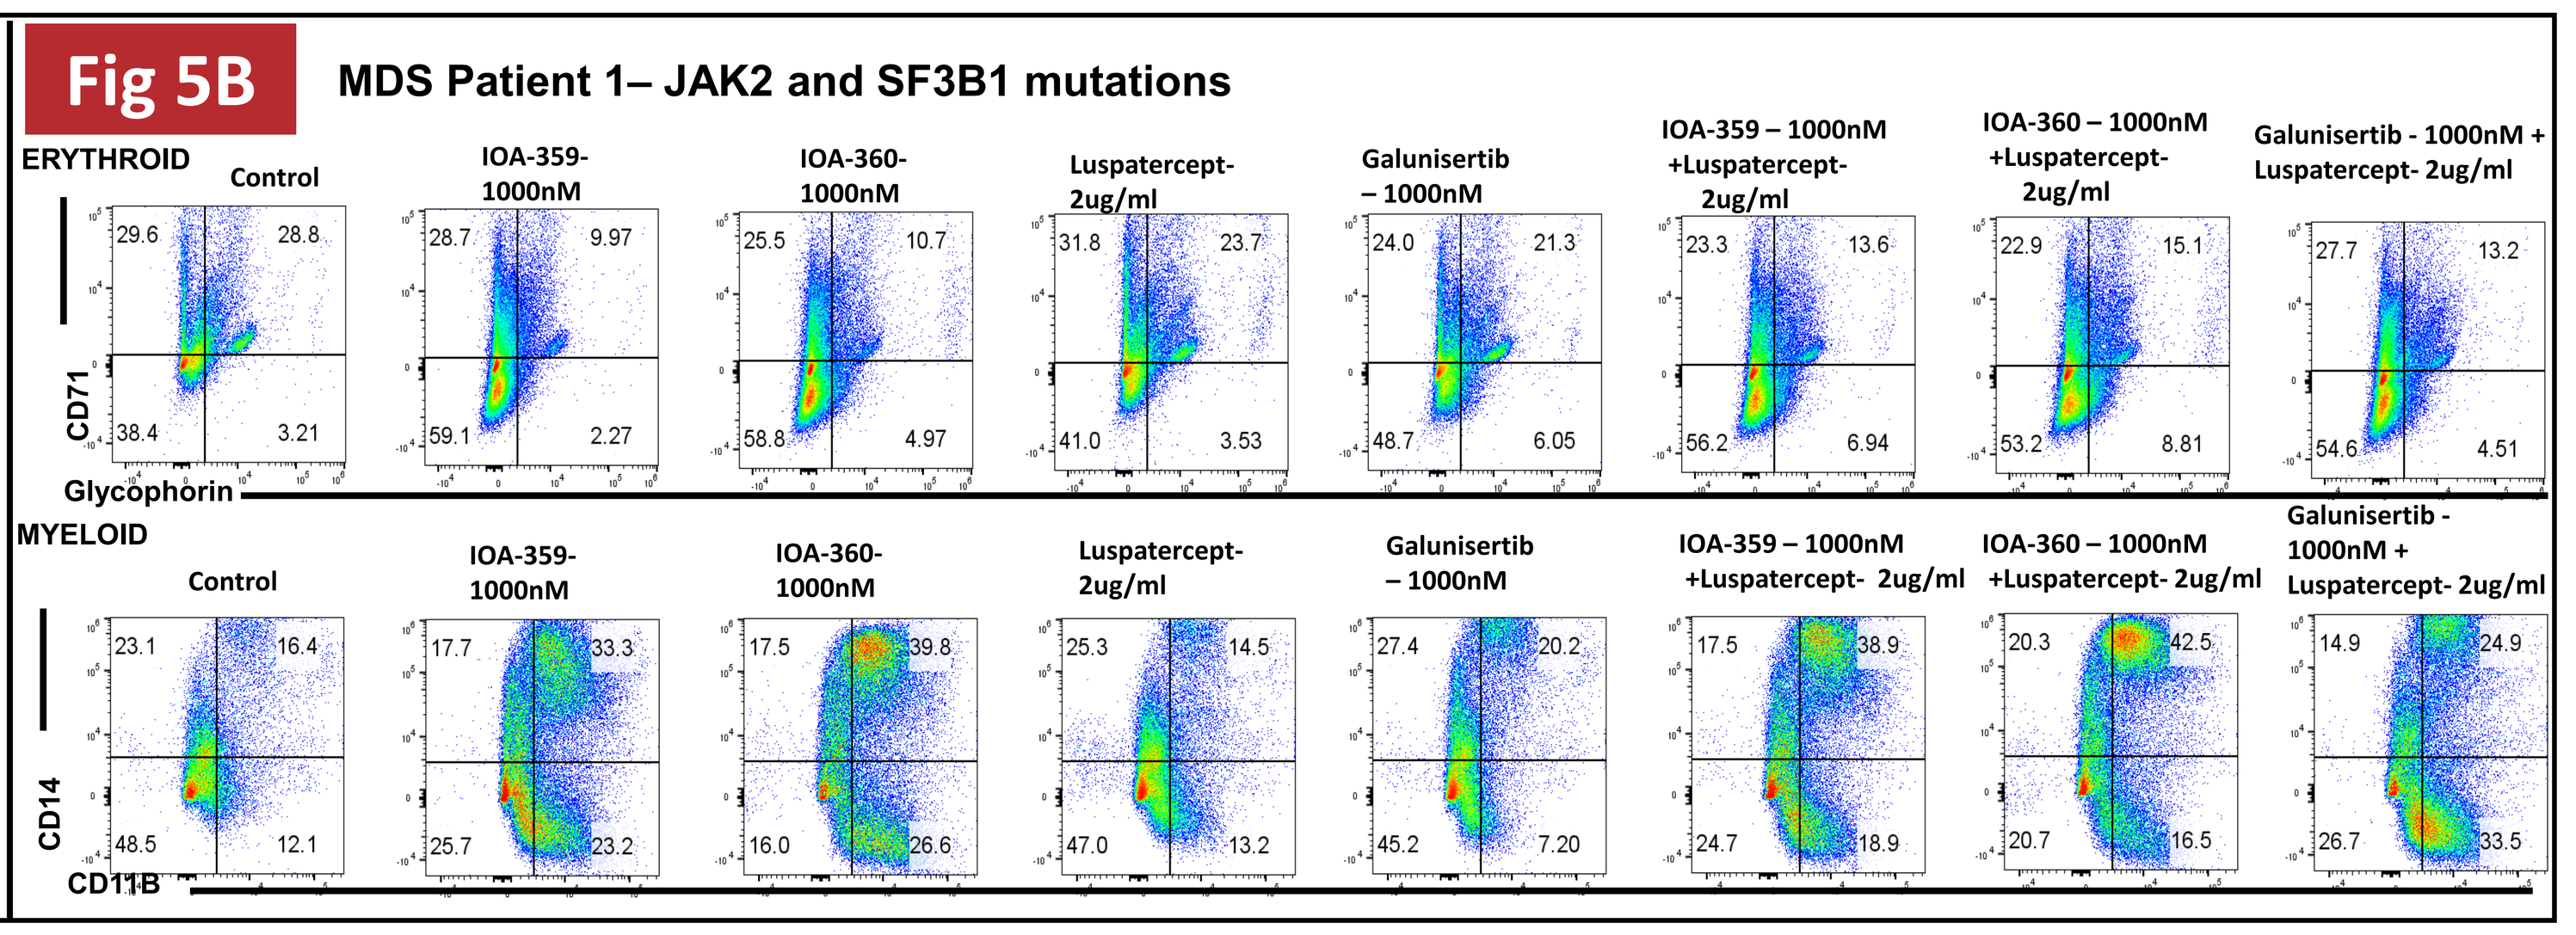
MDS Patient 1– JAK2 and SF3B1 mutations
ERYTHROID
MYELOID
Control
IOA-359-1000nM
IOA-360-1000nM
Luspatercept-2ug/ml
Galunisertib – 1000nM
IOA-359 – 1000nM +Luspatercept-2ug/ml
IOA-360 – 1000nM +Luspatercept-2ug/ml
Galunisertib - 1000nM + Luspatercept- 2ug/ml
CD71
Glycophorin
CD14
CD11B

Fig 5C

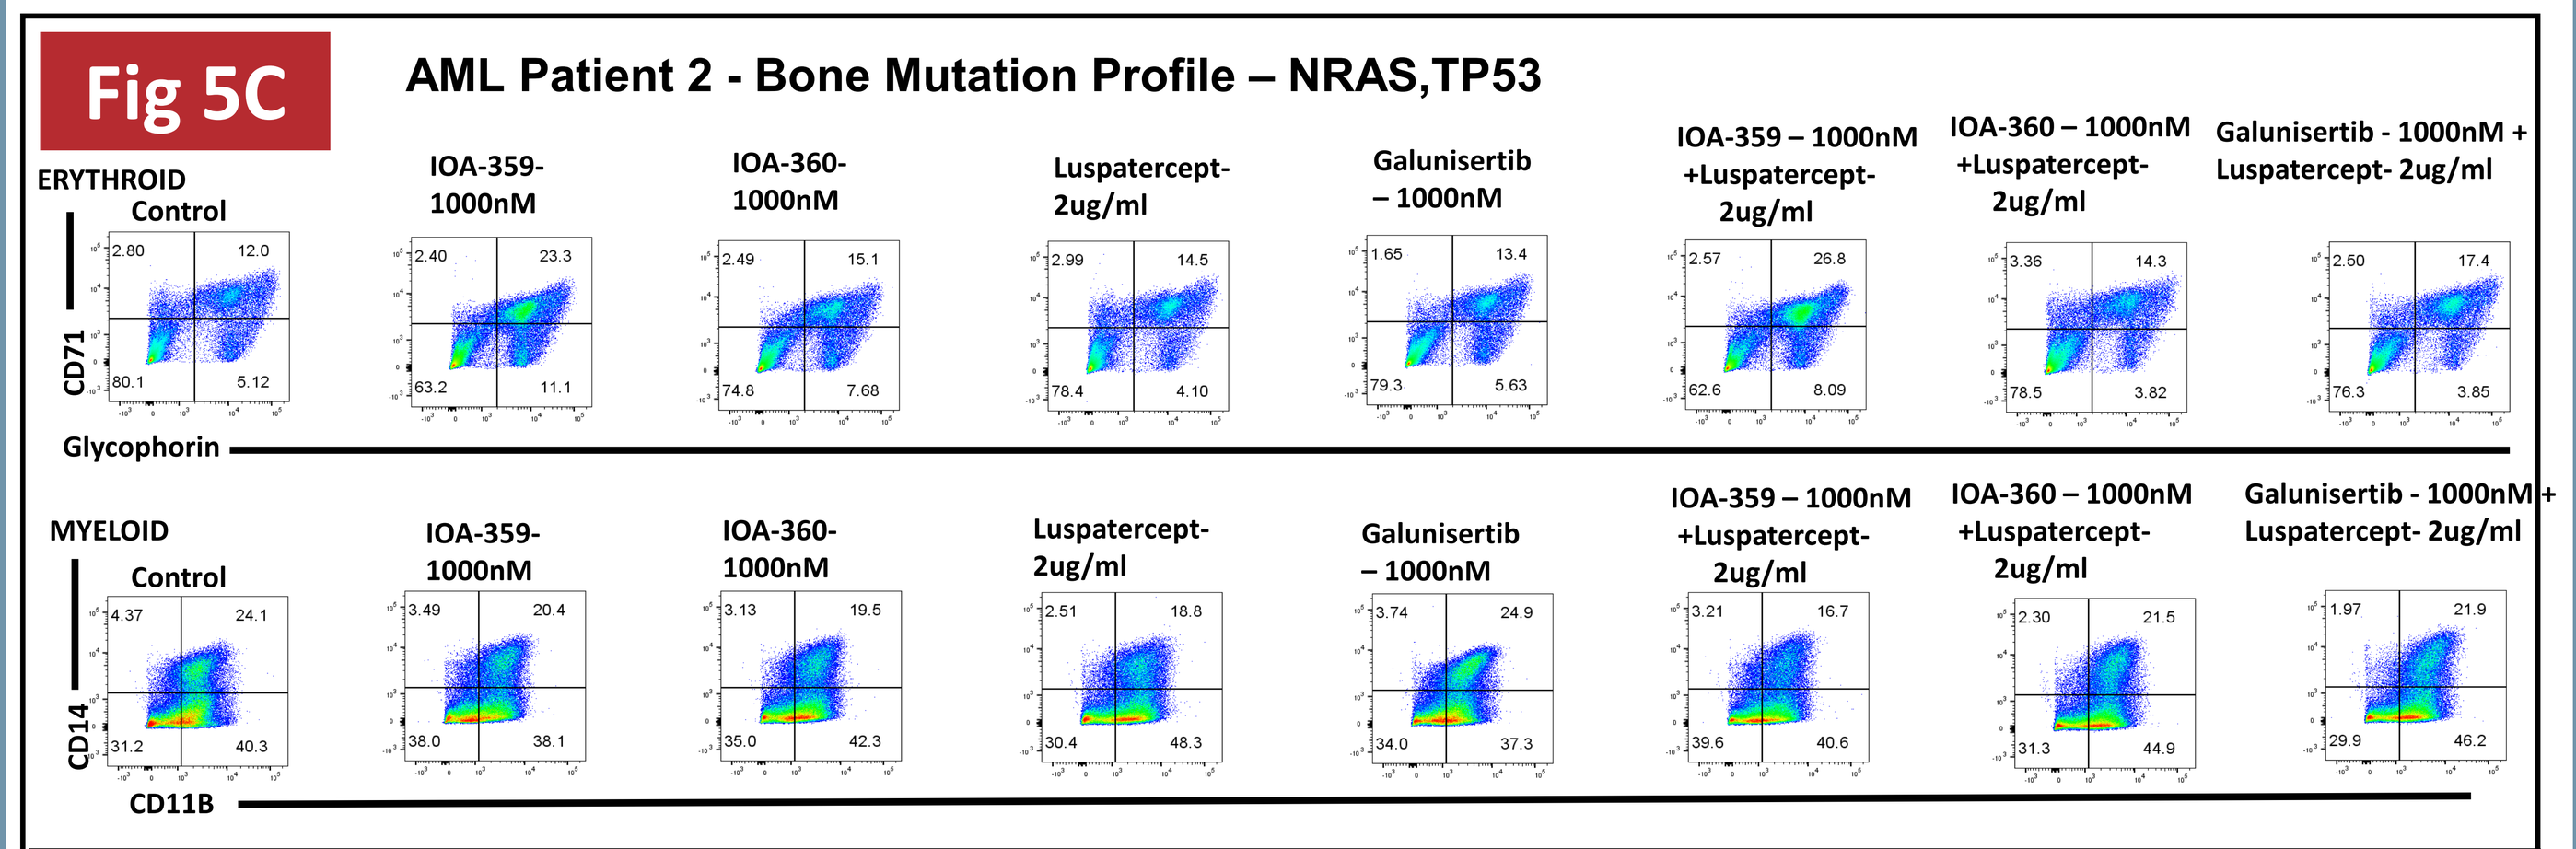
AML Patient 2 - Bone Mutation Profile – NRAS,TP53
ERYTHROID
MYELOID
Control
IOA-359-1000nM
IOA-360-1000nM
Luspatercept-2ug/ml
Galunisertib – 1000nM
IOA-359 – 1000nM +Luspatercept-2ug/ml
