AACR Poster # 1877

IOA-237 – A potent anti-CD73 mAb demonstrates monotherapeutic and combination efficacy in solid tumor models

Marcel A. Deken[1], Alan Carruthers[2], Karolina Niewola-Staszkowska[1], Grainne Gernon[2], Hilary Sandig[2], Pritom Shah[2], Michael Lahn[1] & Zoë Johnson[1]

1. iOnctura SA, Avenue de Sécheron 15, 1202 Geneva, Switzerland; 2. Cancer Research UK Therapeutic Discovery Laboratories, Babraham Research Campus, Cambridge, CB22 3AT, UK

Background

CD73, an ecto-5'-nucleotidase, is frequently overexpressed in the tumor microenvironment and can be found on tumor, stromal cells and immune cells. Its function is to catalyse the conversion of adenosine monophosphate (AMP) to adenosine and phosphate, both at the cell membrane and as a soluble factor. CD73 has gained interest as a therapeutic target in cancer due to the immunosuppressive role of adenosine in the tumor microenvironment. High levels of adenosine cause an immunosuppressive microenvironment in the tumor by signalling via adenosine receptors on a multitude of immune cells.

IOA-237 is a fully human, phage derived monoclonal antibody with high potency against CD73. Inhibition of CD73 by IOA-237 results in reduced levels of adenosine thereby limiting tumor immune suppression. This novel antibody demonstrates in vitro and in vivo characteristics that suggest best-in-class potential (Figure 1).

