A novel autotaxin inhibitor, IOA-289, modulates tumor, immune and stromal cell function and has monotherapy activity in fibrotic cancer models

Marcel A. Deken[1], Karolina Niewola-Staszkowska[1], Elisa Matas-Rico[2], Amy Fraser[3], Ragini Medhi[3], Alan Carruthers[3], Anne Cheasty[3], Olivier Peyruchaud[4], Wouter H. Moolenaar[2], Pritom Shah[3], Lars van der Veen[1], Michael M. Lahn[1] & Zoë Johnson[1]

1. iOnctura SA, Geneva, Switzerland; 2. Division of Biochemistry, Netherlands Cancer Institute, Amsterdam, The Netherlands; 3. Cancer Research UK Therapeutic Discovery Laboratories, Babraham Research Campus, Cambridge, UK; 4. INSERM U.1033 Faculté de Médecine Lyon, France.

Background

Autotaxin (ATX) is a secreted glycoprotein that hydrolyzes lysophosphatidylcholine (LPC) to lysophosphatidic acid (LPA). The expression of both ATX and LPA is elevated in most solid tumors and plasma. LPA signaling directly modulates tumor cell function and contributes to the development of the fibrotic tumor microenvironment, which is a mechanism by which tumors evade host immunity and impairs response to therapy. IOA-289 is a potent and orally available ATX inhibitor, being developed as a novel treatment of solid tumours burdened with a high degree of fibrosis. IOA-289 has completed a phase I clinical trial in healthy volunteers with an acceptable safety and PK profile supporting further clinical development.

Methods

PK/PD studies were performed following a single oral dose of IOA-289 in mice, and plasma LPA was used as a PD biomarker. Inhibition of ATX activity in human plasma was determined by measuring reduction in LPA species as quantified by LC-MS/MS. In vitro activity on biomarkers of fibrosis was assessed using the BioMAP screen and fibroblast cell cultures. T cell migration was measured using 48-well chemotaxis chambers. In vitro activity on fibrotic processes was characterized in the BioMAP® Fibrosis panel. In vivo efficacy was studied in two mouse models of breast cancer, 4T1 and E0771. An ELISA assay was used to analyse CA19-9 and ATX levels in human plasma samples. Bioinformatic analysis on gene expression used TCGA and GTEX publicly available datasets.

IOA-289 inhibits plasma LPA

Male CD1 mice dosed with 3, 10 or 30 mg/kg of IOA-289 p.o. showed dose-dependent reduction of circulating LPA C18:2 with an ED_{50} value at 1 h post-dose of around 3 mg/kg (Figure 1A and B).

Figure 1

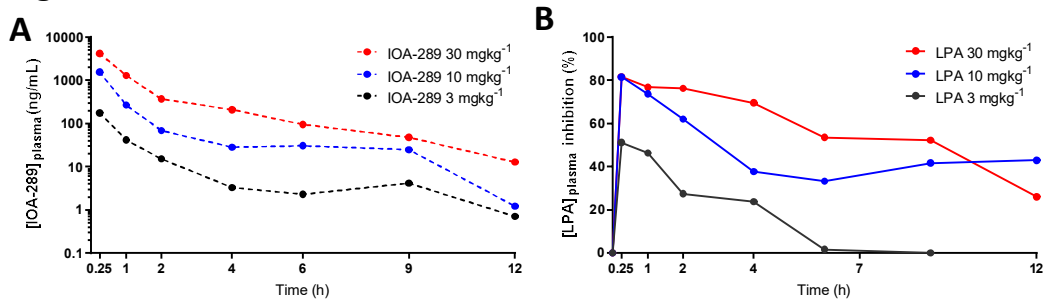
A
B
IOA-289 30 mgkg-1
IOA-289 10 mgkg-1
IOA-289 3 mgkg-1
[IOA-289] plasma (ng/mL)
LPA 30 mgkg-1
LPA 10 mgkg-1
LPA 3 mgkg-1
[LPA] plasma inhibition (%)
Time (h)

Tumor-mediated inhibition of T-cell migration is reversed by ATXi

MDA-MB-435 cells secrete high levels of ATX and conditioned medium from MDA-MB-435 cell cultures inhibits CD8+ T cell migration. Inhibition of ATX by IOA-289 reverses the inhibitory effect of MDA-MB-235 conditioned media on CD8+ T cell migration. CXCL10 is used as a positive control (Figure 2).