IOA-360 – 1000nM +Luspatercept-2ug/ml
Galunisertib - 1000nM + Luspatercept- 2ug/ml
CD71
Glycophorin
CD14
CD11B

Fig 5D

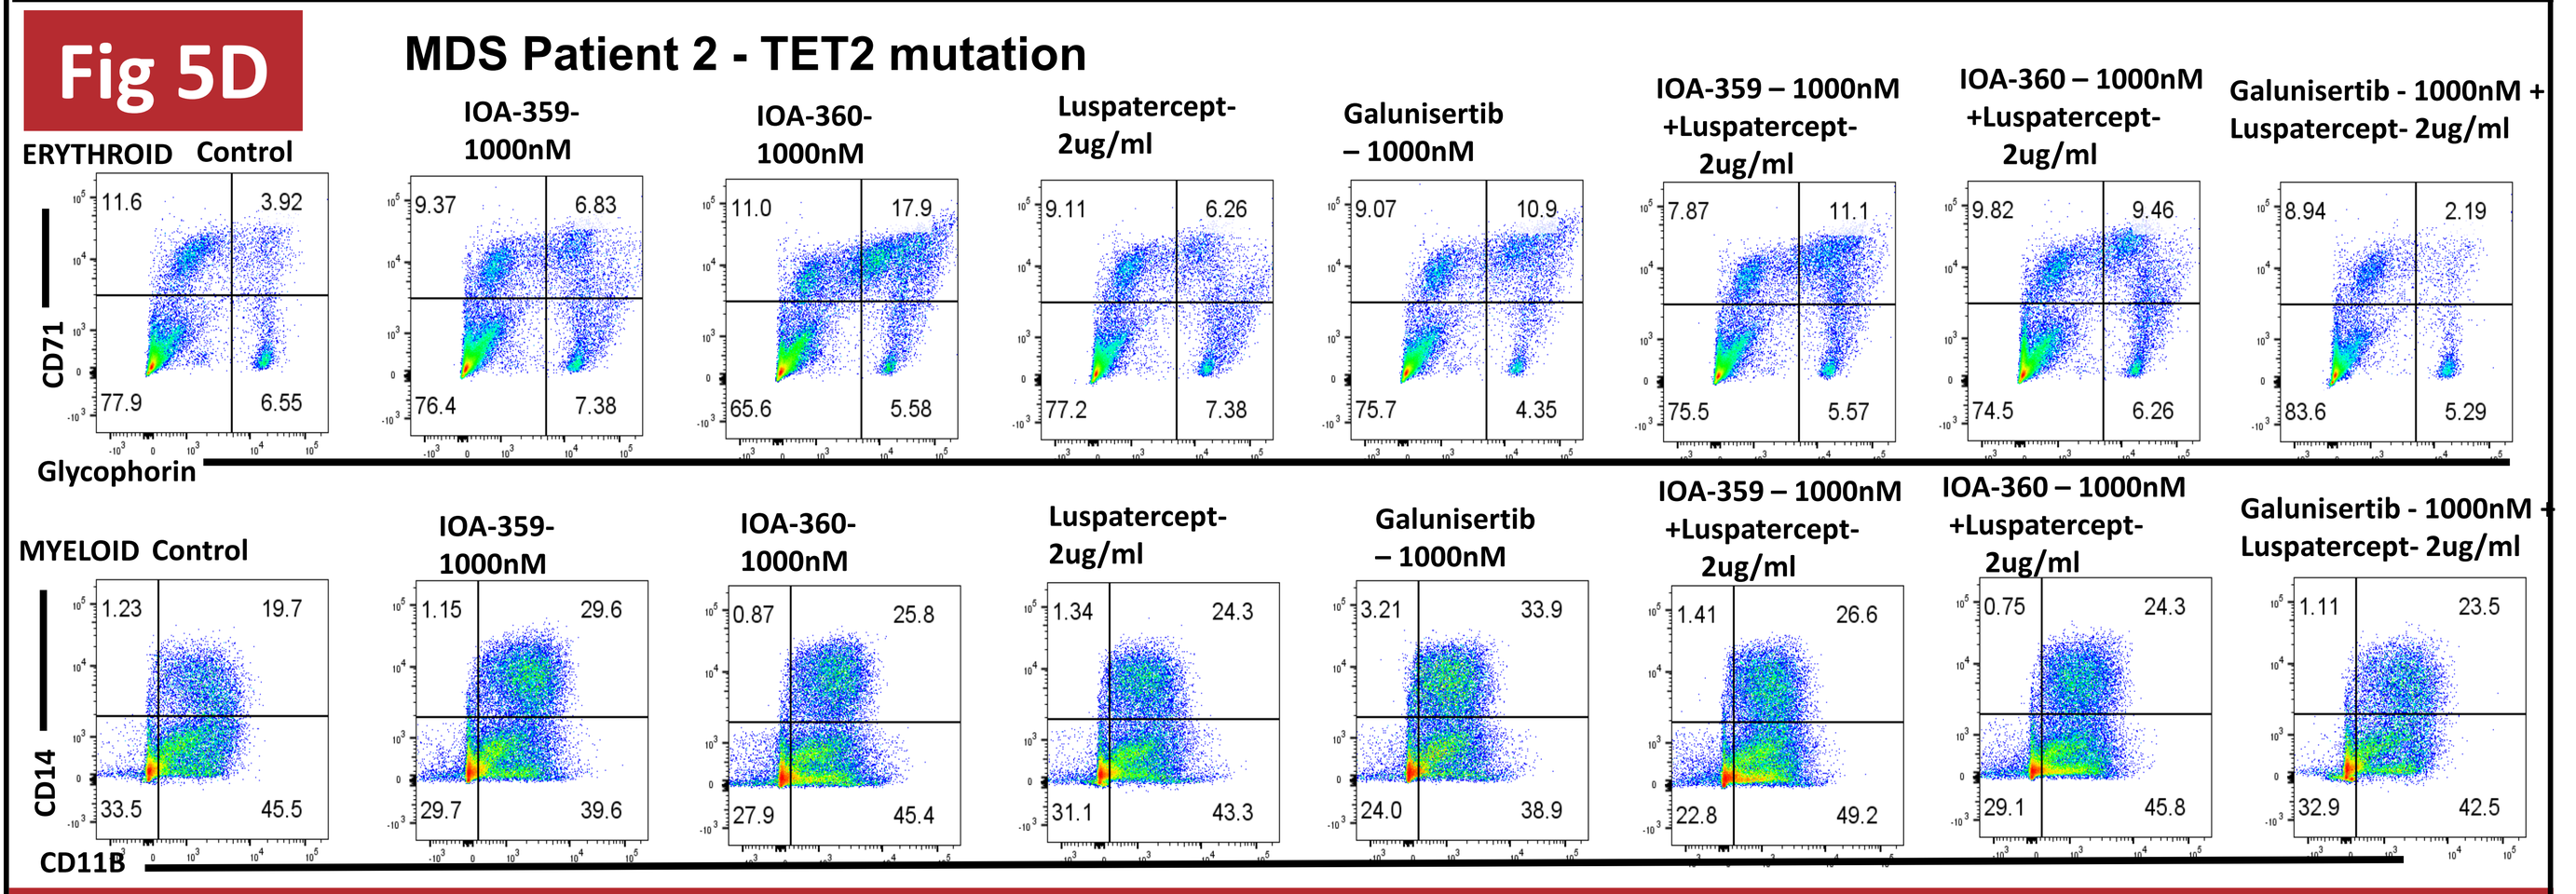
MDS Patient 2 - TET2 mutation
ERYTHROID
MYELOID
Control
IOA-359-1000nM
IOA-360-1000nM
Luspatercept-2ug/ml
Galunisertib – 1000nM
IOA-359 – 1000nM +Luspatercept-2ug/ml
IOA-360 – 1000nM +Luspatercept-2ug/ml
Galunisertib - 1000nM + Luspatercept- 2ug/ml
CD71
Glycophorin
CD14
CD11B

Fig 5 – IOA-359 and IOA-360 augments erythroid differentiation in AML and MDS patient samples– The efficacy of the novel ALK5 inhibitors