Figure 1

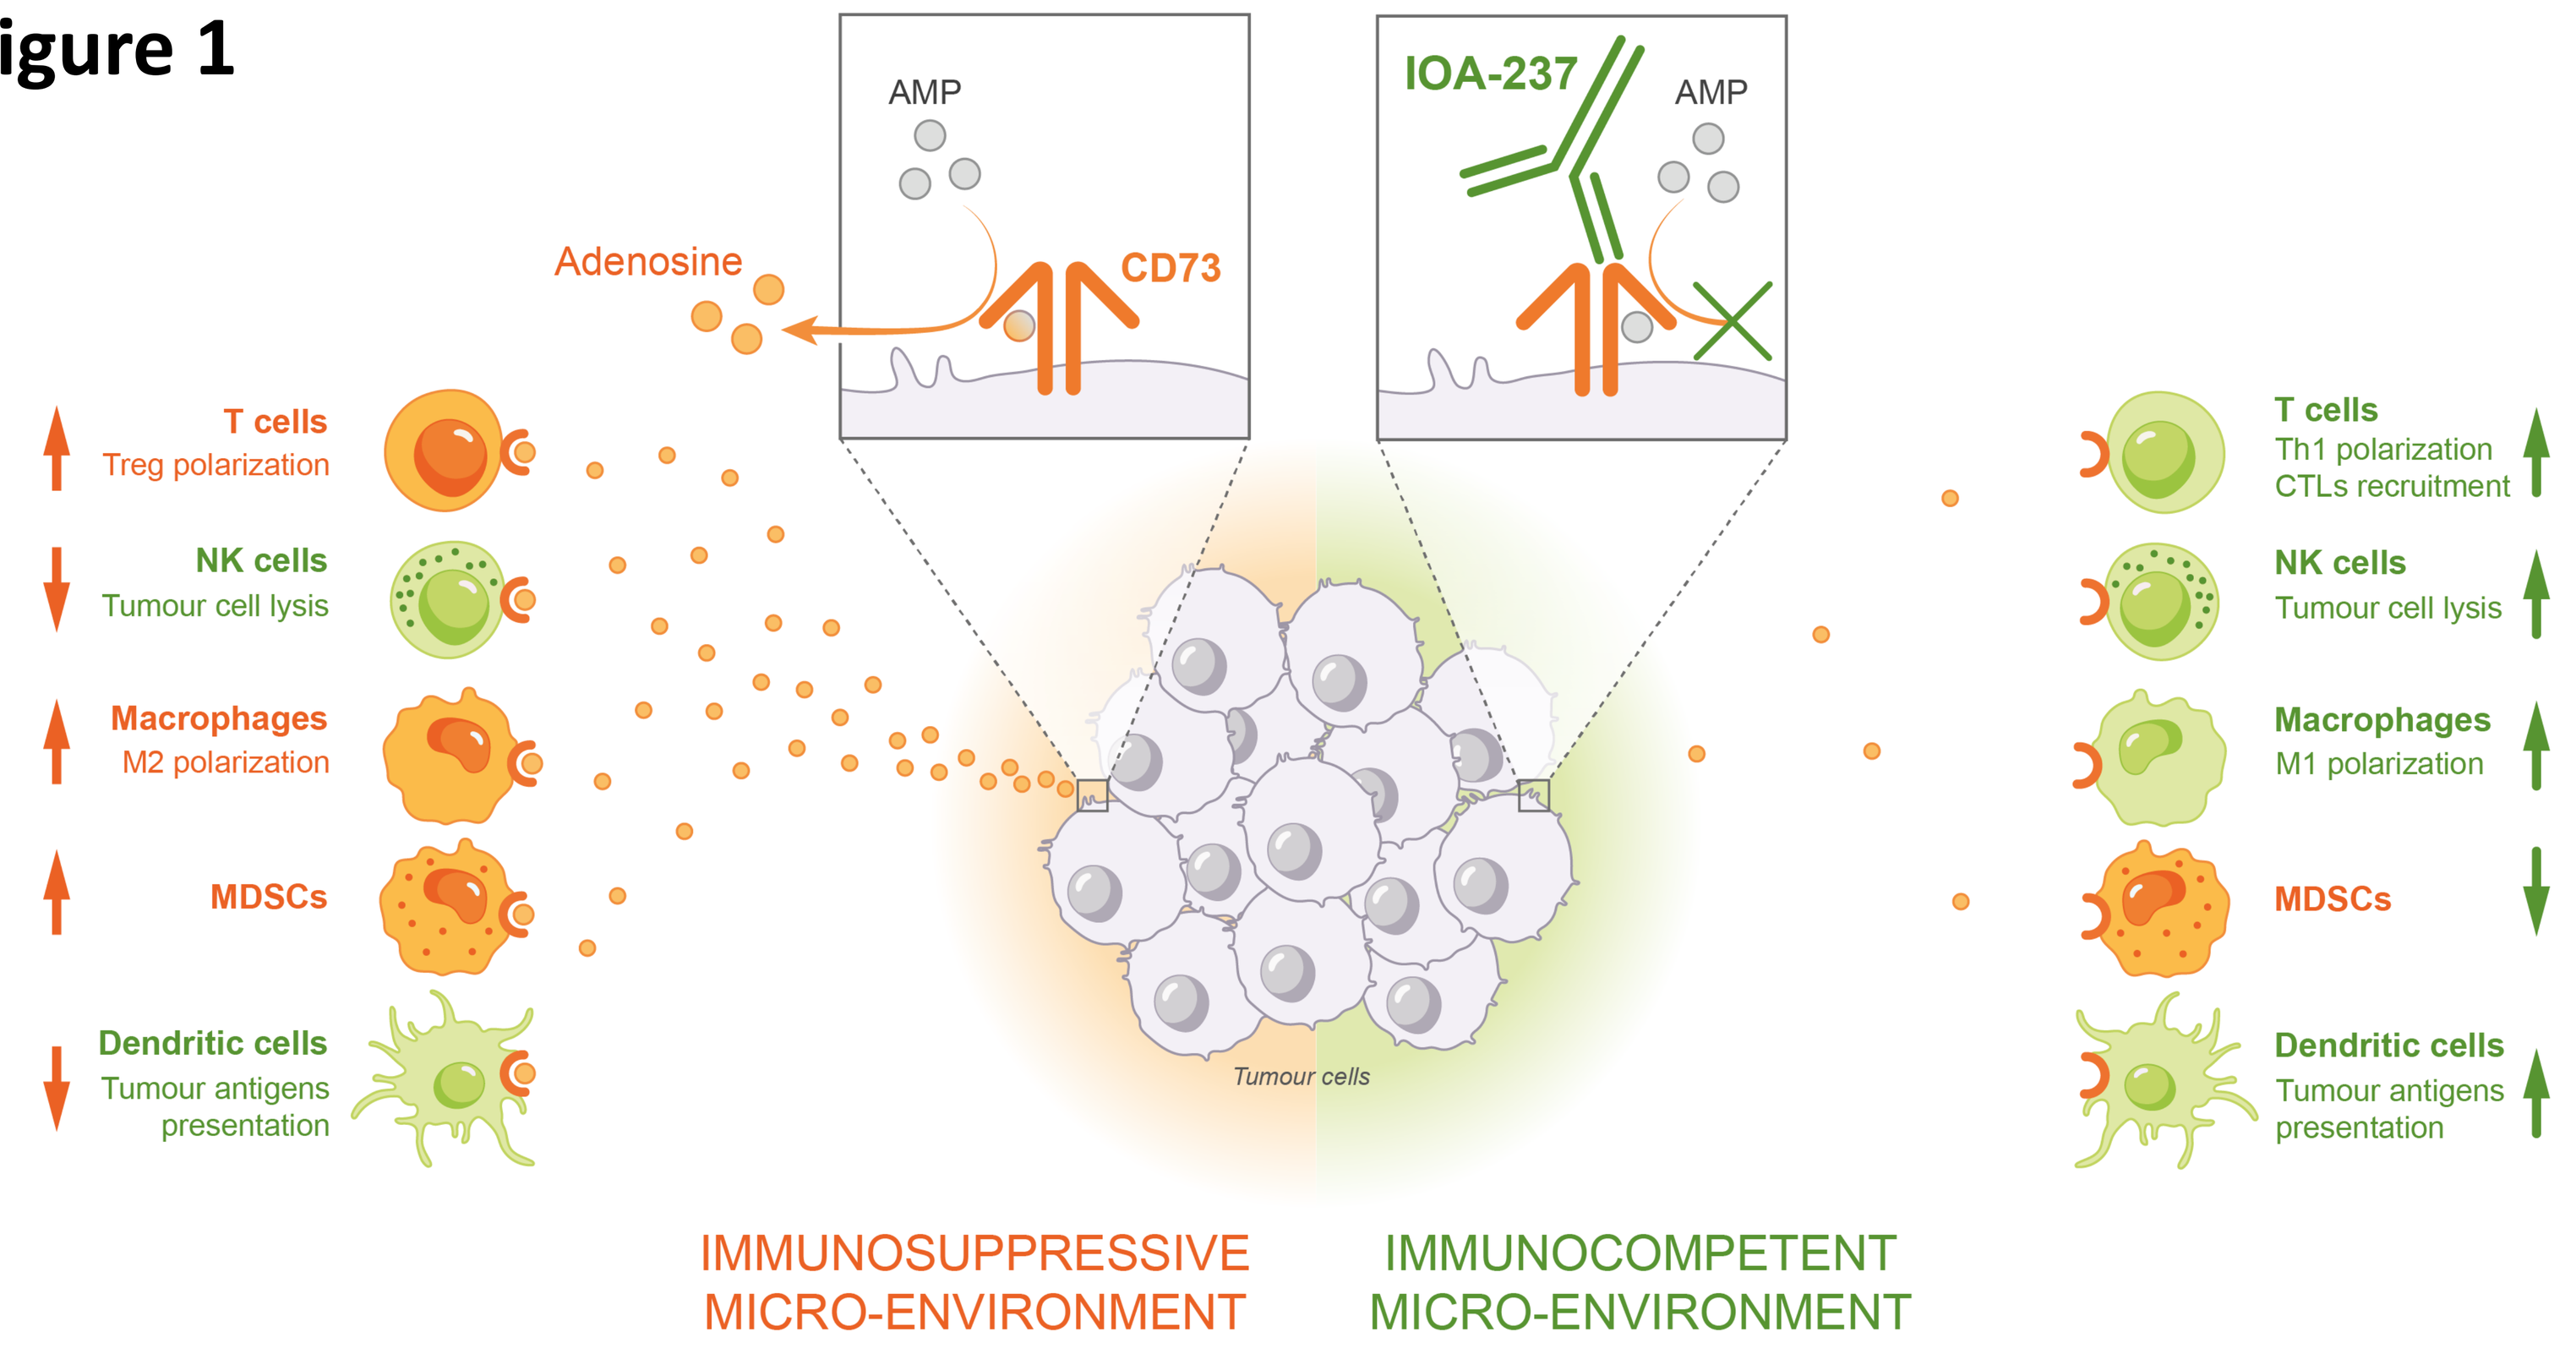
AMP
Adenosine
CD73
IOA-237
AMP
T cells
Treg polarization
NK cells
Tumour cell lysis
Macrophages
M2 polarization
MDSCs
Dendritic cells
Tumour antigens presentation
Tumour cells
T cells
Th1 polarization
CTLs recruitment
NK cells
Tumour cell lysis
Macrophages
M1 polarization
MDSCs
Dendritic cells
Tumour antigens presentation
IMMUNOSUPPRESSIVE MICRO-ENVIRONMENT
IMMUNOCOMPETENT MICRO-ENVIRONMENT

Methods

To evaluate the pharmacokinetics of IOA-237, the antibody was injected intravenously in female Balb/c mice at a concentration of 10mg/kg. MEDI9447 was administrated intravenously at 10mg/kg. The concentration of antibodies present in serum was measured by ELISA using anti-Human IgG, Fcγ fragment specific goat pAb as capture antibodies and anti-human λ chain-HRP mouse mAb for detection.

For hypersensitivity and efficacy studies, Balb/c (n=5/group) were subcutaneously implanted with CT26 cells and treatment with IOA-237 (10mg/kg) was started at day 3 over 4 twice weekly administrations. In the follow-up study, CT26 tumor-bearing mice (n=12/group) were treated with isotype, IOA-237 or MEDI9447 (10mg/kg). Female C57BL/6 mice (n=10/group) were orthotopically injected in the mammary fatpad with E0771 cells and twice weekly treatment with isotype or IOA-237 (10mg/kg) started on day 3 and continued throughout the experiment. Isotype control groups received human isotype antibody. AUC was calculated with an unpaired t-test.