Figure 2

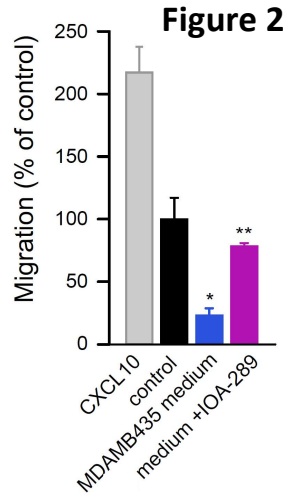
Migration (% of control)
CXCL10
control
MDAMB435 medium
medium +IOA-289

IOA-289 is a potent modulator of fibrotic processes *in vitro*

The BioMAP® Fibrosis panel consists of systems that models fibrotic tissues and disease by co-culturing stimulated human primary cells; lung epithelial cells and fibroblasts, lung fibroblasts only or renal epithelial cells with lung fibroblasts. IOA-289 was tested at concentrations of 6, 1.2 and 0.24 µM. At 6 µM IOA-289 decreased the activity of fibrosis relevant factors, including sIL-6, MCP-1, αSMA, collagen-III, and sVEGF (Figure 3).

Figure 3

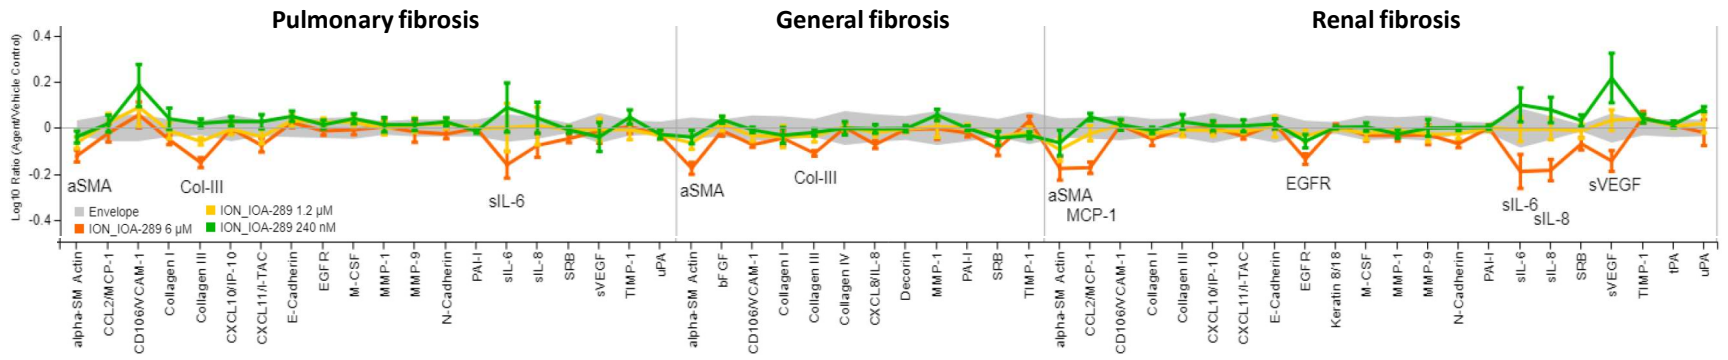
Pulmonary fibrosis
General fibrosis
Renal fibrosis
aSMA
Col-III
sIL-6
MCP-1
EGFR
sIL-8
sVEGF
Envelope
ION_IOA-289 6 µM
ION_IOA-289 1.2 µM
ION_IOA-289 240 nM

IOA-289 inhibits metastasis in 4T1 model of breast cancer

Balb/c mice were orthotopically injected in the mammary fatpad with 4T1 tumor cells. One day before tumor inoculation, BID treatment with IOA-289 by oral gavage started. Fifteen days after tumor inoculation, the primary tumors were surgically removed and mice were taken off treatment. Primary tumor growth was assessed by weighing the excised tumor and did not show an effect (Figure 4A). In an independent experiment, histological analysis showed marked infiltration of CD8+ T cells (Figure 4B). Twenty days after removal of the primary tumor, lungs and bone marrow cells from the mice were harvested. Histological analysis of the lungs showed a marked reduction in metastatic burden for IOA-289 but not PF-8380 (Figure 4C). Colony formation assay from cultured bone marrow showed marked reduction in metastases by IOA-289 and PF-8380 (Figure 4D).