IOA-359 and IOA-360 was next tested in primary patient samples from MDS and AML and compared to Galunisertib in colonogenic assays. Increased erythroid colonies was observed in majority of MDS samples treated with both the ALK5 inhibitors IOA-359 and IOA-360. Colonies were picked and analyzed for erythroid and myeloid differentiation by FACS analysis. A representative flow plot of diverse patient samples (Fig 5A-D) shows enhanced erythroid and myeloid differentiation. Moreover, in specific cases, the inclusion of Luspatercept resulted in enhanced erythrocyte maturation. Representative flow cytometry plots shows enhanced erythroid differentiation with Luspatercept combinatorial therapy of AML1 with CEBPA, KMT20, MYC, NRAS, S1PR2, SMC3, TET2 and TGFBR2 mutations (5A). Representative flow cytometry plot shows erythroid and myeloid differentiation with Luspatercept combinatorial therapy of MDS Patient 1 with JAK2 and SF3B1 mutations (5B). Representative flow cytometry plot shows erythroid and myeloid differentiation with Luspatercept combinatorial therapy of AML Patient 2 with NRAS, TP53 mutations (5C). Representative flow cytometry plot shows erythroid and myeloid differentiation with Luspatercept combinatorial therapy of MDS Patient 2 with TET2 mutation (5D).

CONCLUSIONS

- ALK5 inhibitors IOA-359 and IOA-360 effectively inhibit TGF-β / SMAD2/3 downstream activation in a dose dependent manner in MDS and AML cell lines. Functionally, the ALK5 inhibitors did not lead to any increase in viability or proliferation in leukemic cell lines.
- ALK5 inhibitors reverse the growth inhibitory effects exerted by TGF-β on normal hematopoietic stem cells (HSCs), as demonstrated through CFU assay. Furthermore, treatment enhanced erythroid differentiation in MDS and AML patient samples with diverse mutational profiles.
- Our current results demonstrate the preclinical in vitro efficacy of ALK5 inhibitors IOA-359 and IOA-360, highlighting their potential for further development and clinical testing in MDS/AML.

CONTACT INFORMATION

Nandini Ramachandra MS :- nandini.ramachandra @einsteinmed.edu
Charan Thej Reddy Vegivinti MD- :- cvegivinti@montefiore.org / Twitter - @CharanVegivinti
Michael Lahn MD:- m.lahn@ionctura.com
Amit Verma MD :- amit.verma@einsteinmed.edu