IOA-237 has a favorable PK profile

Figure 2

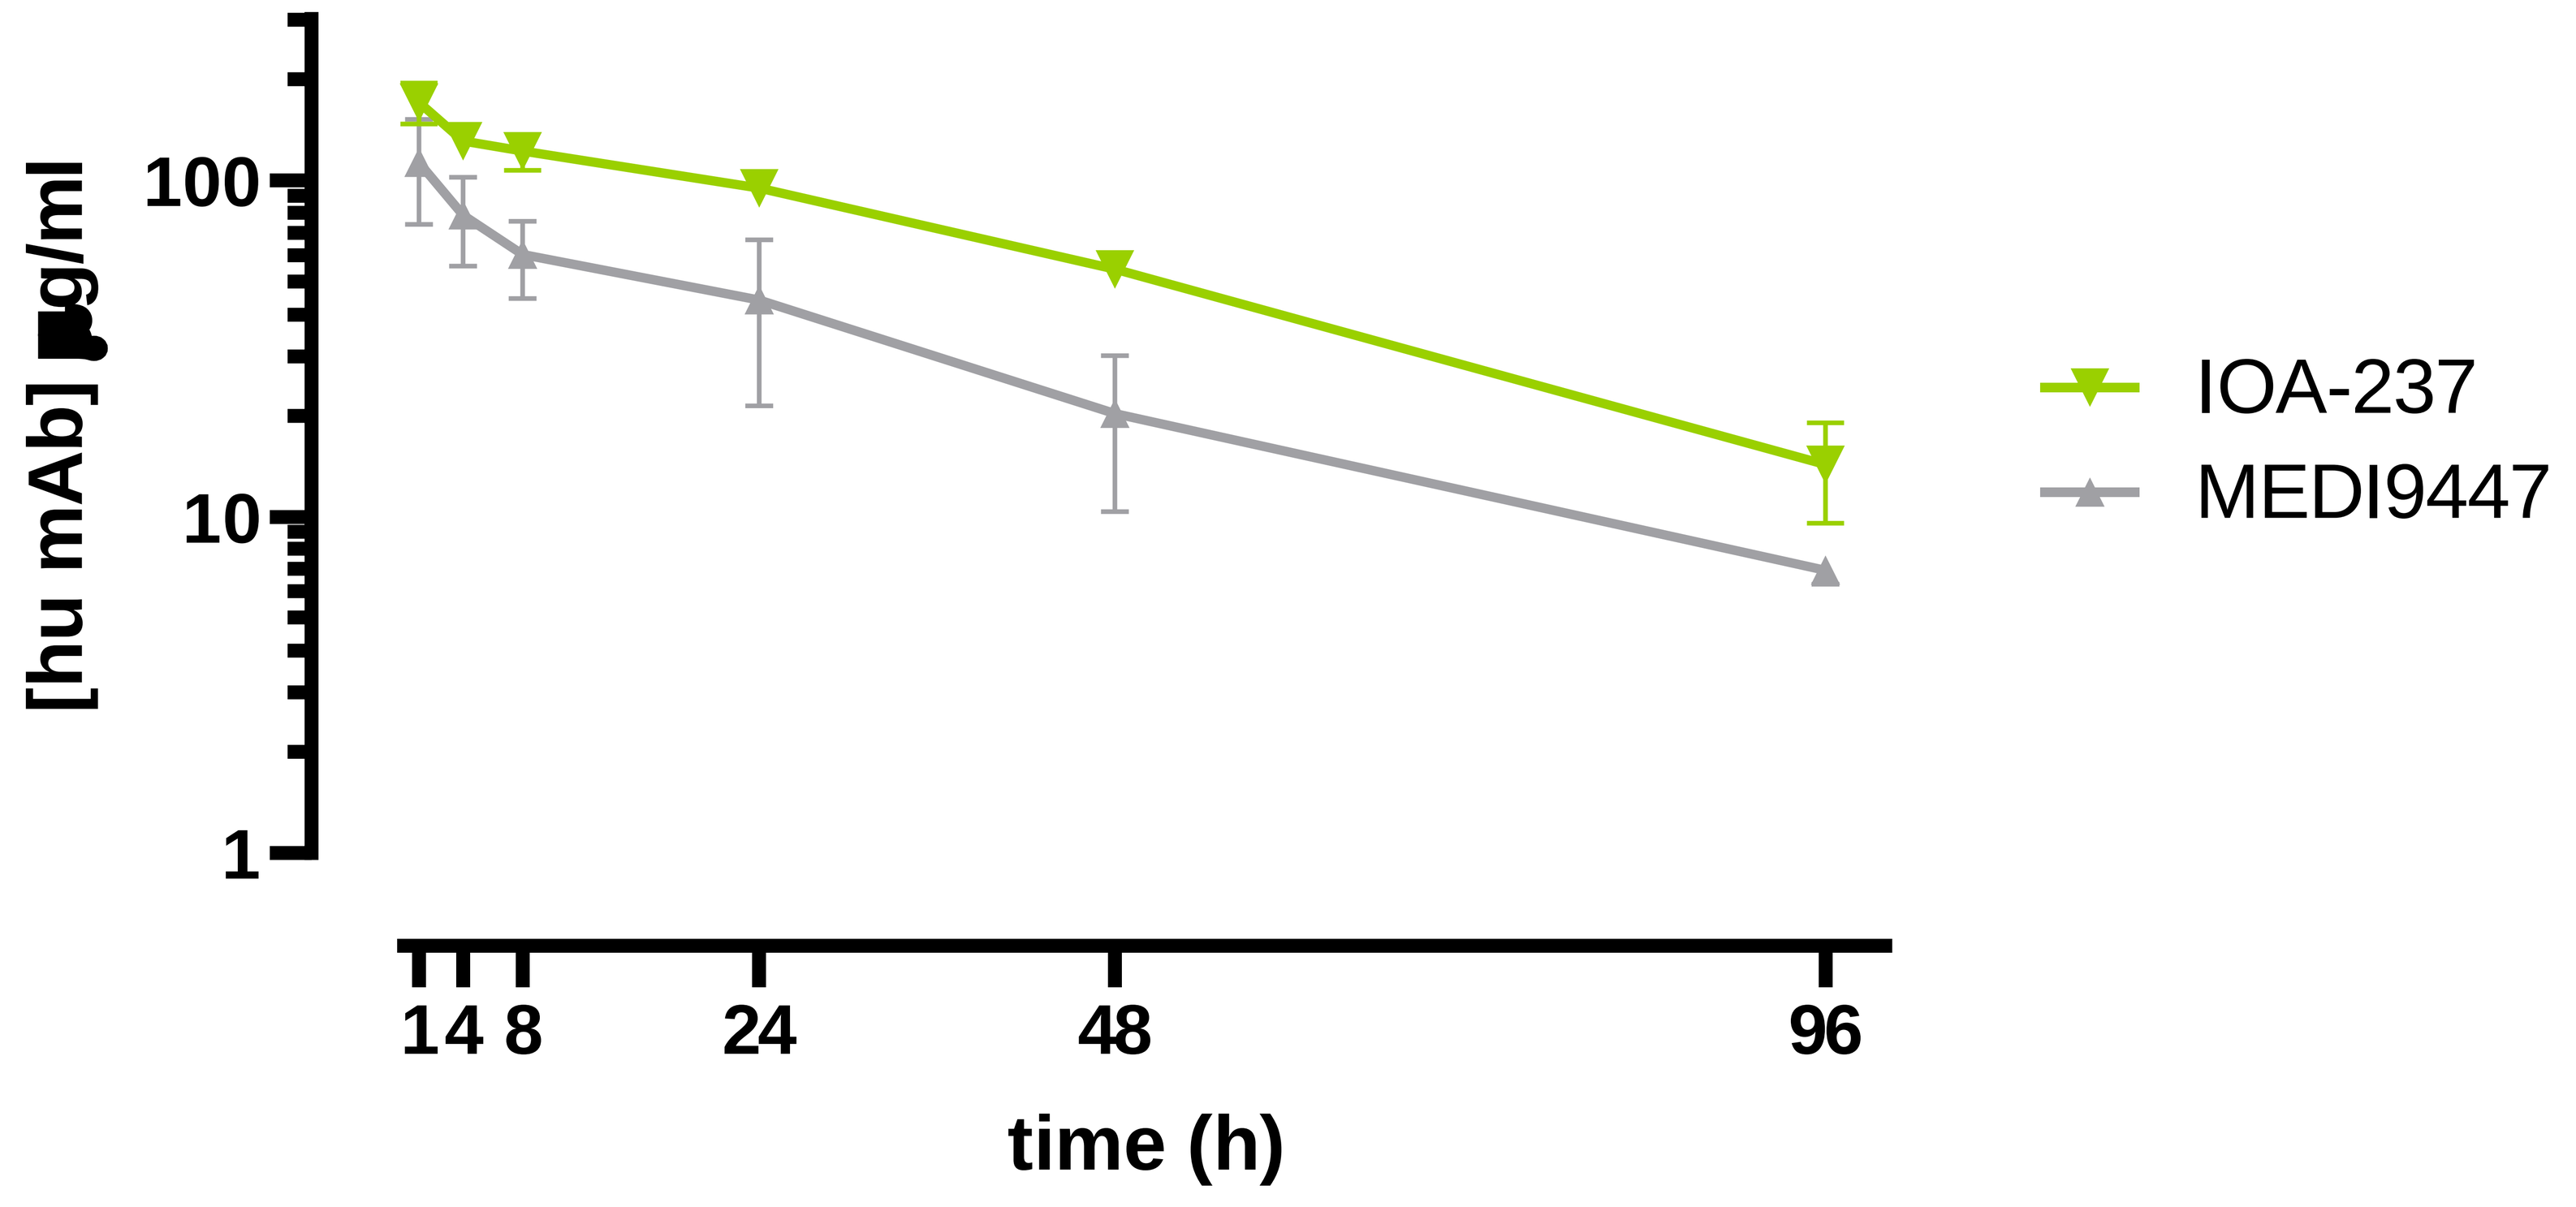
[hu mAb] µg/ml
100
10
1
1 4 8
24
48
96
time (h)
IOA-237
MEDI9447