Figure 4

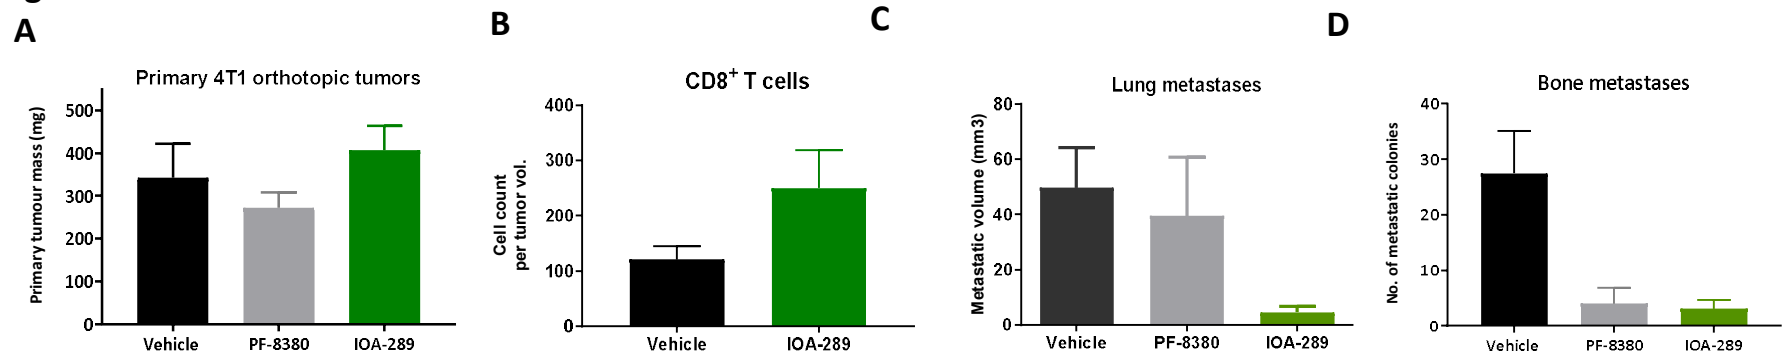
A
Primary 4T1 orthotopic tumors
Primary tumour mass (mg)
B
CD8+ T cells
Cell count per tumor vol.
C
Lung metastases
Metastatic volume (mm3)
D
Bone metastases
No. of metastatic colonies
Vehicle
PF-8380
IOA-289

IOA-289 inhibits tumor outgrowth in E0771 model of breast cancer

C57BL/6 mice were orthotopically injected in the mammary fatpad with E0771 cells and treatment by oral BID started on day 3 and continued throughout the experiment. IOA-289 inhibited tumor outgrowth and induced two mice with complete tumor eradication (Figure 5A-C).

Figure 5

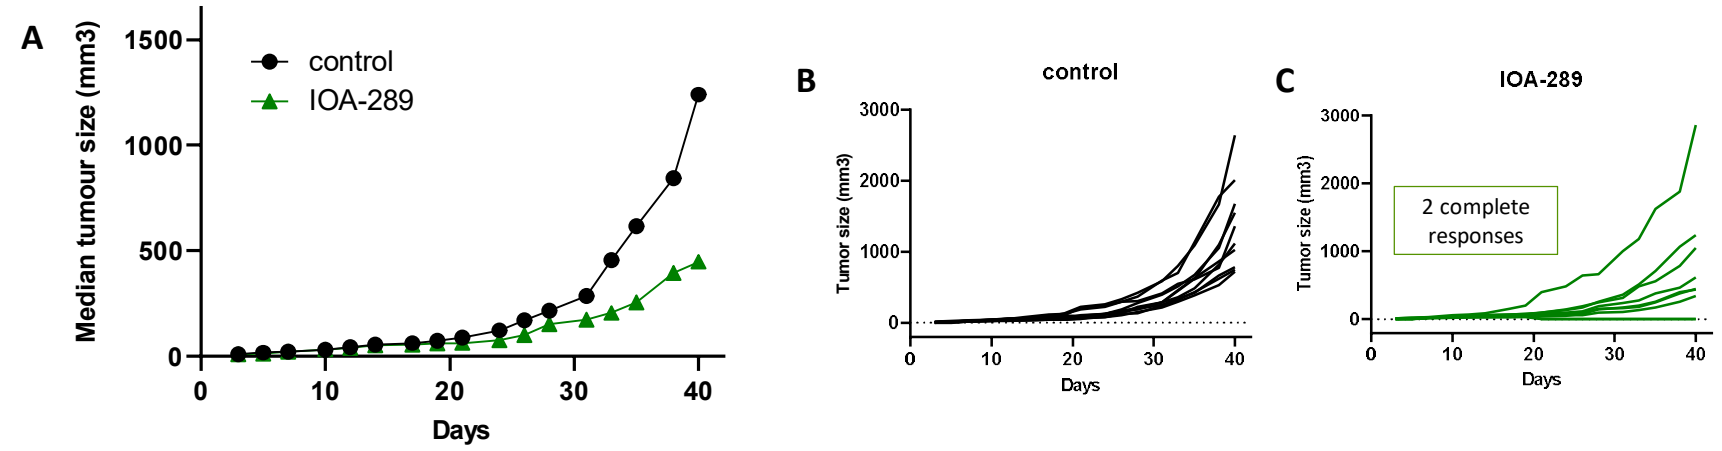
A
Median tumour size (mm3)
control
IOA-289
Days
B
control
Tumor size (mm3)
C
IOA-289
2 complete responses

ATX is highly expressed in PDAC

Analysis of ATX (genename ENPP2) expression in various cancers and sample types using TCGA and GTEX data indicated that ENPP2 expression is increased in tumor tissue in comparison to normal tissue in most cases. Pancreatic adenocarcinoma (PDAC) was identified as a indication of interest with particular occurrence of high ENNP2 expression in tumor tissue compared to normal tissue (Figure 6).