The pharmacokinetic profile of IOA-237 showed to be favorable over the clinical stage competitor MEDI9447 when administrated intravenously to Balb/c mice. (Figure 2).

IOA-237 does not induce hypersensitivity and shows efficacy in mouse model of colon cancer

Figure 3

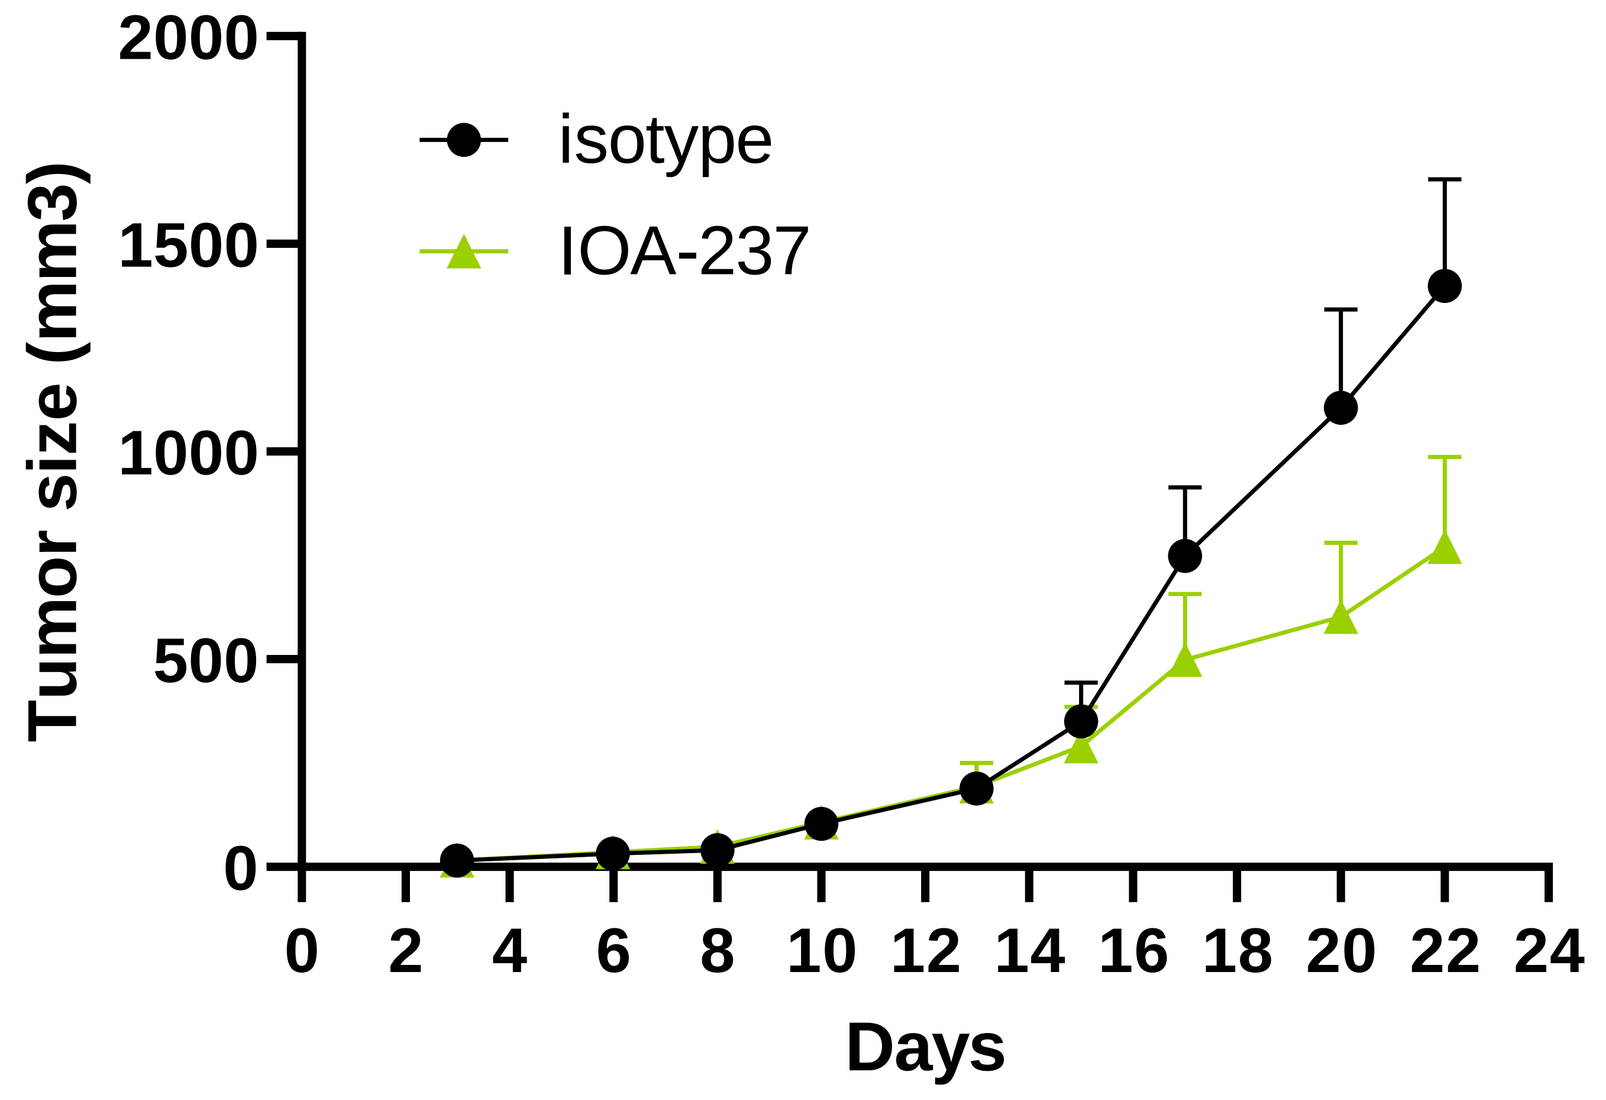
Tumor size (mm3)
2000
1500
1000
500
0
0 2 4 6 8 10 12 14 16 18 20 22 24
Days
isotype
IOA-237

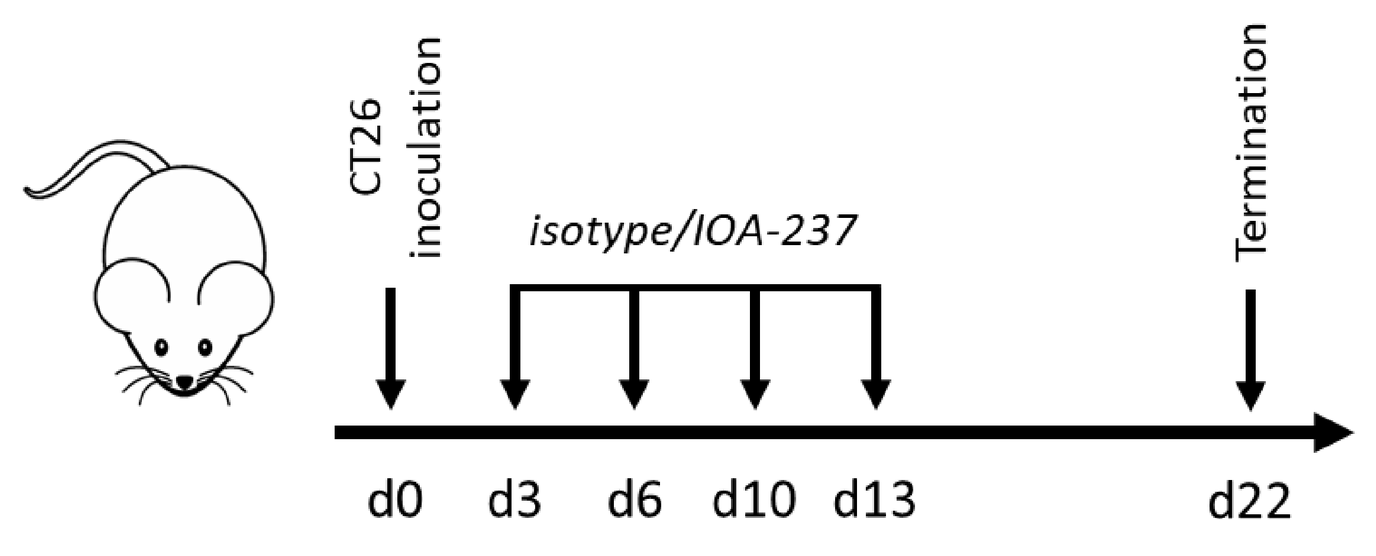
CT26 inoculation
isotype/IOA-237
Termination
d0 d3 d6 d10 d13 d22

To test potential induction of hypersensitivity, IOA-237 was administered to CT26 tumor-bearing Balb/c mice. IOA-237 did not induce hypersensitivity at the treatment schedule used in and showed a trend towards reduced tumor outgrowth (Figure 3).

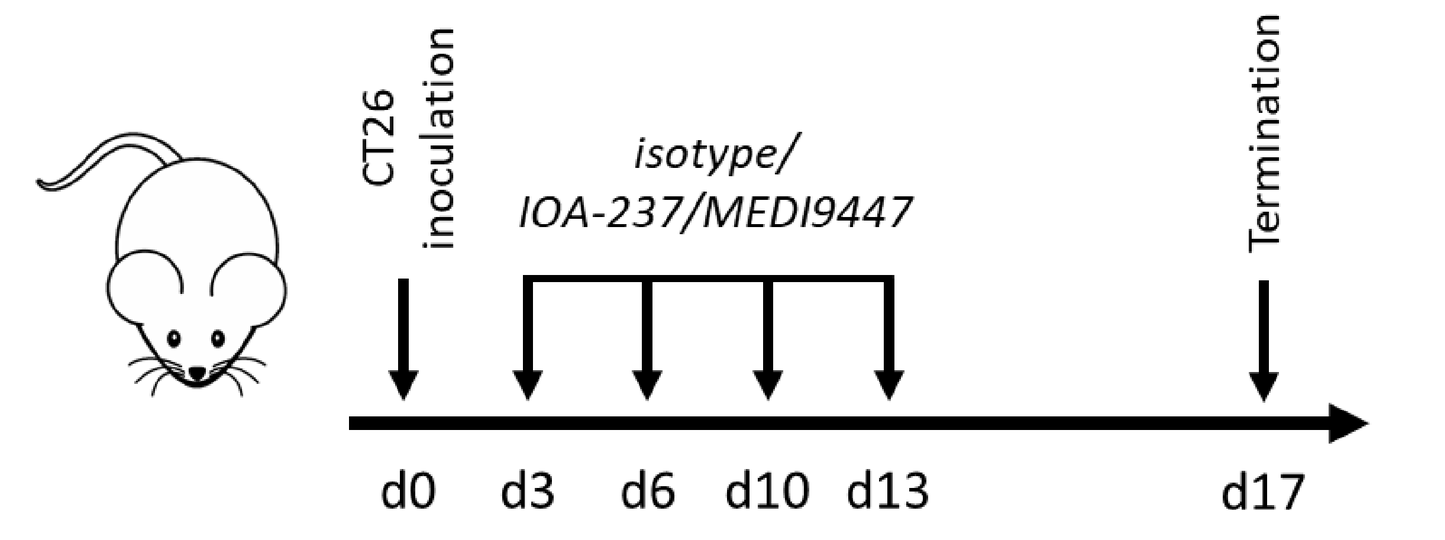
CT26 inoculation
isotype/ IOA-237/MEDI9447
Termination
d0 d3 d6 d10 d13 d17

To directly compare IOA-237 efficacy to MEDI9447, CT26 tumor-bearing Balb/c mice were administered with isotype or the respective antibodies. IOA-237 and MEDI9447 showed a comparable trend towards reduction in tumor outgrowth (Figure 4).