Figure 6

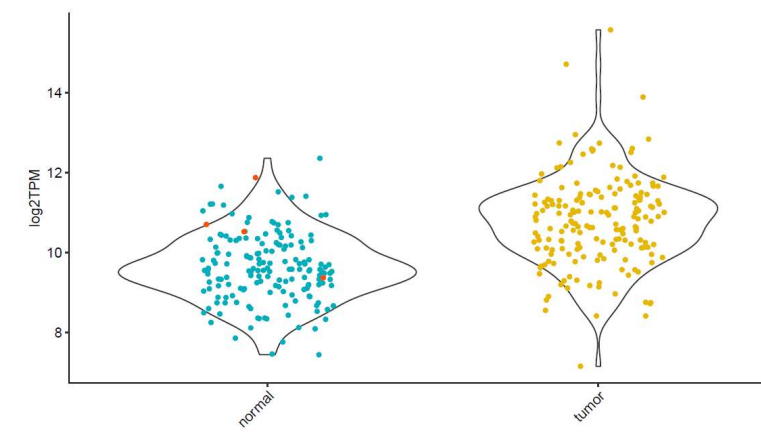
log2TPM
normal
tumor

ATX correlates with PDAC marker CA19-9

Plasma samples from patients with pancreatic cancer (PANC) and NSCLC were analyzed by standard ELISA for CA19-9 levels. Pooled human plasma samples from were used as control. Elevated levels of CA19-9 were found in PANC samples, and to a lesser extend in NSCLC samples compared to control (Figure 7A). ELISA analysis of the same samples for levels of ATX showed a potential correlation between CA19-9 and ATX in pancreatic cancer (Figure 7B) but not NCLSC samples (Figure 7C).

Figure 7

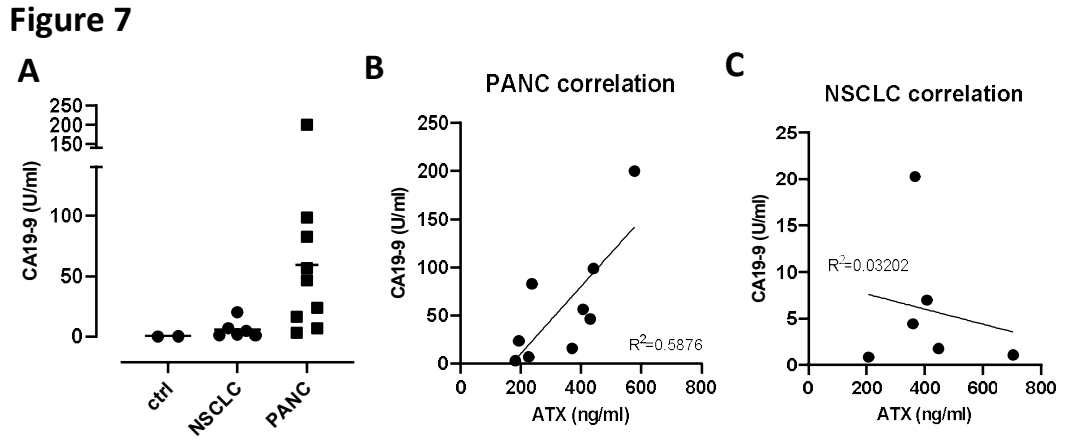
A
CA19-9 (U/ml)
ctrl
NSCLC
PANC
B
PANC correlation
R2=0.5876
ATX (ng/ml)
C
NSCLC correlation
R2=0.03202

Conclusion

The ATX/LPA pathway represents a novel target for anti-cancer therapy with actions on the tumor and immune cells and on the stromal environment. IOA-289 is a highly potent and selective inhibitor of ATX with demonstrated effects on T cell migration and fibrotic processes in vitro. IOA-289 has monotherapy activity in multiple fibrosis and cancer models, including breast and pancreatic cancer. Based on the mechanism of action we are investigating combinations of IOA-289 with chemotherapy, immunotherapy and novel agents in ongoing preclinical studies. IOA-289 has completed a phase I clinical trial in healthy volunteers with an acceptable safety and PK profile supporting further clinical development.

Conflict of interest statement:
MD, KNS, LvdV, ML and ZJ are employees and shareholders of iOnctura

z.johnson@ionctura.com

SITC | 36th Annual Meeting | Nov 10-14, 2021