Figure 4

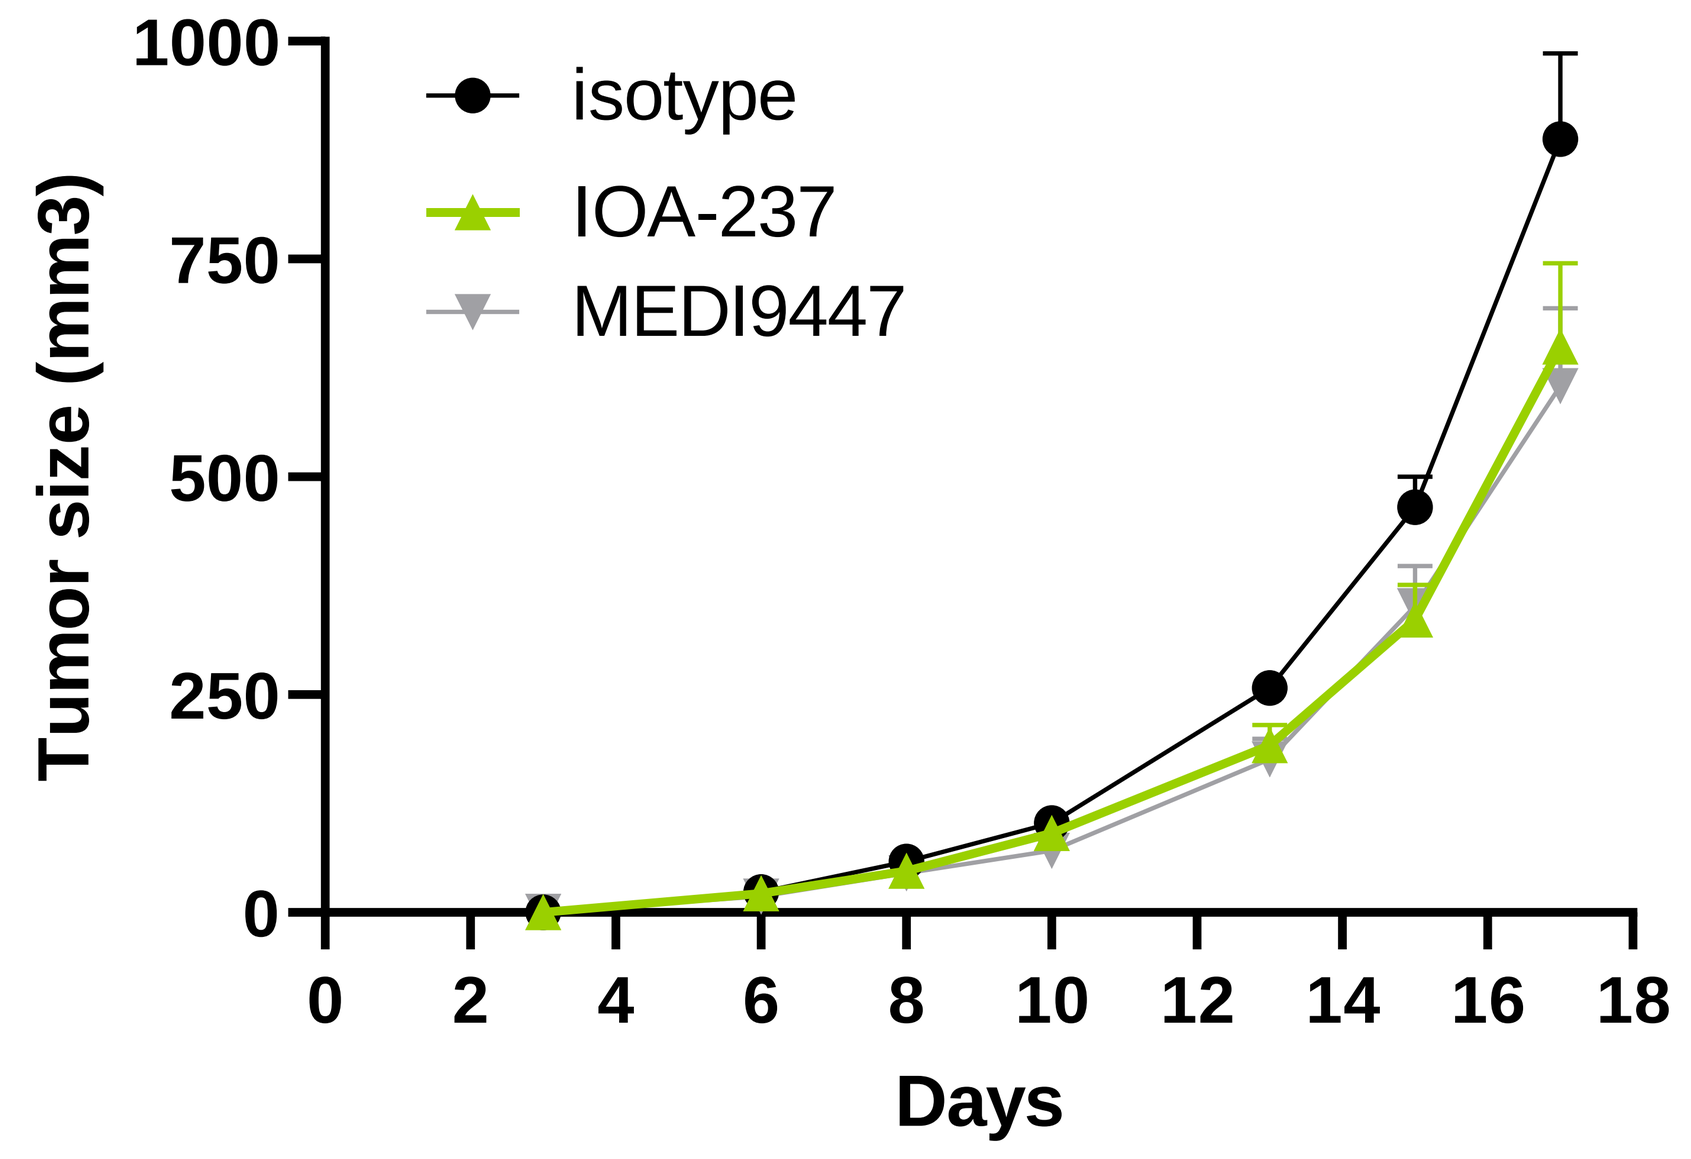
Tumor size (mm3)
1000
750
500
250
0
0 2 4 6 8 10 12 14 16 18
Days
isotype
IOA-237
MEDI9447

Single agent activity of IOA-237 in the E0771 orthotopic mouse model of breast cancer

Figure 5

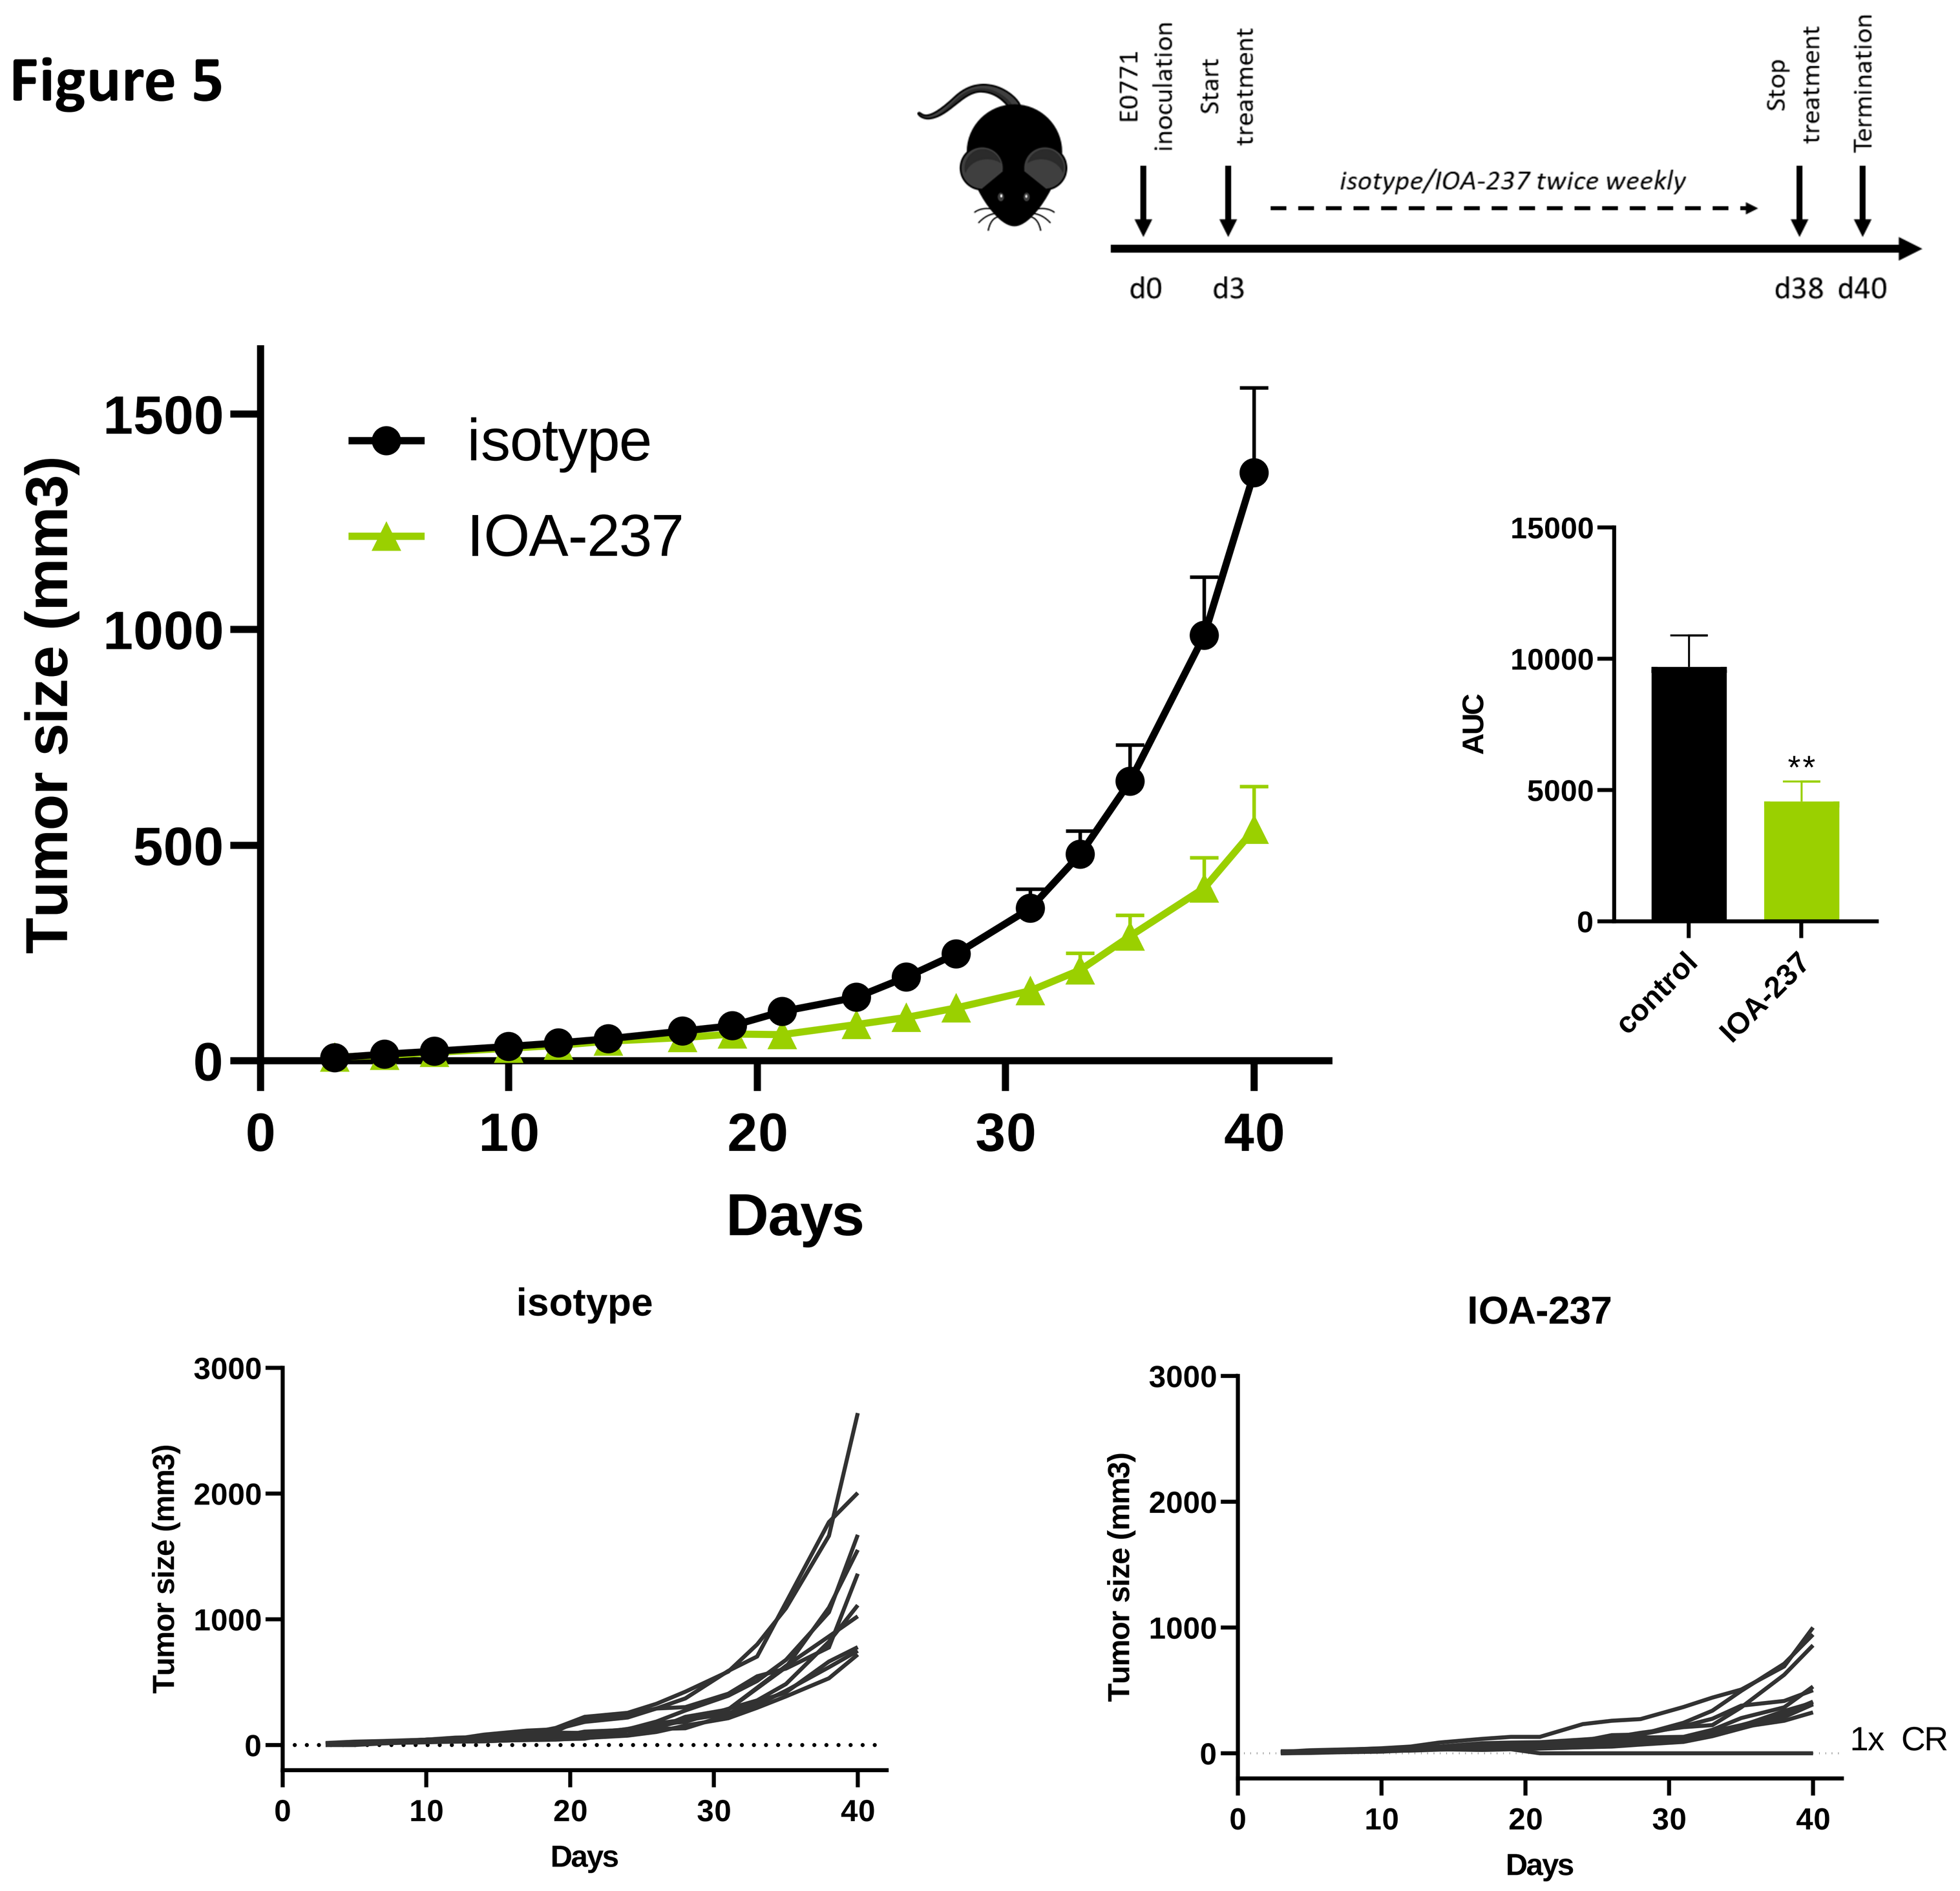
E0771 inoculation
Start treatment
isotype/IOA-237 twice weekly
Stop treatment
Termination
d0 d3 d38 d40
Tumor size (mm3)
1500
1000
500
0
0 10 20 30 40
Days
isotype
IOA-237
AUC
15000
10000
5000
0
**
control
IOA-237
isotype
IOA-237
Tumor size (mm3)
3000
2000
1000
0
0 10 20 30 40
Days
1x CR

In the orthoptic E0771 mouse model of triple negative breast cancer, IOA-237 twice weekly and continuously administered at 10mg/kg showed statistically significant inhibition of tumor outgrowth as measured by area under the curve (AUC). In the IOA-237 treated group one animal showed initial outgrowth of the tumor but regressed upon prolonged treatment and resulted into a complete responder (Figure 5).

Conclusion

- IOA-237 is a novel and potent anti-CD73 mAb that can be used to block adenosine generation in the tumor microenvironment.
- IOA-237 has a favorable PK profile when comparing intravenous administration to MEDI9447.
- No hypersensitivity was induced by the fully human antibody when used in twice weekly administration to tumor-bearing Balb/c mice and continuous twice weekly administration to tumor-bearing C57BL/6 mice.
- IOA-237 showed a trend towards reduced tumor outgrowth in the subcutaneous CT26 mouse model of colon carcinoma and showed comparable efficacy as MEDI9447.
- IOA-237 showed statistically significant inhibition of tumor outgrowth in the orthotopic E0771 mouse model of triple negative breast cancer.
- Efficacy studies to test combinations with checkpoint inhibitors and other agents are ongoing.

Conflict of Interest Statement:
MD, KNS, ML and ZJ are employees and shareholders of iOnctura.

m.deken@ionctura.com / z.johnson@ionctura.com

AACR 2021|Virtual Annual Meeting | April 9-14, 2021